Understanding Protein Normalization: A Case Study in PBMC Fit-for-Purpose Validation by MS

Laurence Mayrand-Provencher, Nicholas Stesco, Mirzo Kanoatov, Gwenael Pottiez, Luca Genovesi
CellCarta, Montreal, Quebec, Canada

POSTER NUMBER: DAY2-P21

CellCarta

INTRODUCTION

- Traditional protein-based analysis methods, such as Western blotting, often struggle to distinguish true biological changes from sample loading variability. In contrast, mass-spectrometry-based proteomics provides multiplexed measurements, enabling the relative and absolute quantification of thousands of proteins across complex biological systems. This broad coverage not only enhances biological interpretation but also allows the identification of stably expressed proteins directly from the dataset[1].
- When combined with a direct digestion approach, MS thus offers an advantage over antibody-based assays like LBA by revealing endogenous proteins that can serve as more reliable loading controls than conventional strategies such as BCA assays, especially in samples prone to red blood cell contamination.
- PARK-7[2] is a highly conserved multifunctional enzyme found across species, from prokaryotes to eukaryotes. Its enzymatic and non-enzymatic functions (e.g., anti-oxidation, anti-glycation, protein quality control), along with its role as a transcriptional coactivator, support its involvement in key cellular processes such as epigenetic regulation, making it a promising therapeutic target for diseases including cancer and Parkinson's disease[3].
- Bromodomain-containing protein 4 (BRD4), a key transcriptional coactivator, drives cancer and inflammation through both bromodomain-dependent and independent mechanisms. Resistance to bromodomain inhibitors has prompted alternative strategies, including BRD4-targeted degraders, non-bromodomain ligands that disrupt scaffolding functions, PTM disruptors, and condensate modulators[4].
- Here, we present a case study of fit-for-purpose validation (FFPV) for BRD4 quantification in PBMCs using PARK-7 as a normalizing protein. Key steps—including PBMC collection, lysis, homogenization, and digestion—were assessed as potential sources of variability. This work highlights the need to tailor sample preparation to both the analyte and normalizer to ensure robust and reproducible performance.

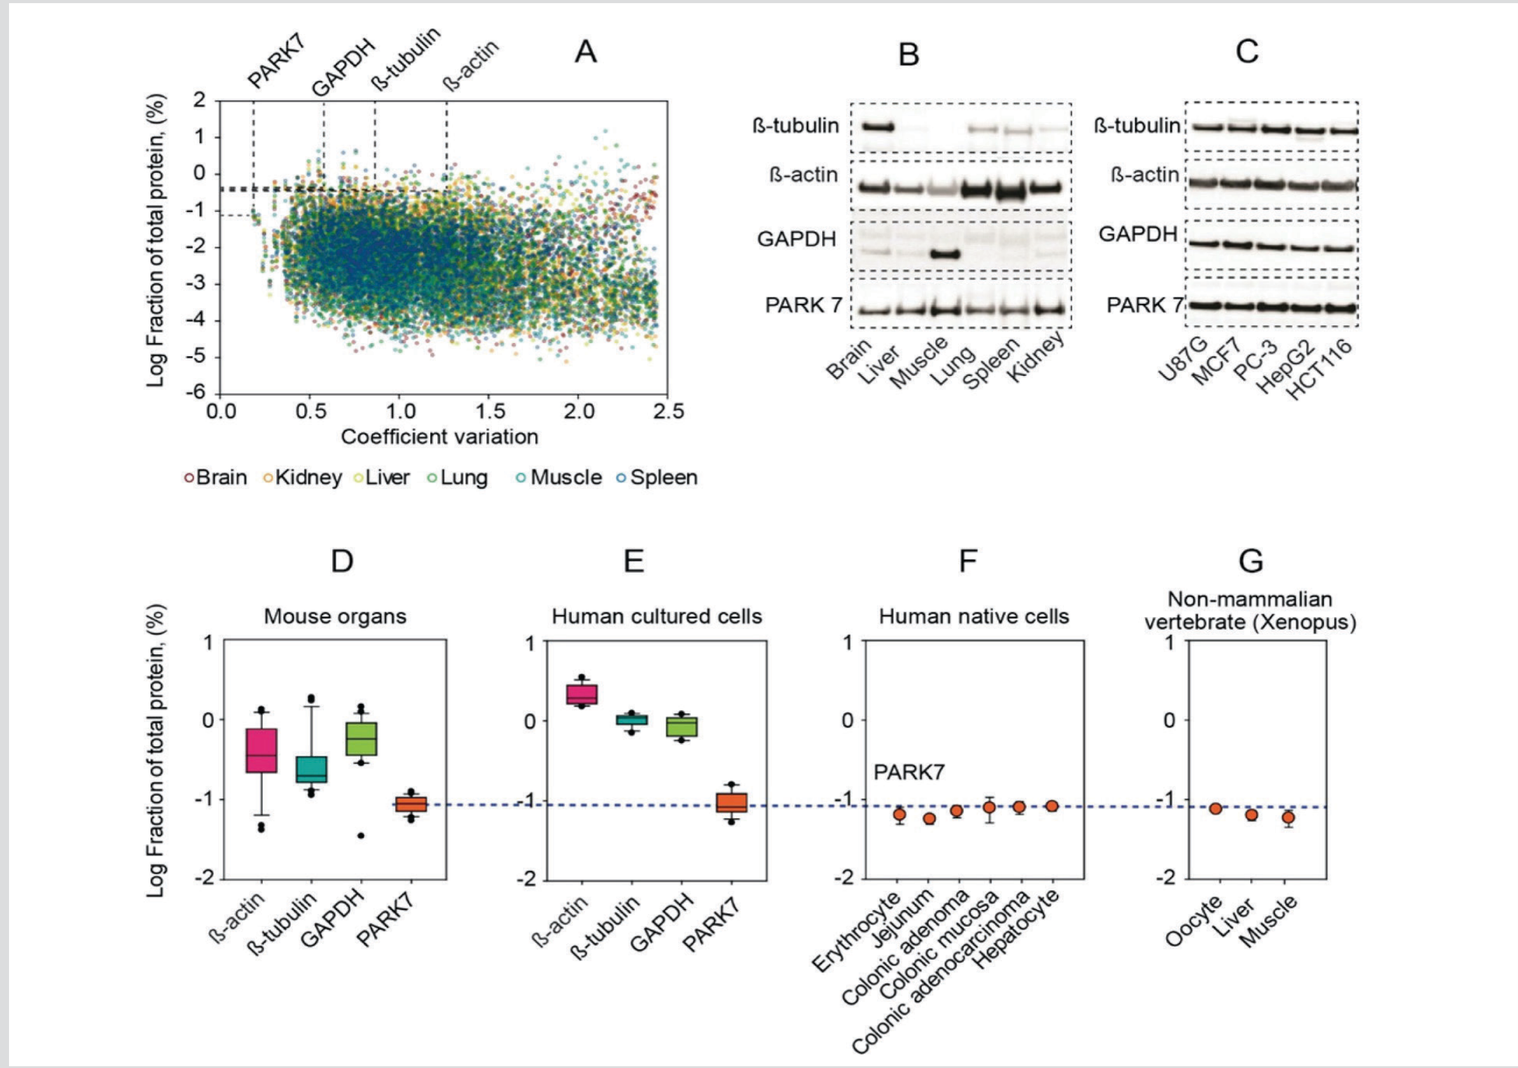
A
B
C
D
E
F
G
Log Fraction of total protein, (%)
Coefficient variation
Brain Kidney Liver Lung Muscle Spleen
ß-tubulin
ß-actin
GAPDH
PARK 7
Mouse organs
Human cultured cells
Human native cells
Non-mammalian vertebrate (Xenopus)

Figure 1: PARK-7 is a ubiquitous protein with constant expression in vertebrate cells.

Sample Preparation and Processing

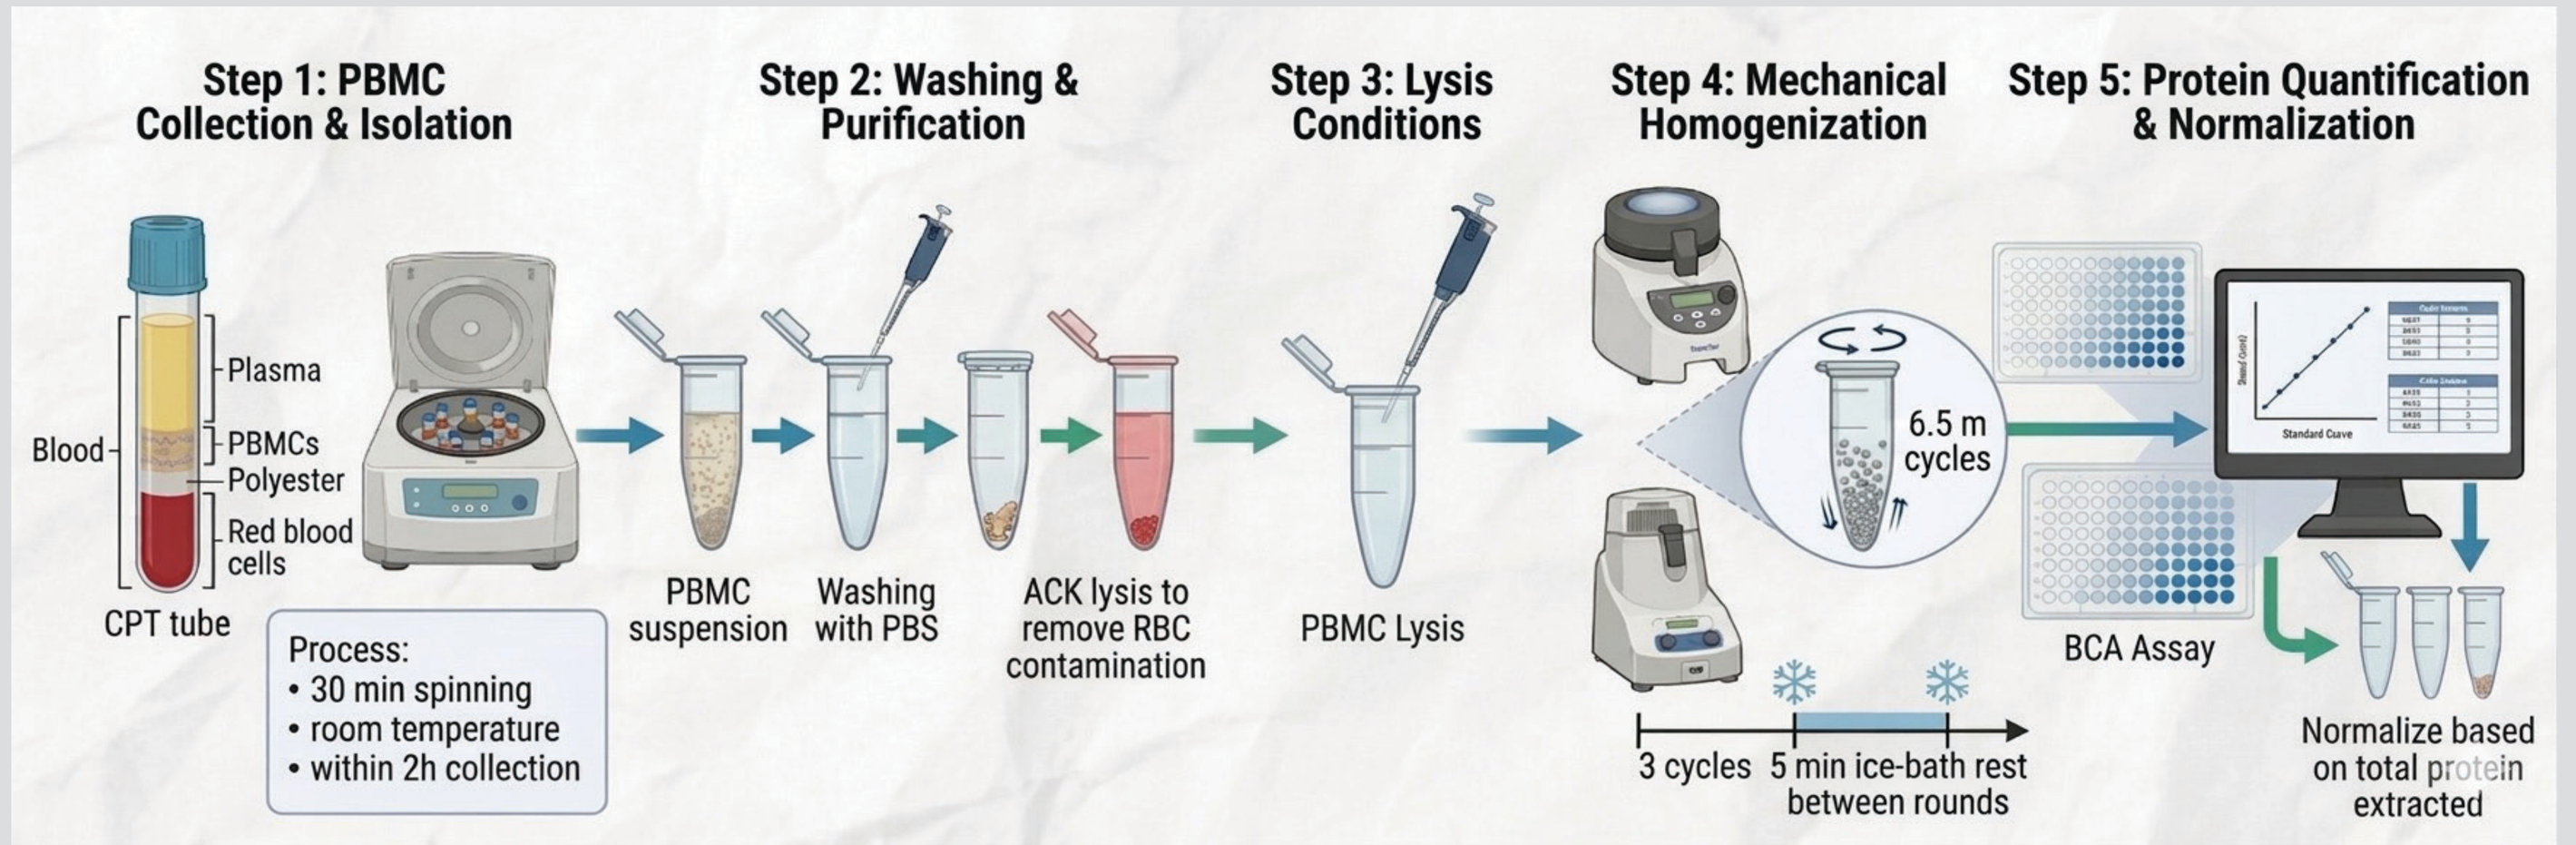
Step 1: PBMC Collection & Isolation
Step 2: Washing & Purification
Step 3: Lysis Conditions
Step 4: Mechanical Homogenization
Step 5: Protein Quantification & Normalization
Blood
Plasma
PBMCs
Polyester
Red blood cells
CPT tube
Process:
• 30 min spinning
• room temperature
• within 2h collection
PBMC suspension
Washing with PBS
ACK lysis to remove RBC contamination
PBMC Lysis
6.5 m cycles
3 cycles
5 min ice-bath rest between rounds
BCA Assay
Normalize based on total protein extracted

Figure 2: Sample Preparation from PBMC Collection to BCA total protein quantitation.

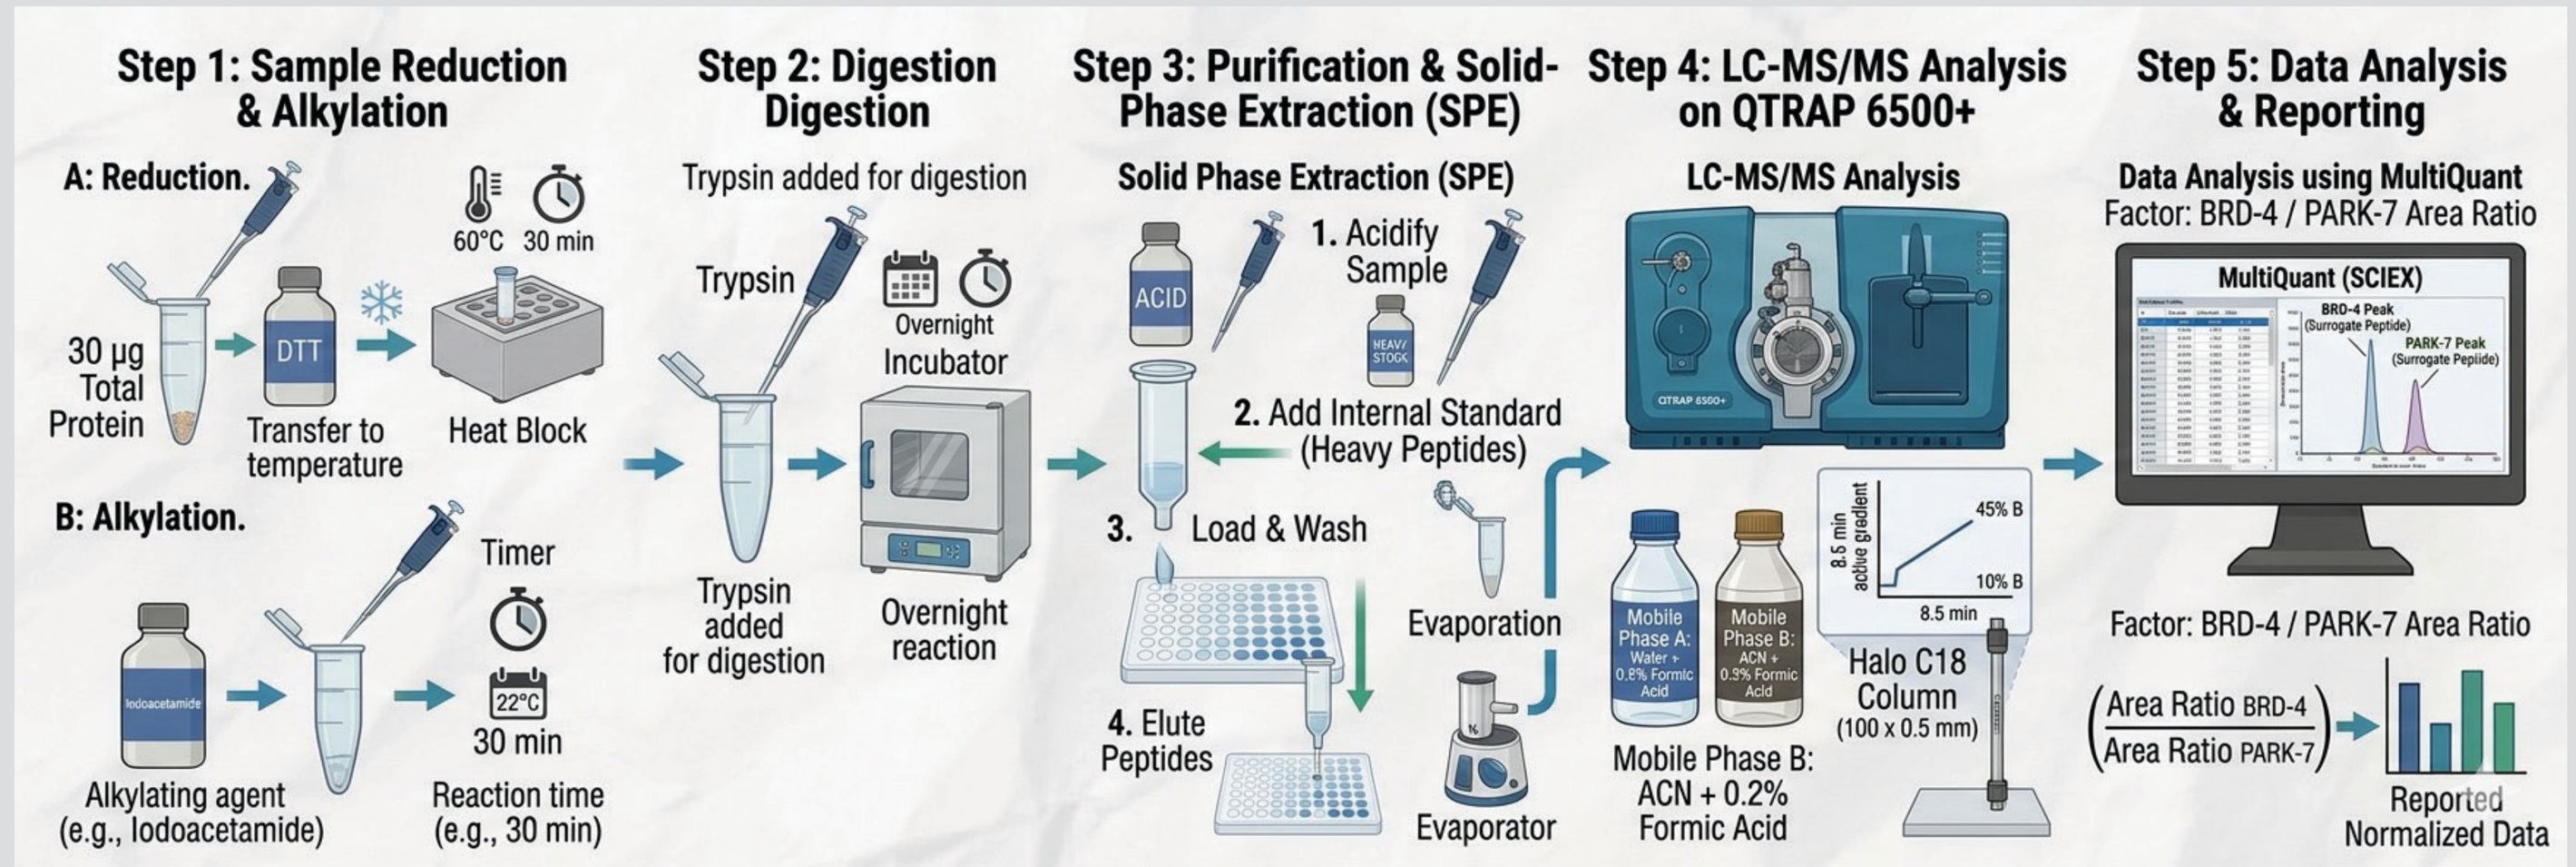
Step 1: Sample Reduction & Alkylation
Step 2: Digestion Digestion
Step 3: Purification & Solid-Phase Extraction (SPE)
Step 4: LC-MS/MS Analysis on QTRAP 6500+
Step 5: Data Analysis & Reporting
A: Reduction.
60°C 30 min
30 µg Total Protein
DTT
Transfer to temperature
Heat Block
B: Alkylation.
Timer
22°C
30 min
Alkylating agent (e.g., Iodoacetamide)
Reaction time (e.g., 30 min)
Trypsin added for digestion
Trypsin
Overnight Incubator
Trypsin added for digestion
Overnight reaction
Solid Phase Extraction (SPE)
1. Acidify Sample
2. Add Internal Standard (Heavy Peptides)
3. Load & Wash
4. Elute Peptides
Evaporation
Evaporator
LC-MS/MS Analysis
Mobile Phase B: ACN + 0.2% Formic Acid
Halo C18 Column (100 x 0.5 mm)
8.5 min
Data Analysis using MultiQuant
Factor: BRD-4 / PARK-7 Area Ratio
MultiQuant (SCIEX)
Factor: BRD-4 / PARK-7 Area Ratio
Area Ratio BRD-4 / Area Ratio PARK-7
Reported Normalized Data

Figure 3: Sample Processing and LC/MS analysis.

Assay Performance

Table 1: Linearity Testing for PARK-7.

PARK7	Reversed Curve STD Calculated Concentrations (fmol/mL)								
	STD 1	STD 2	STD 3	STD 4	STD 5	STD 6	STD 7	STD 8	STD 9
Nominal Concentration	31.3	62.5	125.0	250.0	1875.0	3750.0	8750.0	11250.0	12500.0
Calculated Concentration	RC (20.1)	RC (71.1)	121.7	273.0	2019.4	3686.2	9093.4	11306.5	12206.1
	RC (68.8)	RC (74.0)	120.4	260.0	1652.9	3994.4	7690.6	11037.3	13052.1
Mean	-	-	121.05	266.50	1836.15	3840.30	8392.00	11171.90	12629.10
SD	-	-	0.92	9.19	259.15	217.93	991.93	190.35	598.21
%CV	-	-	0.8	3.4	14.1	5.7	11.8	1.7	4.7
%bias	-	-	-3.2	6.6	-2.1	2.4	-4.1	-0.7	1.0

Table 2: Linearity Testing for BRD4.

BRD4	Reversed Curve STD Calculated Concentrations (fmol/mL)								
	STD-1	STD-2	STD-3	STD-4	STD-5	STD-6	STD-7	STD-8	STD-9
Nominal Concentration	31.3	62.5	125.0	250.0	1875.0	3750.0	8750.0	11250.0	12500.0
Calculated Concentration	RC (33.6)	RC (86.6)	128.2	249.7	1909.3	3984.2	8749.9	11461.0	13220.5
	N/D	RC (84.7)	127.4	226.1	1889.9	3742.4	8618.0	10481.8	12150.8
Mean	-	-	127.80	237.90	1899.60	3863.30	8683.95	10971.40	12685.65
SD	-	-	0.57	16.69	13.72	170.98	93.27	692.40	756.39
%CV	-	-	0.4	7.0	0.7	4.4	1.1	6.3	6.0
%bias	-	-	2.2	-4.8	1.3	3.0	-0.8	-2.5	1.5

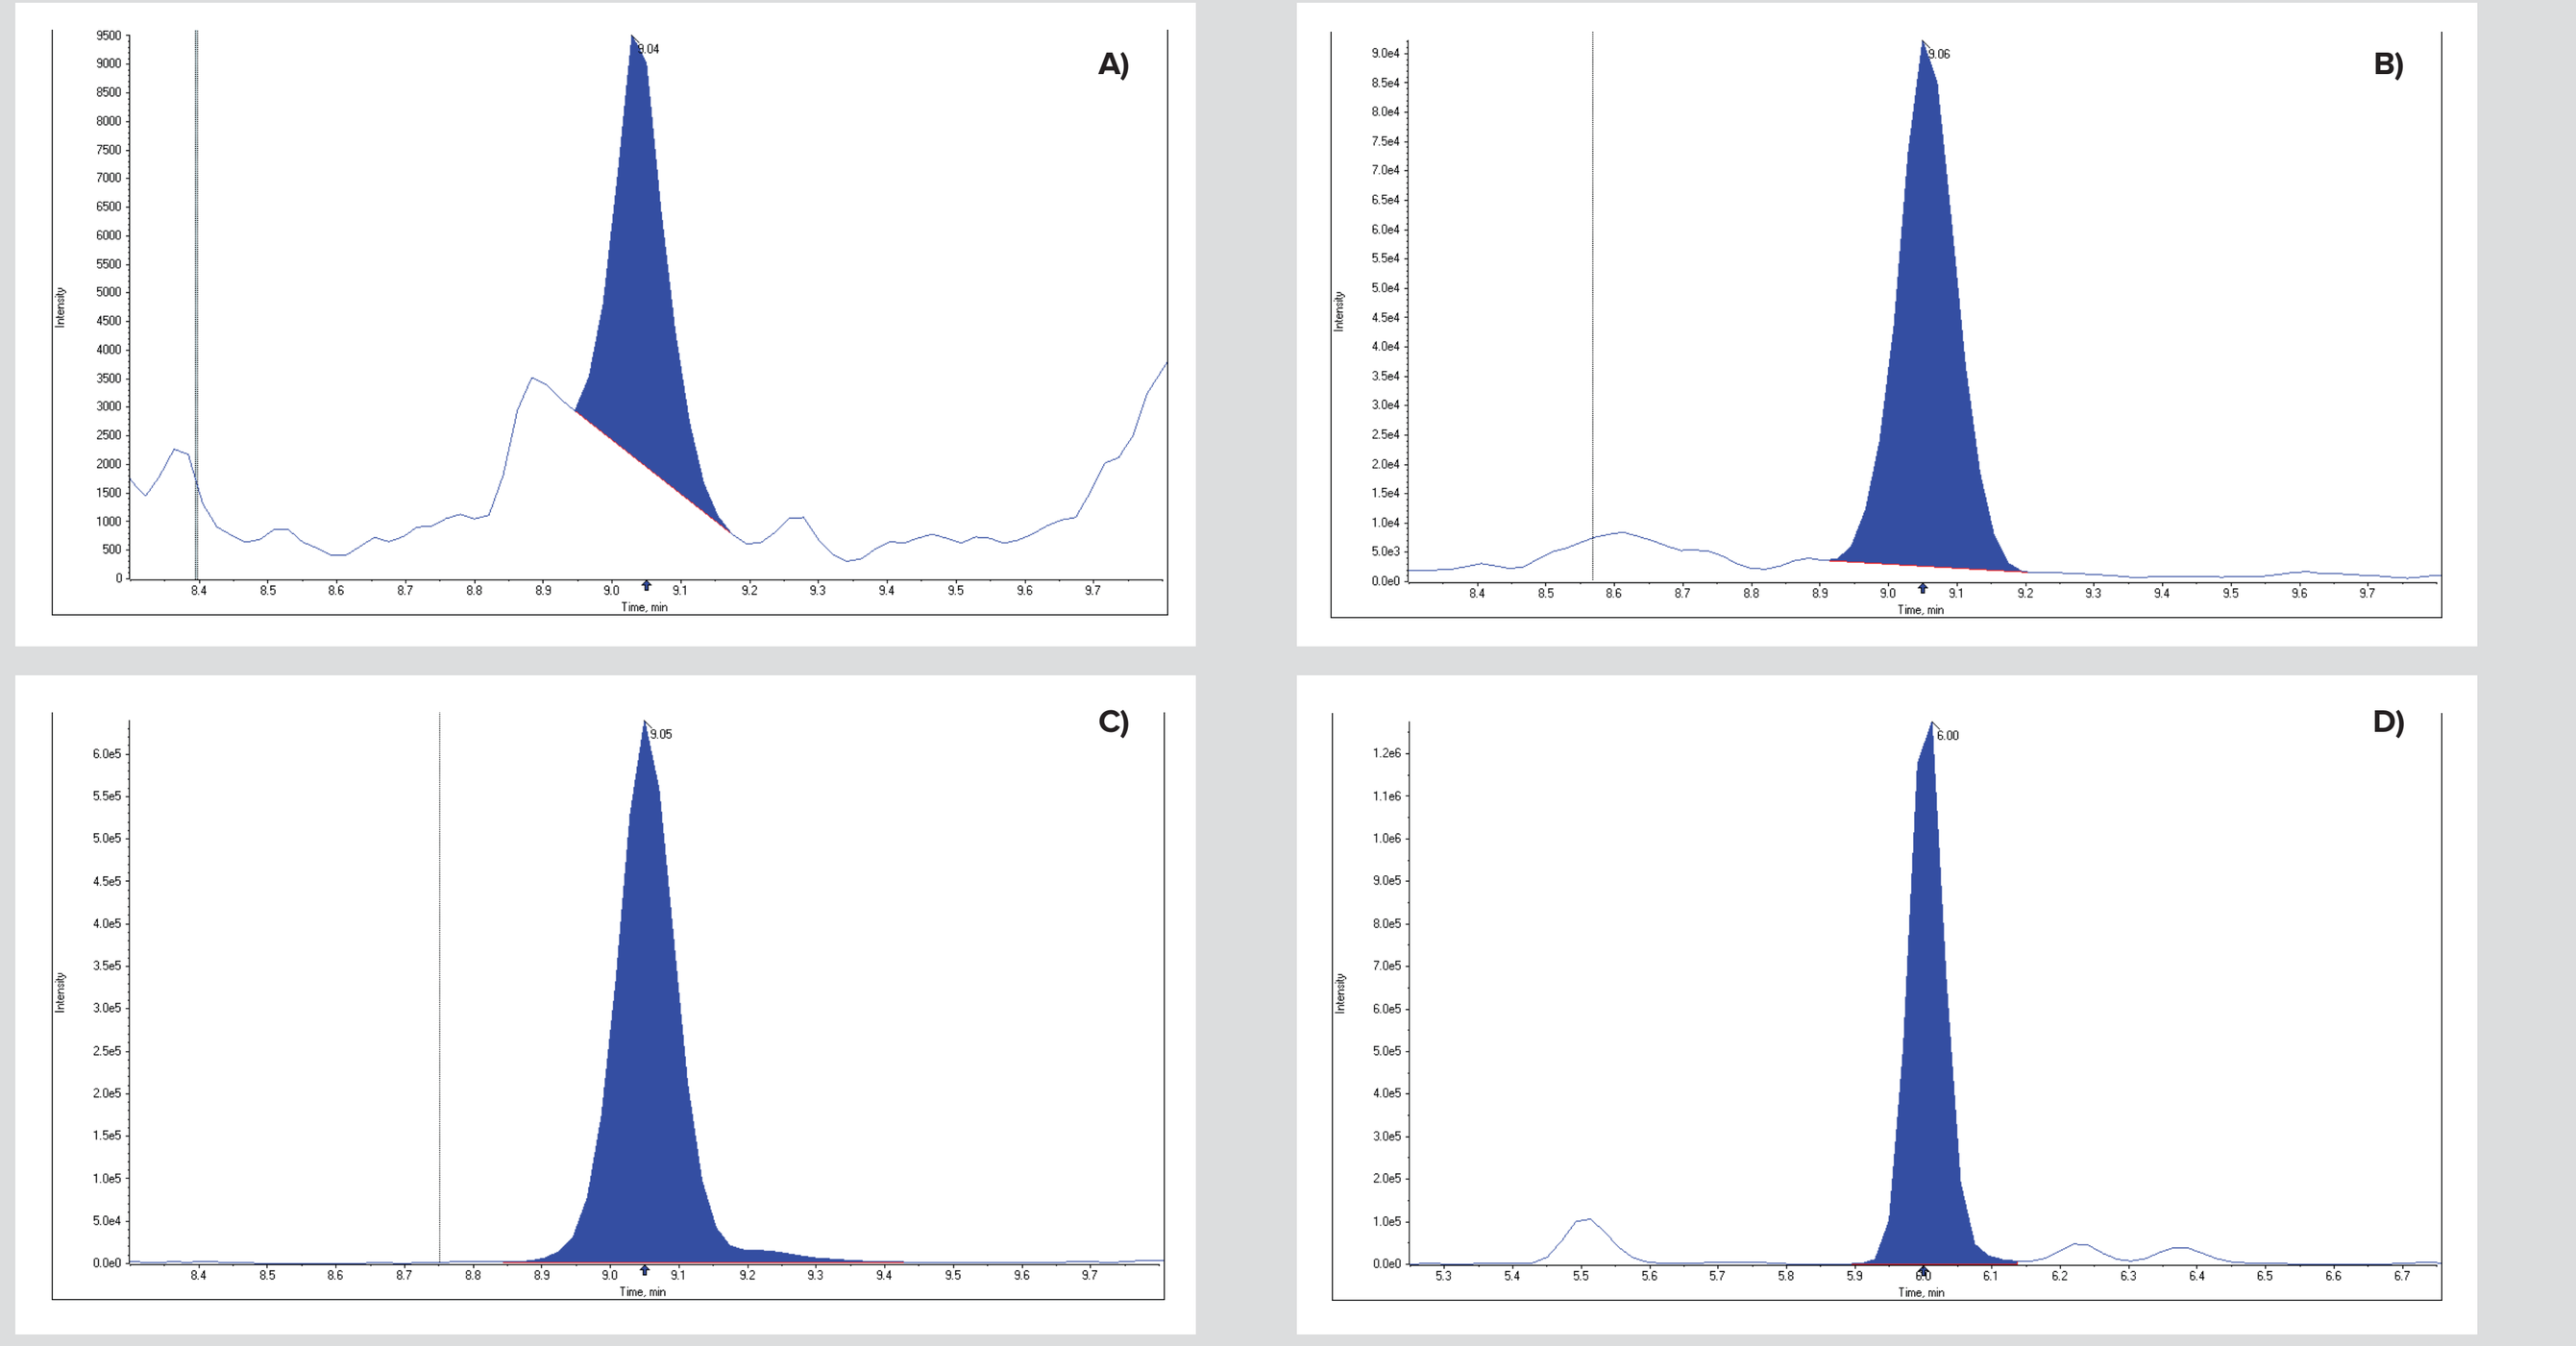
A)
B)
C)
D)
Time, min
Intensity

Figure 4: A) 125fmol/mL for BRD4 (LLOQ); **B)** Endogenous concentration of BRD4 in homogenized PBMC; **C)** 5000fmol/mL for BRD4 (ULOQ); **D)** Endogenous concentration of PARK-7.

Assay Validation Results

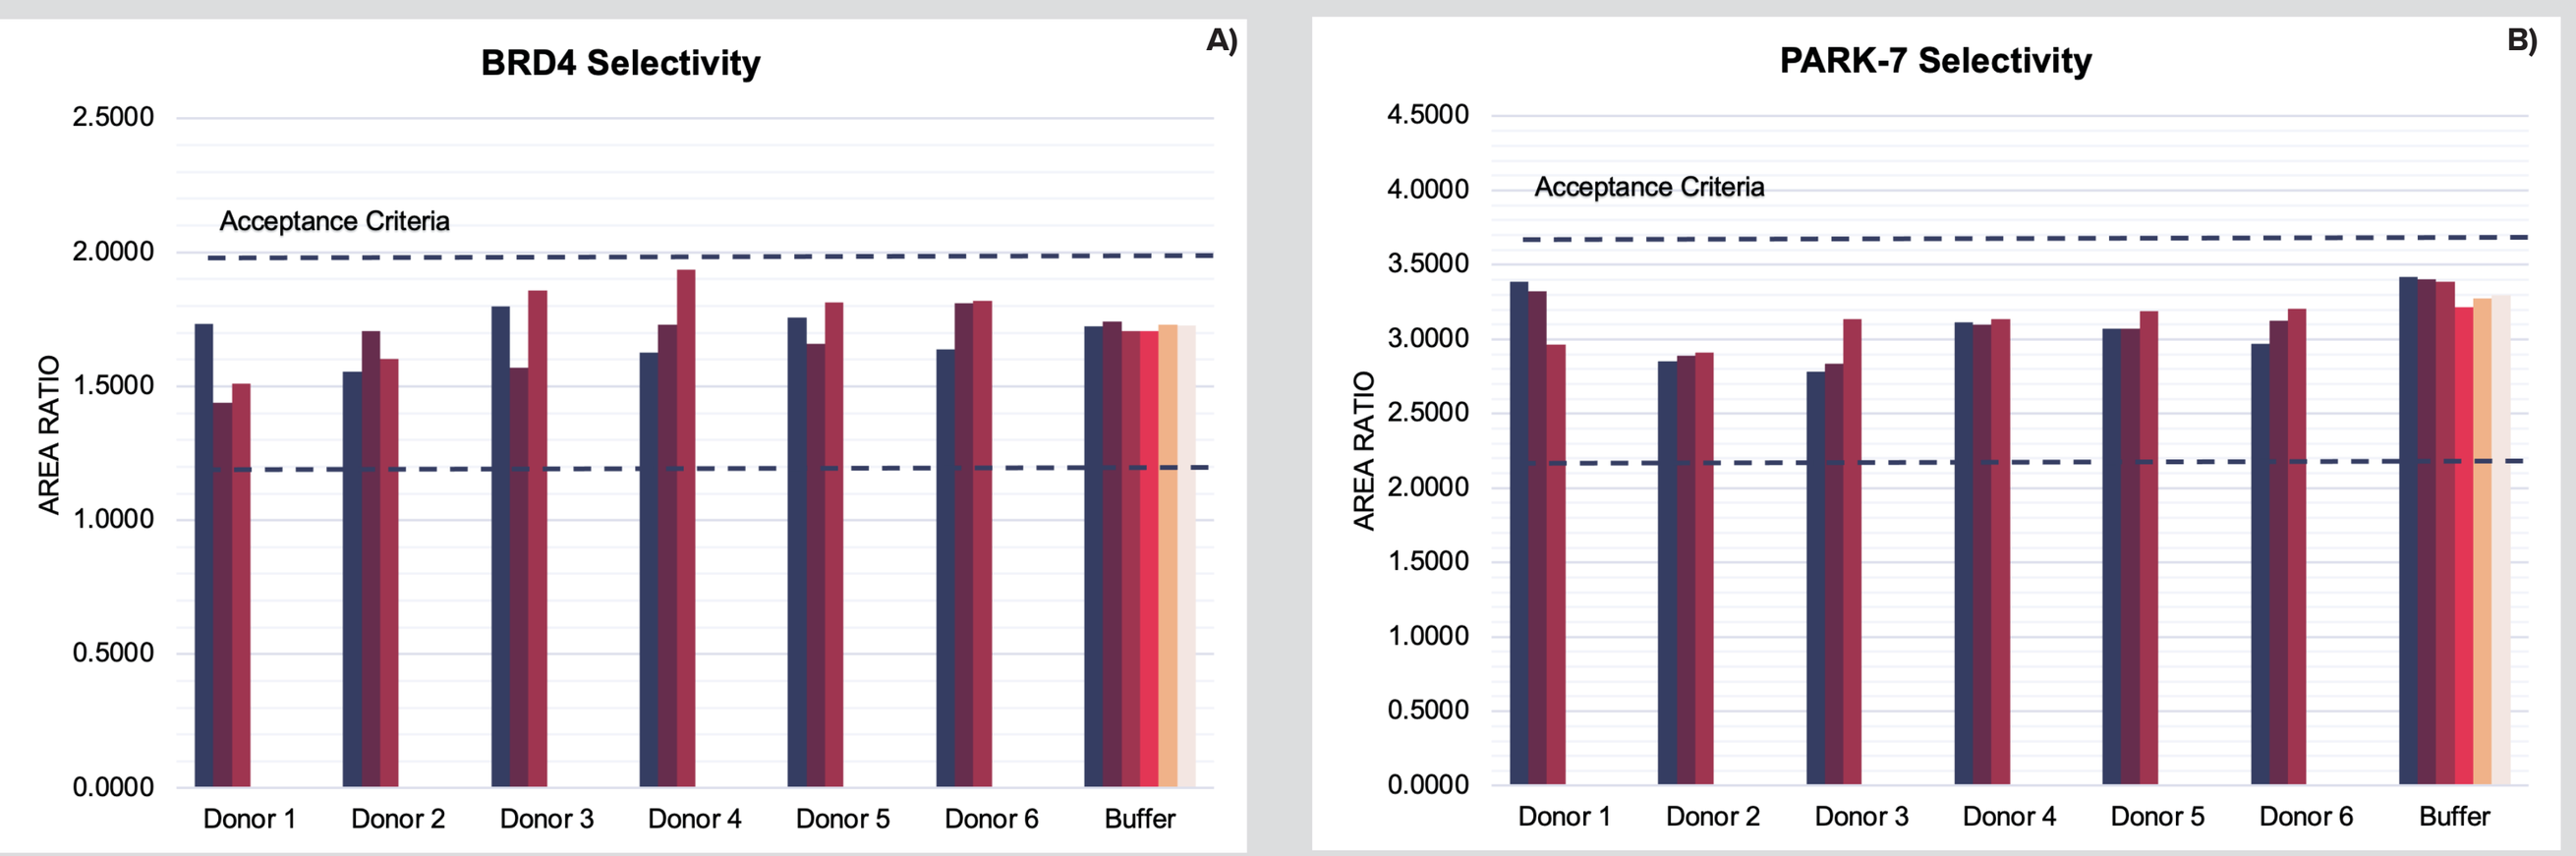
BRD4 Selectivity
A)
Acceptance Criteria
AREA RATIO
Donor 1
Donor 2
Donor 3
Donor 4
Donor 5
Donor 6
Buffer
PARK-7 Selectivity
B)

Figure 5: A) BRD4 assay selectivity: 3 replicates per donor, 6 for buffer samples; **B)** PARK-7 Assay selectivity: 3 replicates per donor, 6 for buffer samples.

Due to their endogenous presence, the selectivity of the two biomarkers is assessed using the peak area ratio of the quantifier to qualifier MRM transitions in buffer vs. extracted matrix samples. The percent difference between the transition ratios in matrix and buffer must not exceed 30.0%.

Testing Parallelism and Assay Troubleshooting

Table 3. Parallelism for the number of processed cells.

PARK-7	Donor 1		Donor 2		Donor 3	
	2X Cells	1X Cells	2X Cells	1X Cells	2X Cells	1X Cells
Area Ratio	18.5874	21.2314	24.1546	26.8097	20.4082	24.1546
	18.1488	21.2314	21.9780	27.7008	19.6464	25.9740
	19.4932	20.5339	24.6914	25.1889	20.4499	23.9808
Mean	18.7431	20.9989	23.6080	26.5665	20.1682	24.7031
%CV	3.7%	1.9%	6.1%	4.8%	2.2%	4.5%
%Difference	-	12.0%	-	12.5%	N/Ap	22.5%
BRD4	**Donor 1 2X Cells**	**Donor 1 1X Cells**	**Donor 2 2X Cells**	**Donor 2 1X Cells**	**Donor 3 2X Cells**	**Donor 3 1X Cells**
Area Ratio	0.0546	0.0683	0.0481	0.0455	0.0590	0.0648
	0.0602	0.0614	0.0473	0.0386	0.0572	0.0729
	0.0499	0.0729	0.0517	0.0337	0.0622	0.0781
Mean	0.0549	0.0675	0.0490	0.0393	0.0595	0.0719
%CV	9.4%	8.6%	4.8%	15.1%	4.3%	9.3%
%Difference	-	23.0%	-	-19.8%	N/Ap	20.8%
BRD4 Normalized	**Donor 1 2X Cells**	**Donor 1 1X Cells**	**Donor 2 2X Cells**	**Donor 2 1X Cells**	**Donor 3 2X Cells**	**Donor 3 1X Cells**
BRD4 Area Ratio vs. PARK-7 Area Ratio	0.0029	0.0032	0.0020	0.0017	0.0029	0.0027
	0.0033	0.0029	0.0022	0.0014	0.0029	0.0028
	0.0026	0.0036	0.0021	0.0013	0.0030	0.0033
Mean	0.0029	0.0032	0.0021	0.0015	0.0029	0.0029
%CV	12.1%	11.0%	4.8%	13.9%	2.0%	11.1%
%Difference	-	10.3%	-	-28.6%	N/Ap	0.0%

Parallelism assessment of PBMC isolations performed from varying cell inputs revealed inconsistent outcomes. Normalized BRD4 levels did not display a consistent trend, with responses either improving or deteriorating depending on the donor. These findings prompted a thorough re-evaluation of the entire workflow, from PBMC collection and stability through homogenization, total protein processing, and digestion efficiency. Pinpointing the specific steps at which the two target proteins diverge in behavior is critical to control the assay performances and confidently complete the method validation.

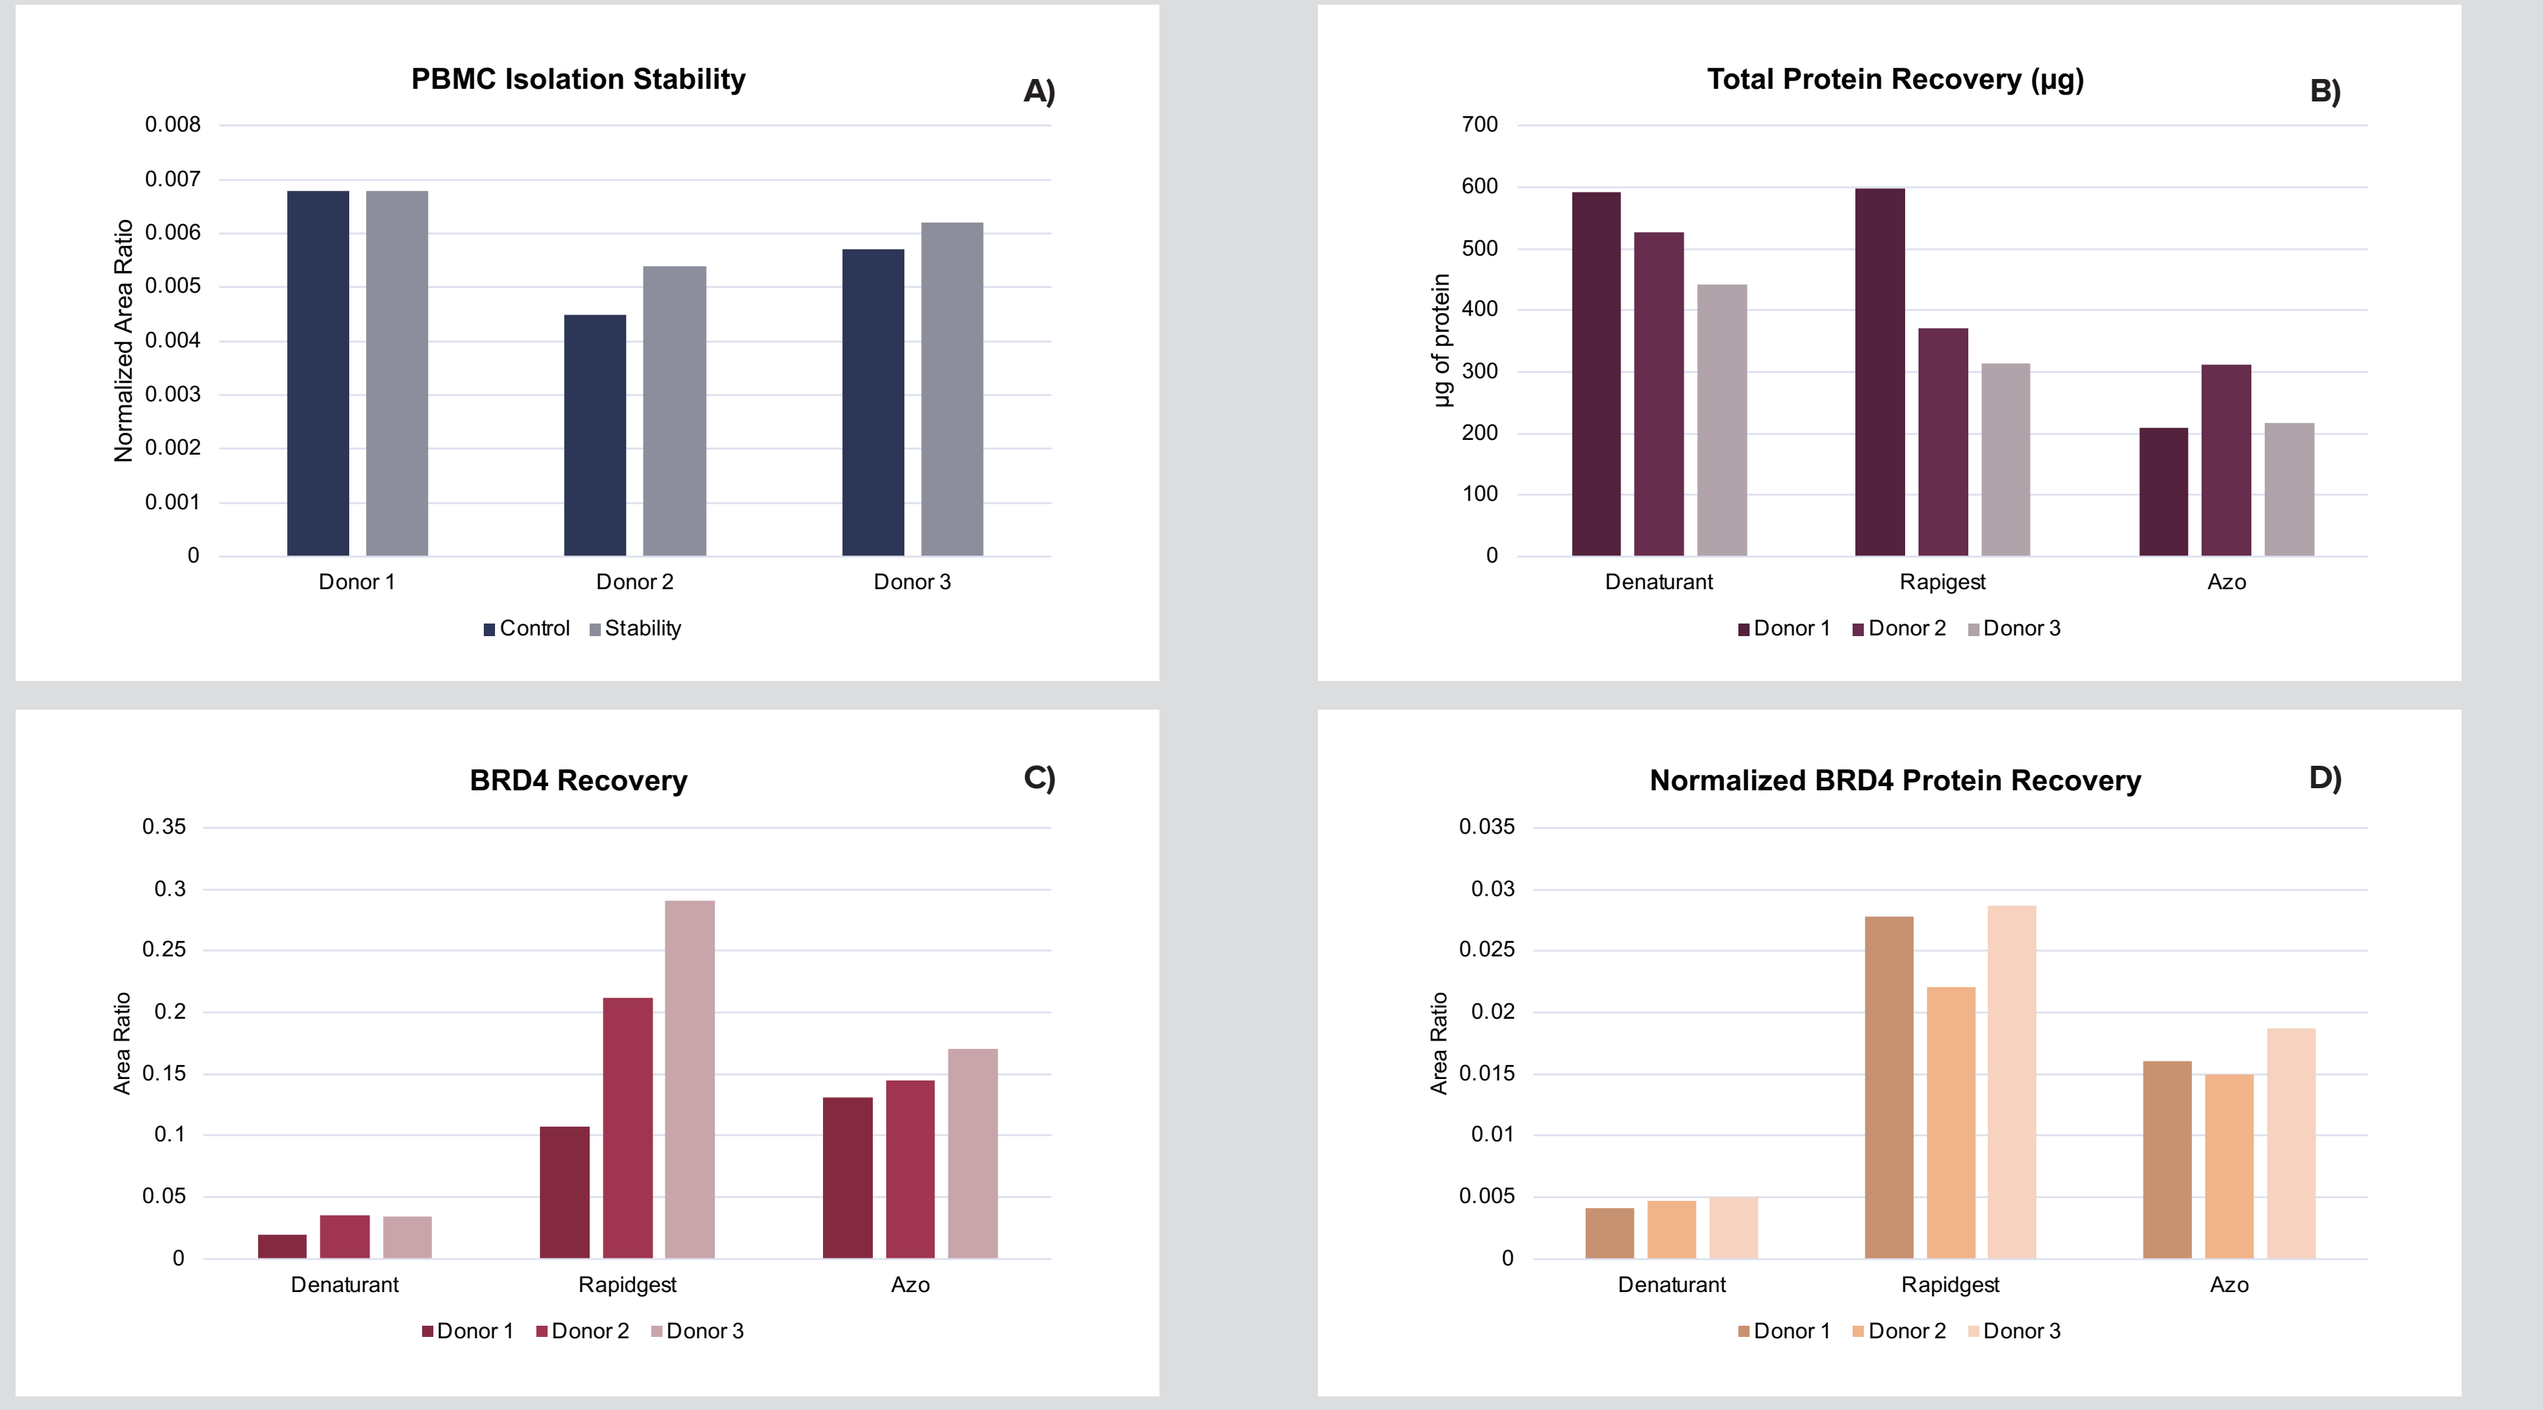
PBMC Isolation Stability
A)
Normalized Area Ratio
Donor 1
Donor 2
Donor 3
Control Stability
Total Protein Recovery (µg)
B)
µg of protein
Denaturant
Rapigest
Azo
Donor 1 Donor 2 Donor 3
BRD4 Recovery
C)
Area Ratio
Denaturant
Rapidgest
Azo
Normalized BRD4 Protein Recovery
D)

Figure 6. A) Stability test of normalized BRD4 in PBMC with 2h benchtop stability at 5°C post centrifugation; **B)** Total protein recovery per homogenization condition; **C)** BRD4 recovery per tested homogenization condition; **D)** Normalized BRD4 recovery per homogenization condition.

Experimental data indicate that the only point where the 2 protein differs is the PBMC lysis. The denaturant-based homogenization protocol yields the highest total protein recovery but the lowest recovery for BRD4. In contrast, surfactant-based homogenization approaches improve analyte recovery and enable more consistent normalization of BRD4 relative to PARK7. Based on overall performance, RapiGest was selected for further validation, as it provided improved sensitivity for BRD4 compared to the other conditions.

Table 4. A) Parallelism for the number of processed cells during PBMC isolation; **B)** Parallelism for the amount of matrix loaded during sample processing.

A) PARK-7	5X Cells	2.5 X Cells	1X Cells
Area Ratio	0.5331	0.5112	0.5101
	0.5020	0.4920	0.4588
	0.4941	0.5031	0.4907
Mean	0.5097	0.5021	0.4865
%CV	4.0	1.9	5.3
%Difference	-	-1.5	-4.6
BRD4	**5X Cells**	**2.5 X Cells**	**1X Cells**
Area Ratio	0.0564	0.0470	0.0600
	0.0572	0.0496	0.0507
	0.0587	0.0469	0.0515
Mean	0.0574	0.0478	0.0541
%CV	2.0	3.2	9.5
%Difference	-	-16.7	-5.7
Normalized BRD4	**5X Cells**	**2.5 X Cells**	**1X Cells**
BRD4 Area Ratio vs. PARK-7 Area Ratio	0.1058	0.0919	0.1176
	0.1139	0.1008	0.1105
	0.1188	0.0932	0.1050
Mean	0.1128	0.0953	0.1110
%CV	5.8	5.0	5.7
%Difference	-	-15.5	-1.6

B) PARK-7	25µg Processed	6µg Processed	25µg Processed	6µg Processed
Area Ratio	2.3981	0.5331	2.3652	0.5112
	2.4102	0.5020	2.2619	0.4920
	2.3348	0.4941	2.1867	0.5031
Mean	2.3810	0.5097	2.2713	0.5021
%CV	1.7	4.0	3.9	1.9
%Difference	-	-10.1	-	-7.2
BRD4	**25µg Processed**	**6µg Processed**	**25µg Processed**	**6µg Processed**
Area Ratio	0.2407	0.0564	0.2197	0.0470
	0.2606	0.0572	0.2174	0.0496
	0.2635	0.0587	0.2091	0.0469
Mean	0.2549	0.0574	0.2154	0.0478
%CV	4.9	2.0	2.6	3.2
%Difference	-	-5.4	-	-6.8
Normalized BRD4	**25µg Processed**	**6µg Processed**	**25µg Processed**	**6µg Processed**
BRD4 Area Ratio vs. PARK-7 Area Ratio	0.1004	0.1058	0.0929	0.0919
	0.1081	0.1139	0.0961	0.1008
	0.1129	0.1188	0.0956	0.0932
Mean	0.1071	0.1128	0.0949	0.0953
%CV	5.9	5.8	1.8	5.0
%Difference	-	5.3	-	0.4

Table 5. A) Processed reconstituted stability at 5°C for 4 days; **B)** PBMC homogenate stability at -20°C for 11 days.

A) PARK-7	QC Low		QC High	
	Control	Stability	Control	Stability
Area Ratio	0.0047	0.0046	0.1375	0.1320
	0.0046	0.0046	0.1407	0.1317
	0.0045	0.0045	0.1381	0.1380
	0.0045	0.0047	0.1420	0.1398
Mean	0.0046	0.0046	0.1396	0.1354
%CV	2.1%	1.8%	1.5%	3.1%
%Difference	-	0.0%	N/Ap	3.0%
BRD4	**QC Low Control**	**QC Low Stability**	**QC High Control**	**QC High Stability**
Area Ratio	0.3203	0.3089	8.3194	8.1428
	0.3158	0.2995	8.7684	8.4120
	0.3127	0.3119	8.7721	8.4891
	0.3143	0.3060	8.6009	8.5681
Mean	0.3158	0.3066	8.6152	8.4030
%CV	1.0%	1.7%	2.5%	2.2%
%Difference	-	2.9%	-	2.5%
Normalized BRD4	**QC Low Control**	**QC Low Stability**	**QC High Control**	**QC High Stability**
BRD4 Area Ratio vs. PARK-7 Area Ratio	68.1489	67.1522	60.5047	61.6879
	68.6522	65.1087	62.3198	63.8724
	69.4889	69.3111	63.5199	61.5152
	69.8444	65.1064	60.5697	61.2883
Mean	69.0336	66.6696	61.7285	62.0910
%CV	1.1%	3.0%	2.4%	1.9%
%Difference	-	3.4%	-	0.6%

B) PARK-7	QC Low		QC High	
	Control	Stability	Control	Stability
Area Ratio	0.0044	0.0047	0.1303	0.1368
	0.0051	0.0043	0.1308	0.1386
	0.0047	0.0043	0.1311	0.1340
	0.0044	0.0047	0.1318	0.1357
Mean	0.0047	0.0045	0.1310	0.1363
%CV	7.1%	5.1%	0.5%	1.4%
%Difference	-	4.3%	-	4.0%
BRD4	**QC Low Control**	**QC Low Stability**	**QC High Control**	**QC High Stability**
Area Ratio	0.3472	0.2971	8.2790	7.9339
	0.3093	0.3034	7.8641	7.8526
	0.3235	0.3046	7.8322	8.2465
	0.3075	0.3129	8.0861	7.8417
Mean	0.3219	0.3045	8.0154	7.9687
%CV	5.7%	2.1%	2.6%	2.4%
%Difference	-	5.4%	-	0.6%
Normalized BRD4	**QC Low Control**	**QC Low Stability**	**QC High Control**	**QC High Stability**
BRD4 Area Ratio vs. PARK-7 Area Ratio	78.9091	63.2128	63.5380	57.9963
	60.6471	70.5581	60.1231	56.6566
	68.8298	70.8372	59.7422	61.5410
	69.8864	66.5745	61.3513	57.7870
Mean	69.5681	67.7957	61.1887	58.4952
%CV	10.7%	5.3%	2.8%	3.6%
%Difference	-	2.5%	-	4.4%

CONCLUSION

- The developed method met all fit-for-purpose validation criteria, including up to 90% target reduction for detection of target degrader activity, parallelism, and specificity, demonstrating its suitability for quantitative applications. The assay showed consistent performance across multiple runs and donors in complex biological matrices.
- Careful optimization of the workflow—particularly lysis and digestion—was critical to overall assay performance. The inclusion of surfactants significantly improved protein recovery and enabled reliable normalization of BRD4 using PARK-7 by standardizing the extraction efficiency of both proteins. The identification of a condition where both proteins were extracted with similar yield reduced variability introduced during sample preparation and contributed to improved consistency across experimental conditions.
- Mass spectrometry–based normalization provided a robust and precise alternative to conventional BCA approaches, minimizing the impact of contaminants, red blood cells, and reagent variability while improving overall accuracy and reproducibility.

REFERENCES

[1] Wiśniewski, J.(2016). Journal of Proteome Research, 15(7), 2321-2326.
[2] https://www.proteinatlas.org/ENSG00000116288-PARK7/tissue
[3] Sun, M. (2023). International Journal of Molecular Sciences, 24(8):7409.
[4] Zhang, W. (2026). Trends in Pharmacological Sciences, 47(1), 15–39

Send inquiries to ***isales@cellcarta.com*** with WRIB 2026 in email subject.

DAY2-P21

www.cellcarta.com