Accelerating spectral flow cytometry assay development for clinical trials with a flexible panel design

Xavier Dagenais-Lussier, Bader Yassine Diab, Maude Boisvert, Matt Clutter

POSTER NUMBER: P112

CellCarta

INTRODUCTION

Flow cytometry provides valuable clinical trial biomarker information, but the antibody panels used often require customization to capture the most relevant readouts for a particular therapy. Custom panel development, especially for spectral cytometry, is a resource-intensive process that offers limited flexibility once validated. To address this challenge, we developed a **modular approach** that streamlines assay validation when sets of interchangeable markers are modified. The two main aspects needed for the success of this panel design are as follow:

- A small Core panel designed to be **insulated** from changes to the modules, which can be used on **different matrices** (PBMCs, Whole Blood).
- For the modules, we need to find bright fluorophores to accommodate potentially dim markers, which are also **widely available**/conjugated to different antibodies.

The resulting panel **de-risks** and **accelerates** new panel design phase.

Study Design and Methodology

The following process was done to assemble a modular flow cytometry panel on cryopreserved human PBMCs. Acquisition was performed using Cytek® Aurora 5 Lasers Spectral Flow Cytometer, unmixed using SpectroFlo® Software v3.3.0 and analyzed on CellEngine:

1. Fluorophore selection and assignment.
2. Titration of all reagents for selection of titers with highest Signal-to-Noise ratios. In-vitro stimulated PBMC were used for Dim-expressed markers.
3. Full panel testing was performed. At this step, a full set of single stain references (cells and beads) were generated to optimize unmixing.
4. FMO control testing for fluorescence spill over and staining specificity.
5. Full panel testing for intra-assay precision and stability assessments.

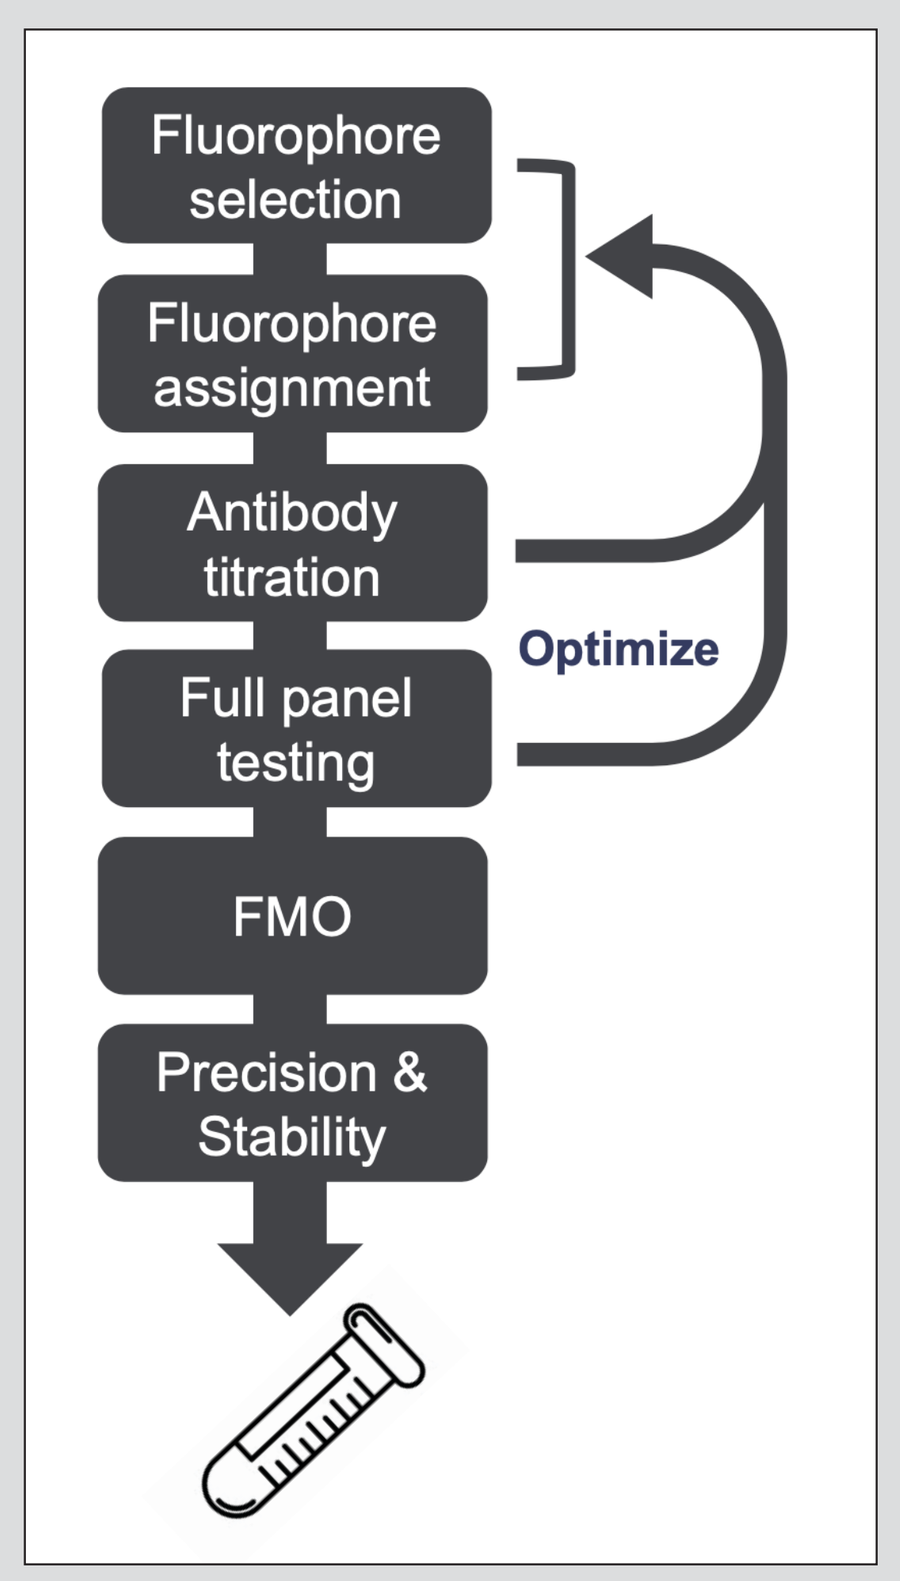
Fluorophore selection
Fluorophore assignment
Antibody titration
Full panel testing
Optimize
FMO
Precision & Stability

Iterative fluorophore assignment

Leveraging both online resources (such as CytekCloud and BD Research cloud) for stain index ranking and our internal surface profiling (see below), we can assign bright fluorophores to lowly expressed markers.

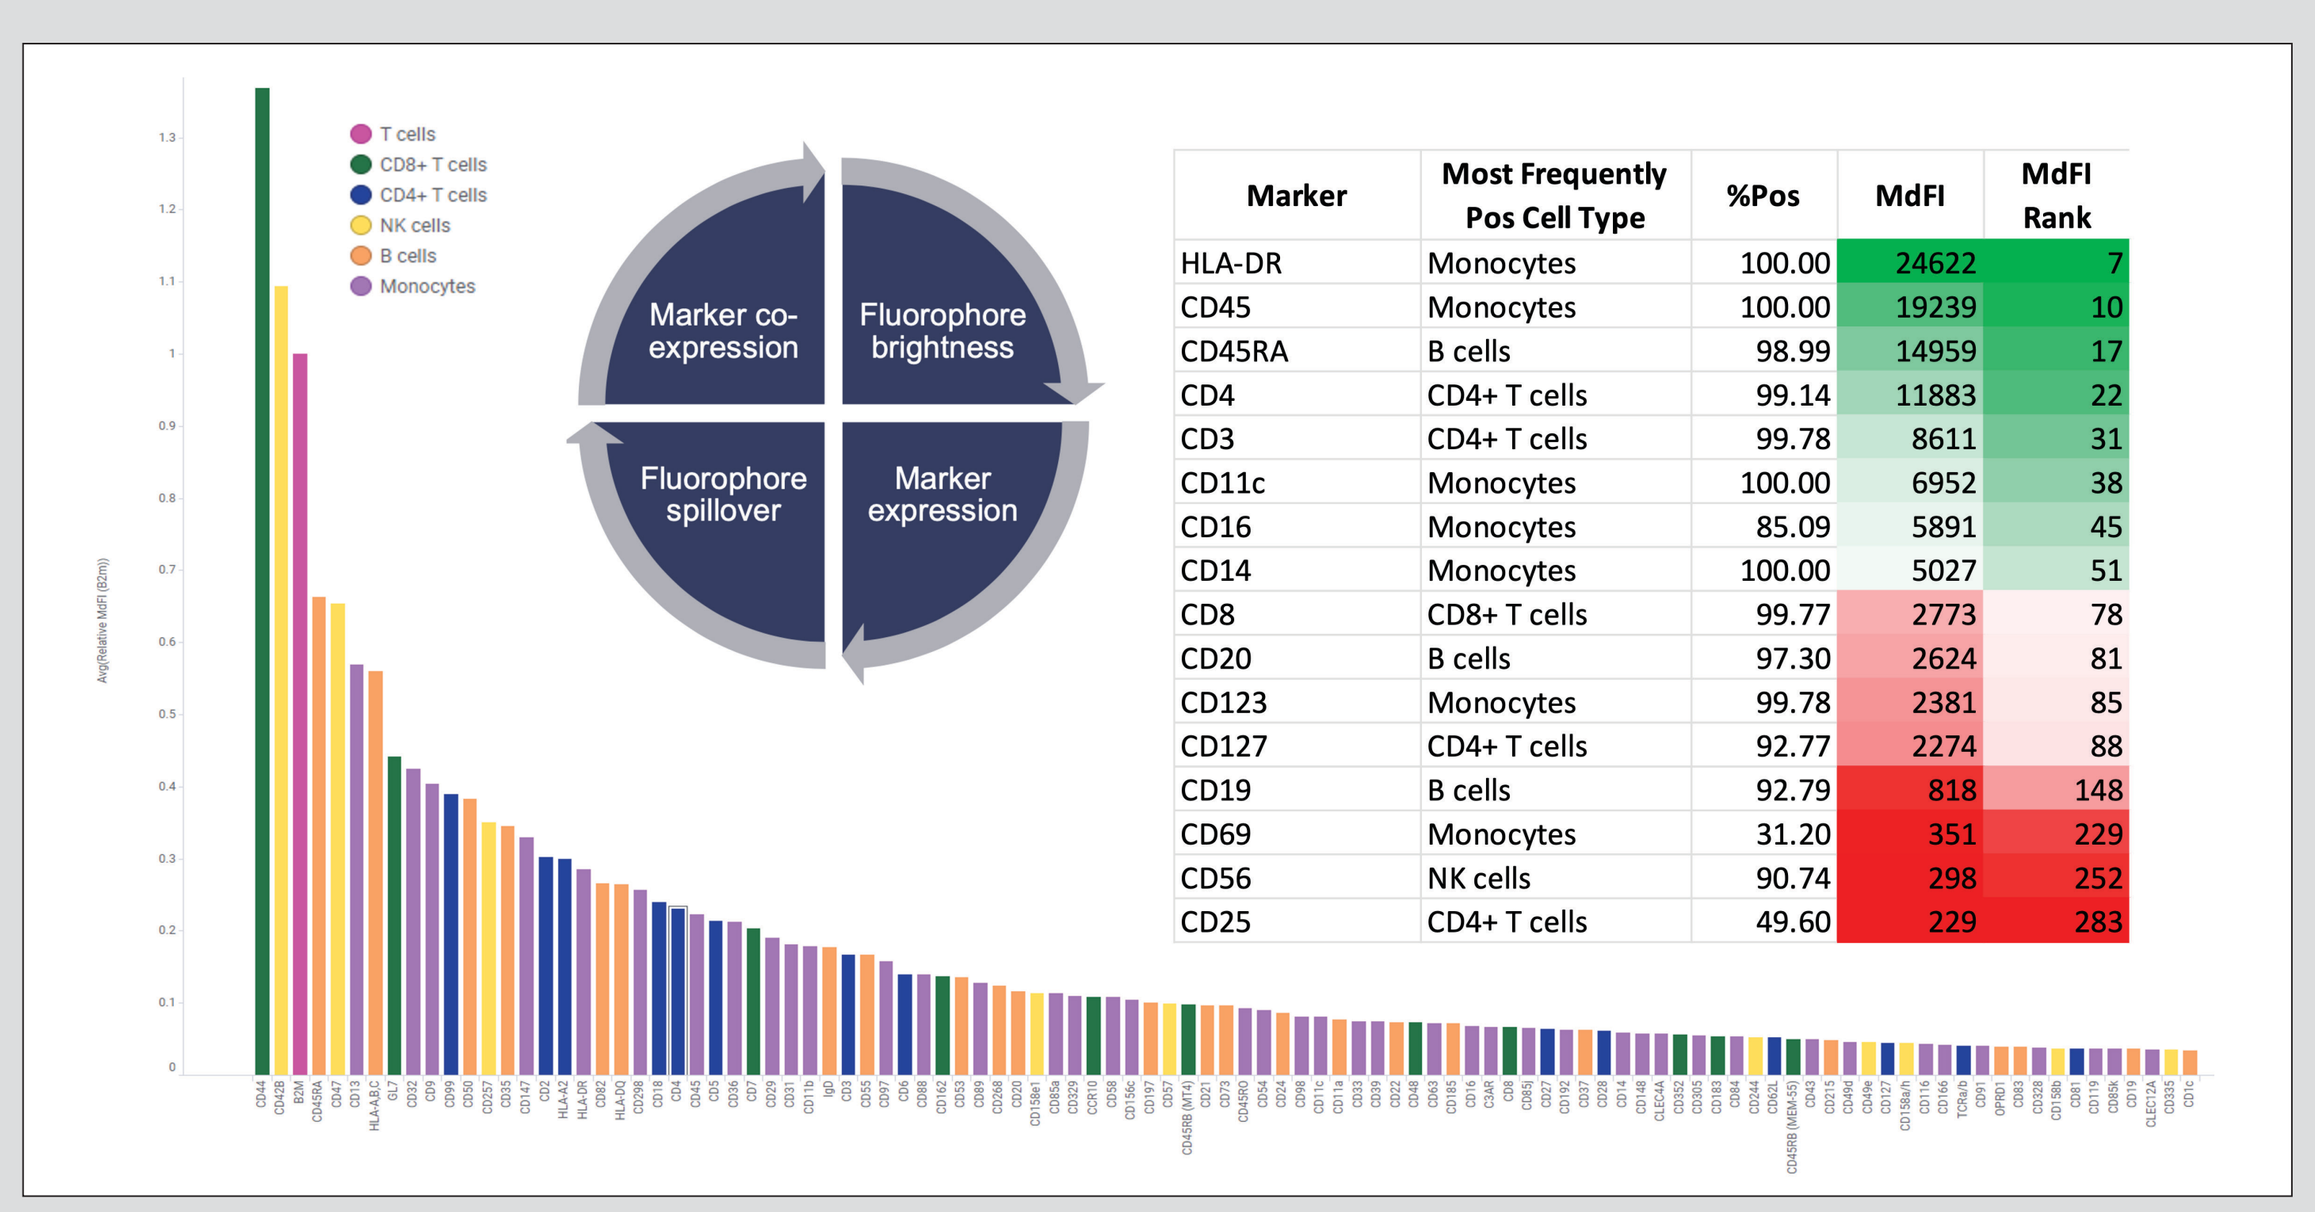
T cells
CD8+ T cells
CD4+ T cells
NK cells
B cells
Monocytes
Marker co-expression
Fluorophore brightness
Fluorophore spillover
Marker expression

Marker	Most Frequently Pos Cell Type	%Pos	MdFI	MdFI Rank
HLA-DR	Monocytes	100.00	24622	7
CD45	Monocytes	100.00	19239	10
CD45RA	B cells	98.99	14959	17
CD4	CD4+ T cells	99.14	11883	22
CD3	CD4+ T cells	99.78	8611	31
CD11c	Monocytes	100.00	6952	38
CD16	Monocytes	85.09	5891	45
CD14	Monocytes	100.00	5027	51
CD8	CD8+ T cells	99.77	2773	78
CD20	B cells	97.30	2624	81
CD123	Monocytes	99.78	2381	85
CD127	CD4+ T cells	92.77	2274	88
CD19	B cells	92.79	818	148
CD69	Monocytes	31.20	351	229
CD56	NK cells	90.74	298	252
CD25	CD4+ T cells	49.60	229	283

Figure 1: Surface Marker Profiling. Internal data base of relative expression of surface markers. Markers are stained with PE-labelled antibodies in a minimal cocktail to identify main cell populations.

Assessment of Panel SSM

Theoretical spillover is estimated during panel design and confirmed during full panel testing. Unsatisfactory unmixing, spillover, or poor population resolution leads to fluorophore/marker shuffling or changes.

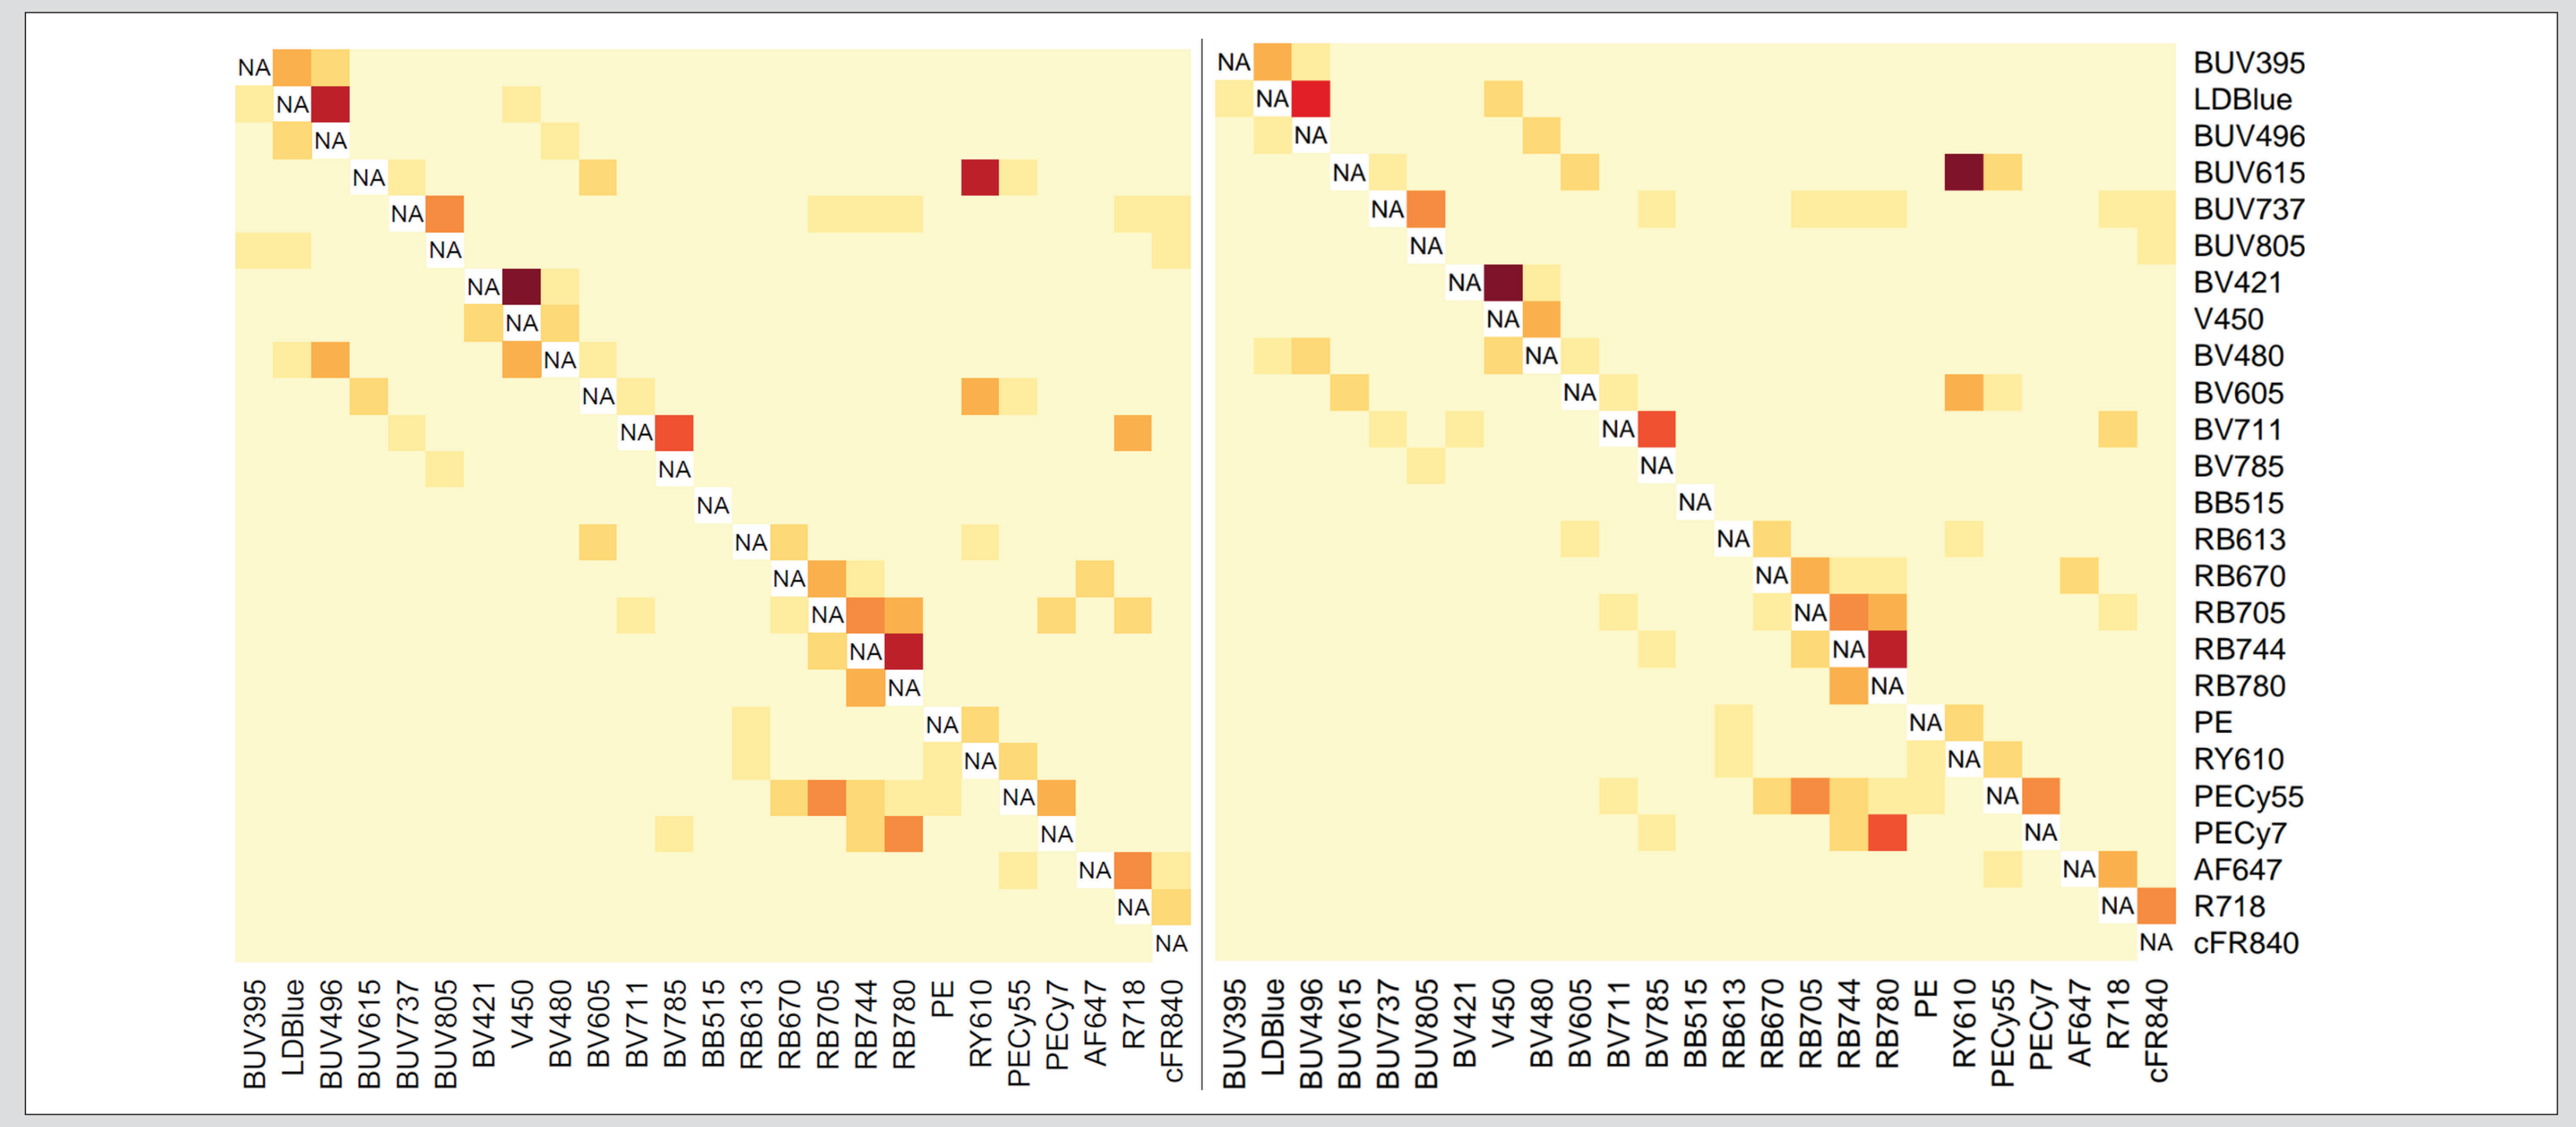
BUV395
LDBlue
BUV496
BUV615
BUV737
BUV805
BV421
V450
BV480
BV605
BV711
BV785
BB515
RB613
RB670
RB705
RB744
RB780
PE
RY610
PECy55
PECy7
AF647
R718
cFR840

Figure 2: Theoretical and Actual Spillover Spread Matrix (SSM). SSM generated using single stained PBMC stained with anti-CD4 antibodies (left) compared to SSM generated using the single stained references (beads and PBMC) used for the unmixing of the panel.

Panel Design

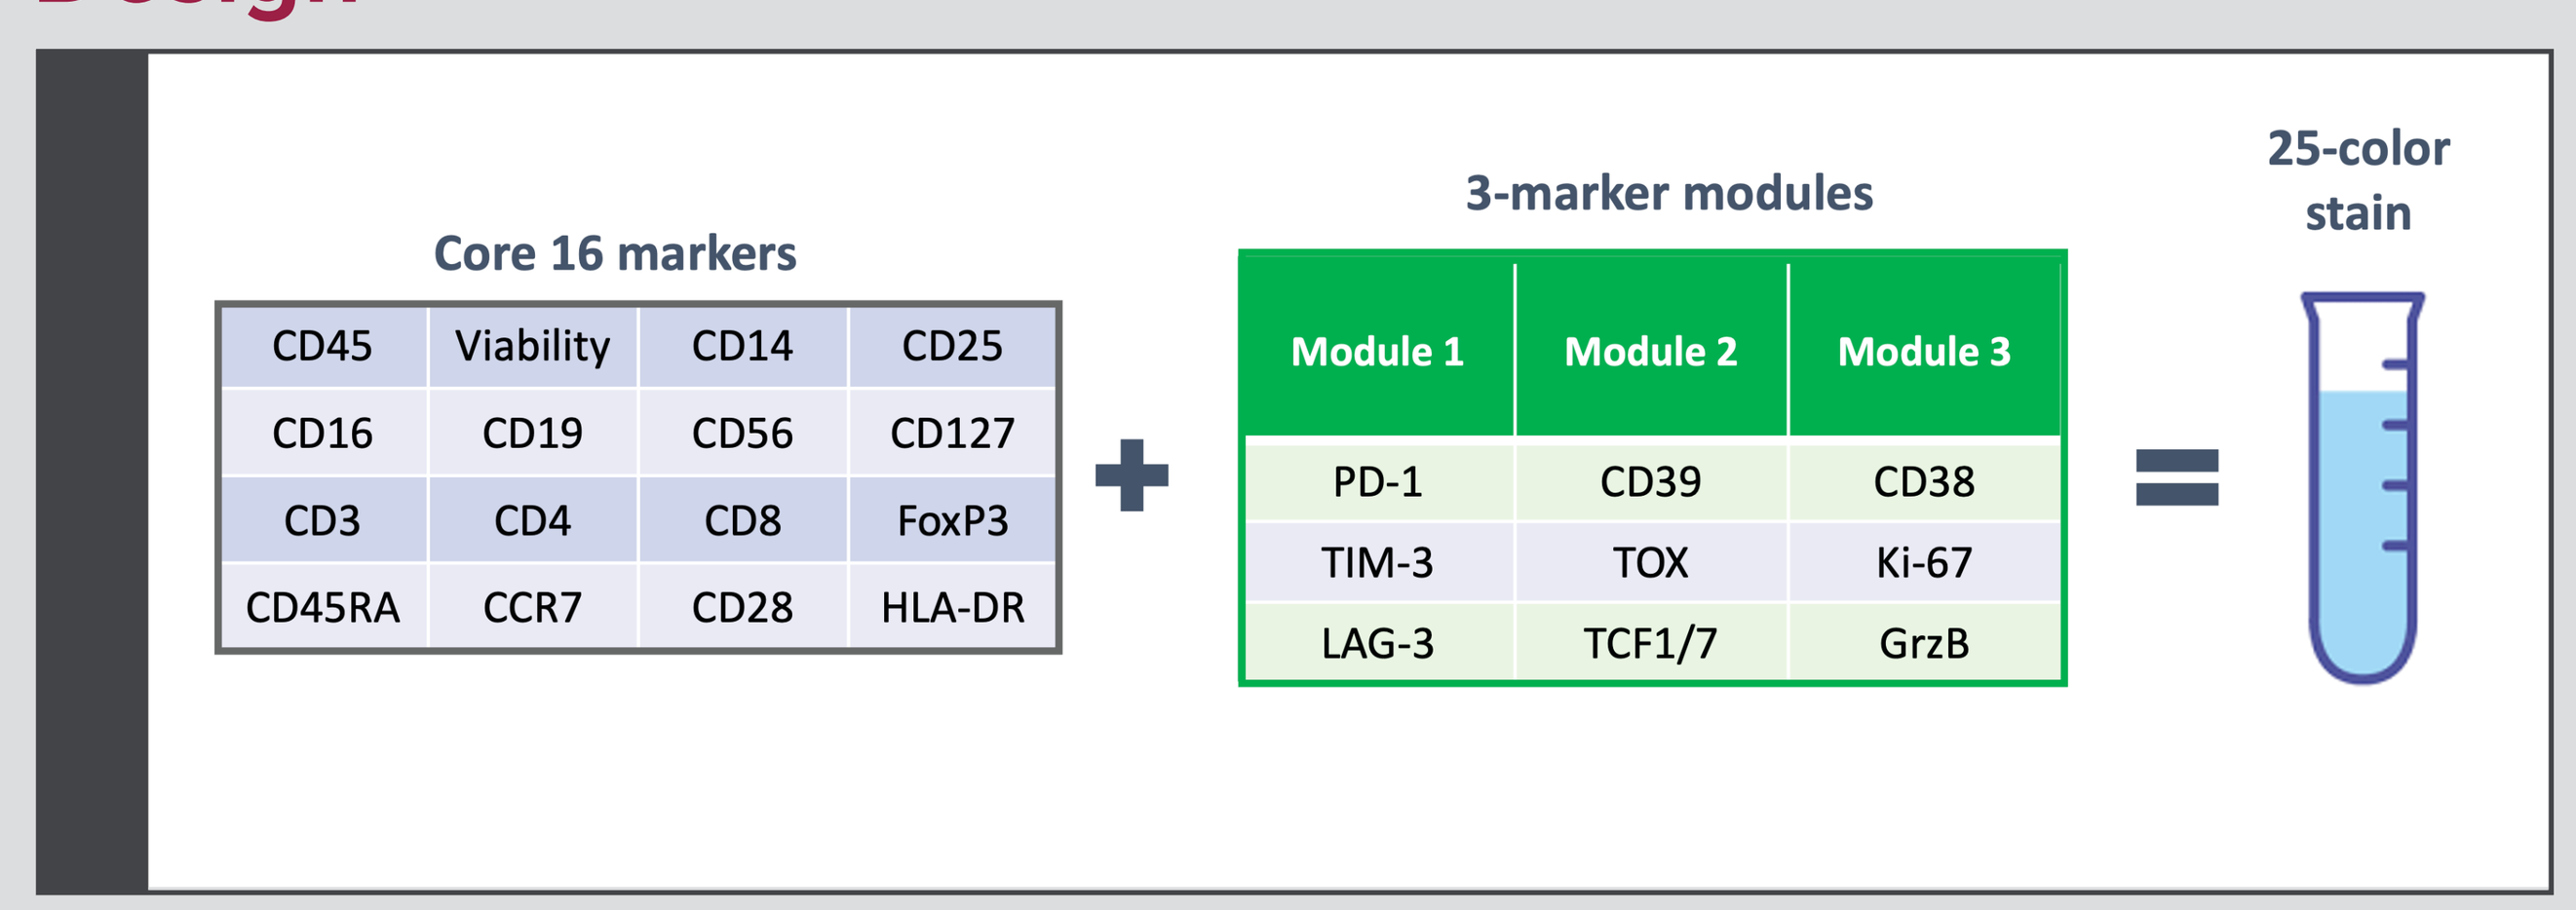
Core 16 markers
CD45 Viability CD14 CD25
CD16 CD19 CD56 CD127
CD3 CD4 CD8 FoxP3
CD45RA CCR7 CD28 HLA-DR
3-marker modules
Module 1: PD-1, TIM-3, LAG-3
Module 2: CD39, TOX, TCF1/7
Module 3: CD38, Ki-67, GrzB
25-color stain

Figure 3: Final Panel Design. 25-marker panel with 3 modules of 3 markers each. The modules have pre-defined fluorophores chosen to minimizes the impact of changing the markers of a module on the other modules or the core.

Modularity Proof of Concept

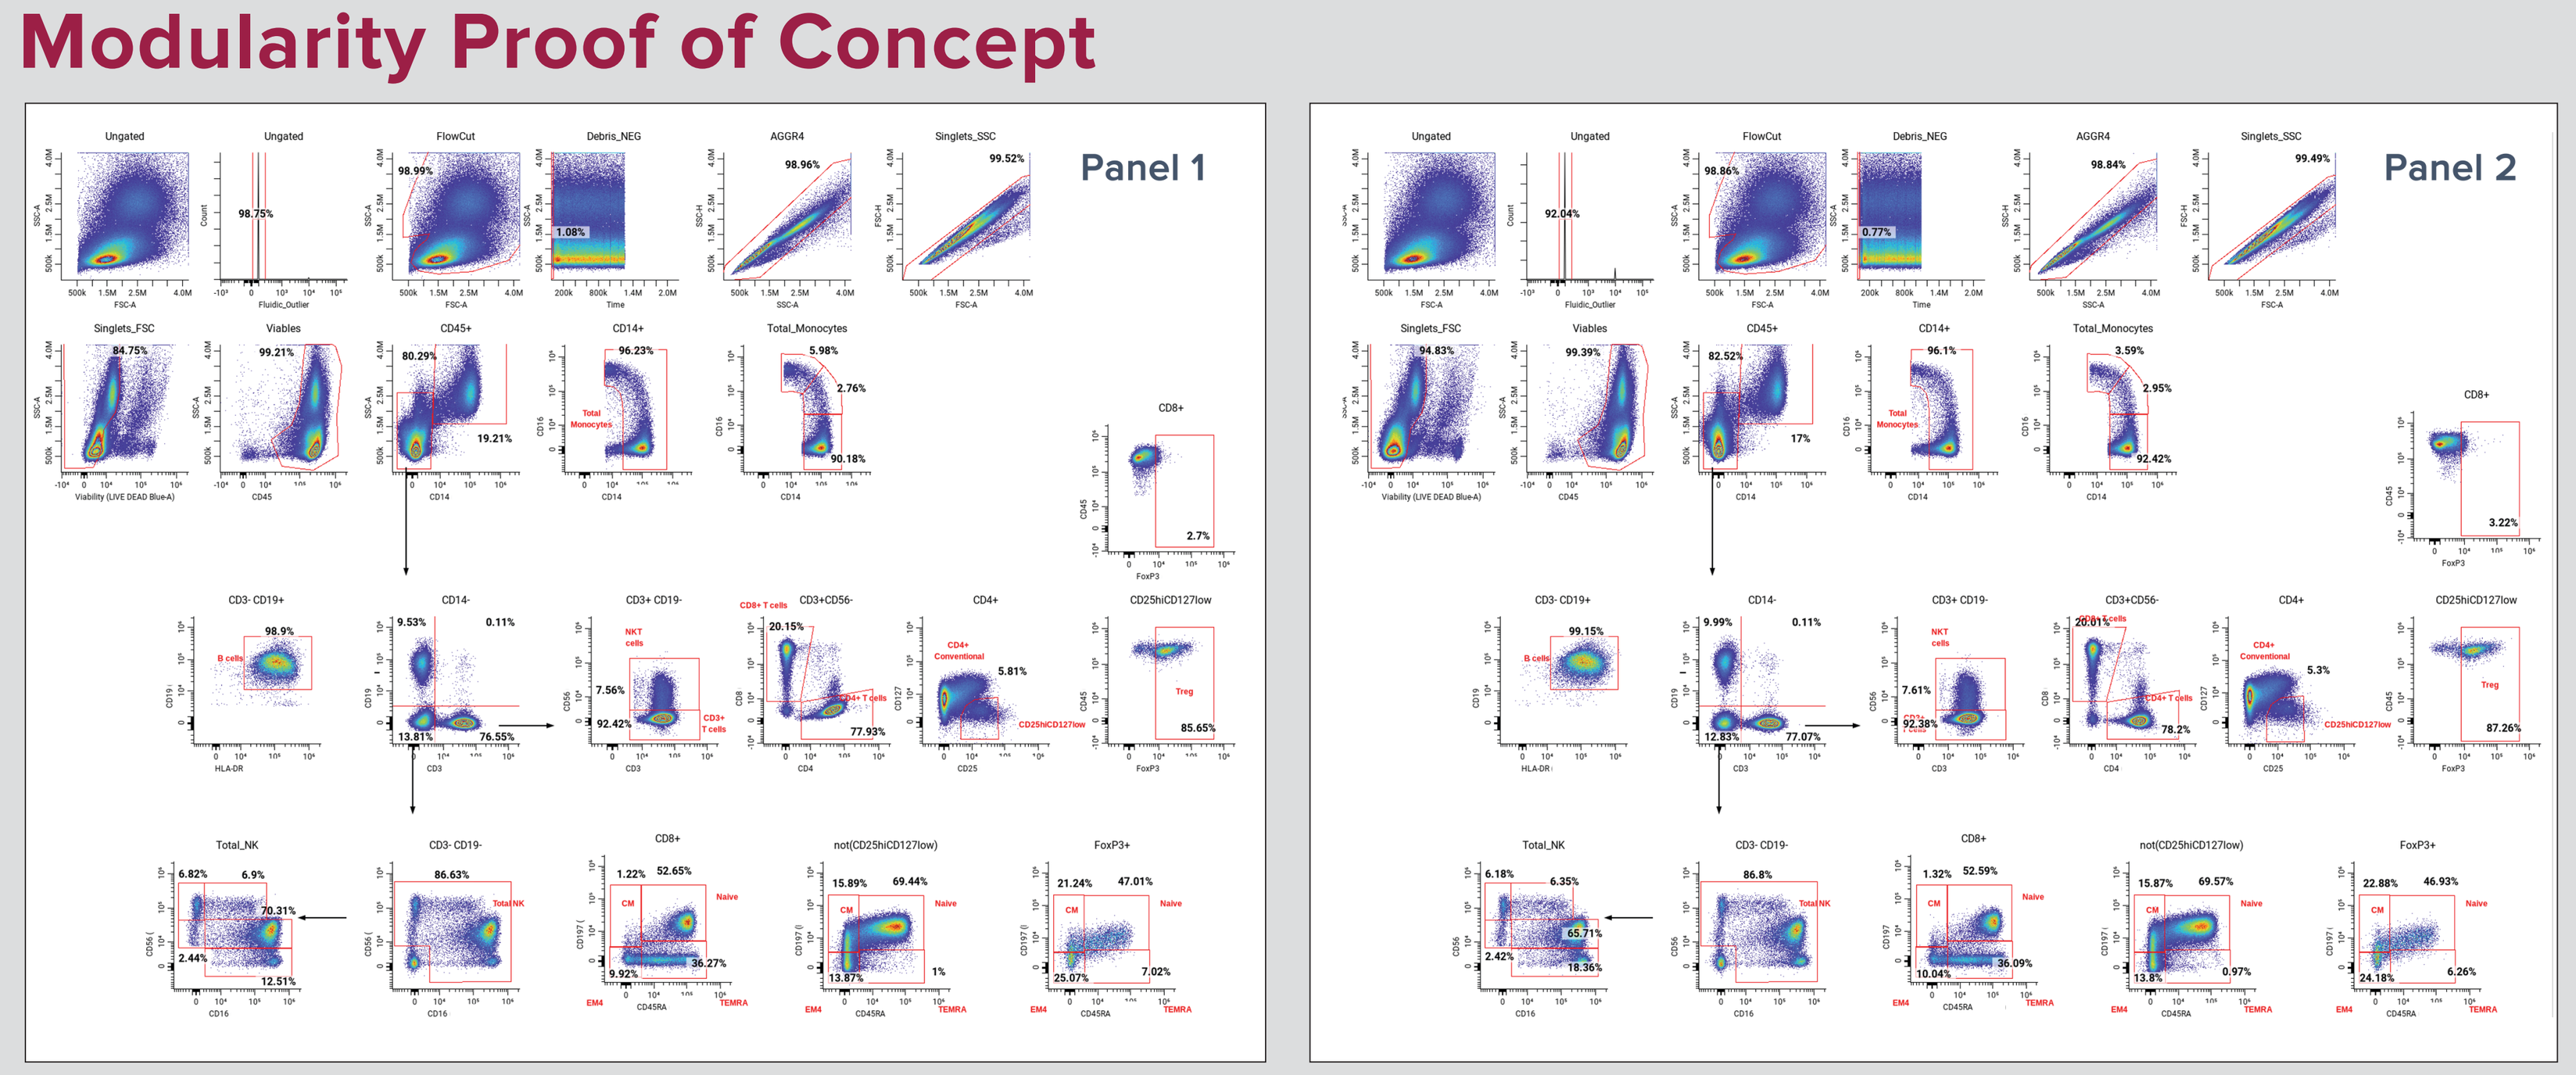
Panel 1
Panel 2

Figure 4: Hierarchical gating of modular panels. Comparison of hierarchical gating between two panels with a swapped module showing minimal changes between panels.

Modularity Proof of Concept

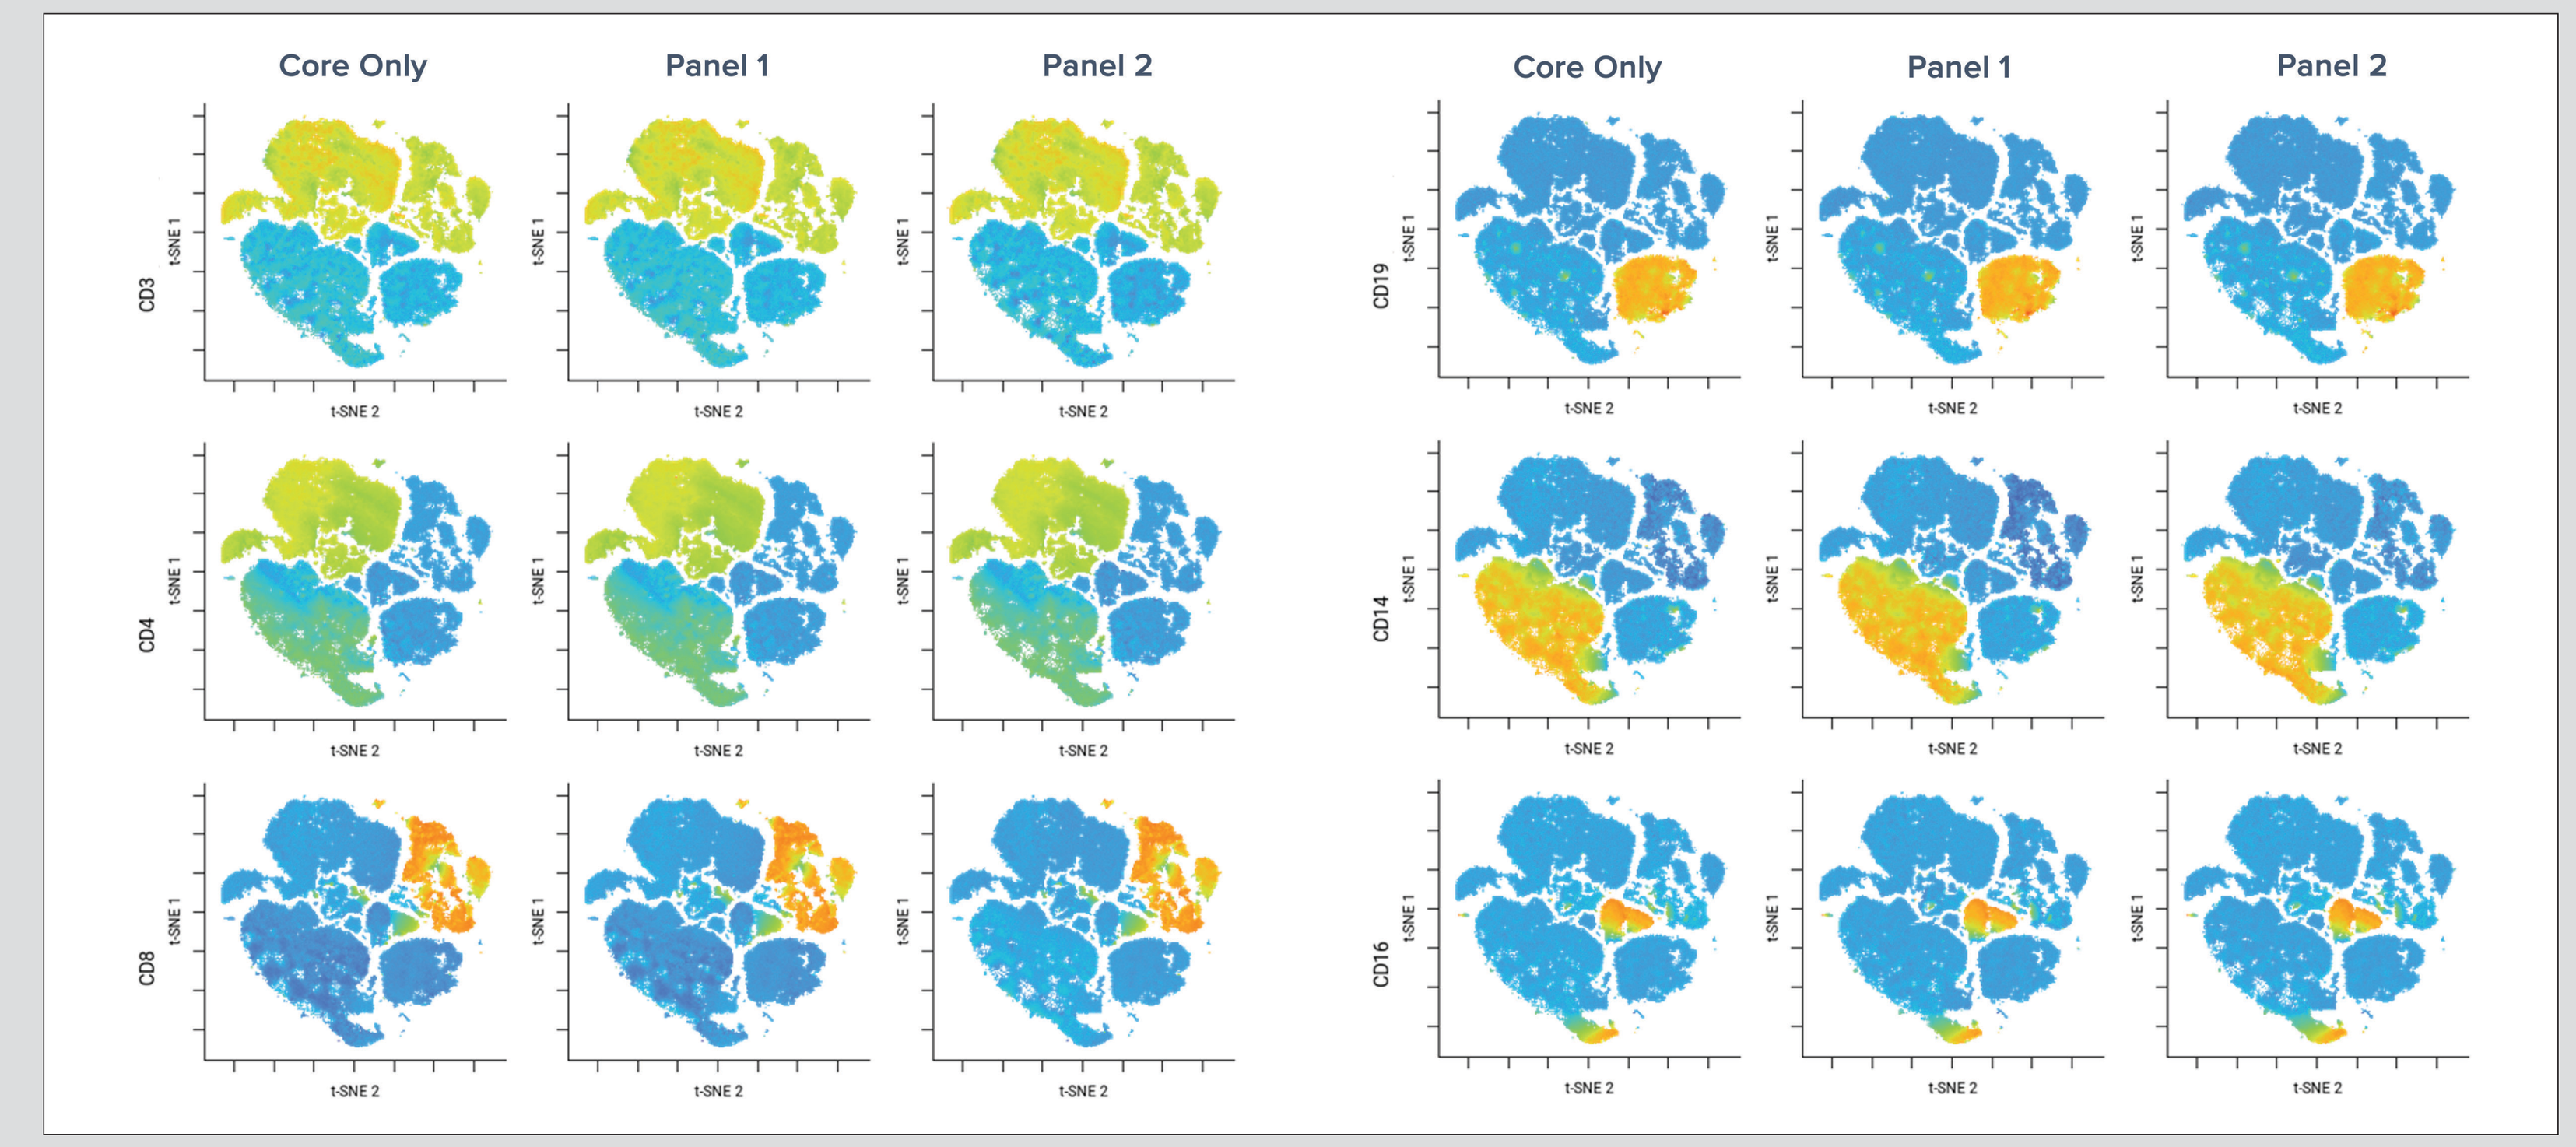
Core Only
Panel 1
Panel 2
CD3
CD4
CD8
CD19
CD14
CD16
t-SNE 1
t-SNE 2

Figure 5: Relative Expression of Markers Using t-SNE Clustering. Comparison between the core and two panels with a marker-swapped module using CellEngine unsupervised clustering highlighting standard cell populations markers.

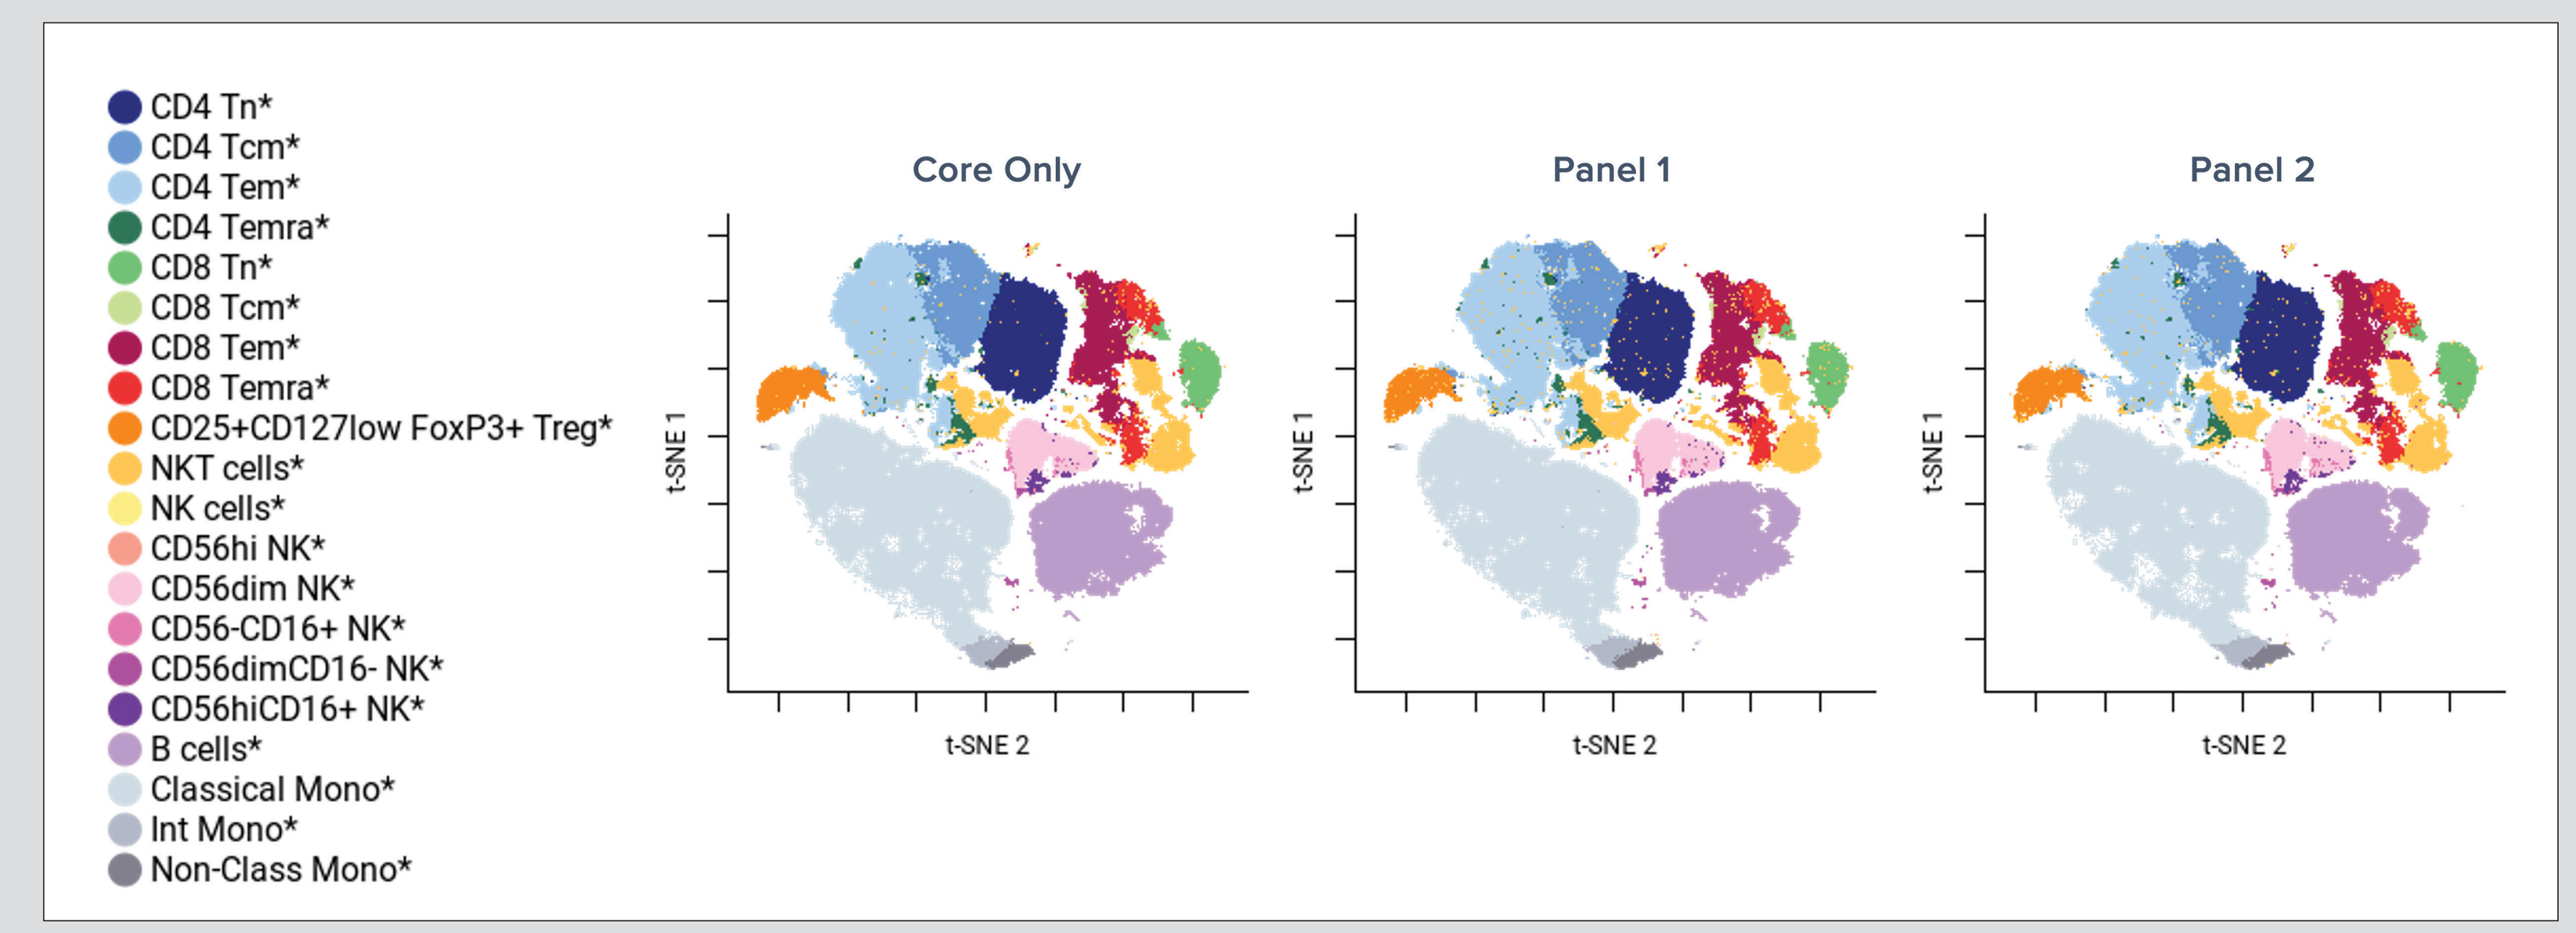
CD4 Tn*
CD4 Tcm*
CD4 Tem*
CD4 Temra*
CD8 Tn*
CD8 Tcm*
CD8 Tem*
CD8 Temra*
CD25+CD127low FoxP3+ Treg*
NKT cells*
NK cells*
CD56hi NK*
CD56dim NK*
CD56-CD16+ NK*
CD56dimCD16- NK*
CD56hiCD16+ NK*
B cells*
Classical Mono*
Int Mono*
Non-Class Mono*
Core Only
Panel 1
Panel 2
t-SNE 1
t-SNE 2

Figure 6: Cell Populations Distribution Using t-SNE Clustering. Comparison between the core and two panels with a marker-swapped module using CellEngine unsupervised clustering highlighting the different populations identifiable by the panel.

CONCLUSION

Overall, this work provides a **flexible** spectral cytometry panel design which reduces the need for repeated optimization and enables **rapid adaptation** to the diverse needs of different clinical programs.

The minimal changes across panel configurations supports **modularity** and likely provides a robust core usable across different matrices. Careful assignments of new markers to the existing fluorophore module remains an aspect to consider, especially in the case of extremely high or low expressed markers.

Send inquiries to *isales@cellcarta.com* with CYTO 2026 in email subject.

P112 www.cellcarta.com