Fit-for-purpose validation and inter-laboratory cross-validation of a TBNK flow cytometry assay with improved B cell monitoring capabilities

Marylène Fortin[1], Katiane Roversi[1], Maude Boisvert[1], Laïla-Aïcha Hanafi[1], David Possamaï[1], Audrey Connolly[1], Da Woon Park[1], Fatma Babikr[1], Christophe Henrard[2], Aude Ytebrouck[2], Shu-Hui Gao[3], Xin-Xin Wei[3], Eilidh Main[4], Lachlan Hyde[4]

1. CellCarta Biosciences, Inc. (Montreal, Quebec, Canada); 2. CellCarta Biosciences, SA (Gosselies, Belgium); 3. CellCarta China Ltd (Jining, Shandong Province, China); 4. CellCarta Pty Ltd (Baulkham Hills, Sydney, Australia)

POSTER NUMBER: DAY2-P18

CellCarta

INTRODUCTION

Standard flow cytometry TBNK assays are widely used in oncology clinical trials for the monitoring of T, B and NK lymphocytes populations in patients. Commercially available TBNK kits rely solely on CD19 for the identification of B cells. In this work, we validated a new assay designed for lymphocyte subsets enumeration in peripheral whole blood. The addition of an anti-CD20 antibody to the standard TBNK panel provided enhanced B cell monitoring capabilities, particularly relevant in the context of clinical trials involving B-cell depleting therapies where the target antigen (CD19) can be masked by the drug or downregulated.

METHODS

Samples, staining method, acquisition and data analysis

Whole blood from 6 healthy donors (HD) collected in Cyto-Chex™ blood collection tubes were used for all validation assessments (except for the linearity & LLOQ assessments where samples from 3 HD were used). The BD Multi-Check™ Controls (Normal and CD4 Low) were included in each run and used as run controls. Sample staining was performed in triplicates using the BD Multi-test™ 6-color TBNK reagent (CD45, CD3, CD4, CD8, CD19 and CD16/56) with an anti-CD20 antibody drop-in (Table 1). The methodology for the assay uses a lyse/no wash staining procedure with BD Trucount™ tubes to determine absolute counts reported in cells/µL. Sample acquisition was performed using the BD FACSLyric™ instrument and data analysis was performed using the CellEngine® software (CellCarta) using the gating strategy shown in Figure 1.

Table 1: Panel composition.

Marker	Fluorochrome
CD20	V450
CD4*	PE-Cy7
CD45*	PerCP-Cy5.5
CD16*	PE
CD56*	PE
CD3*	FITC
CD8*	APC-Cy7
CD19*	APC

Markers part of the BD Multitest™ 6-color TBNK reagent.

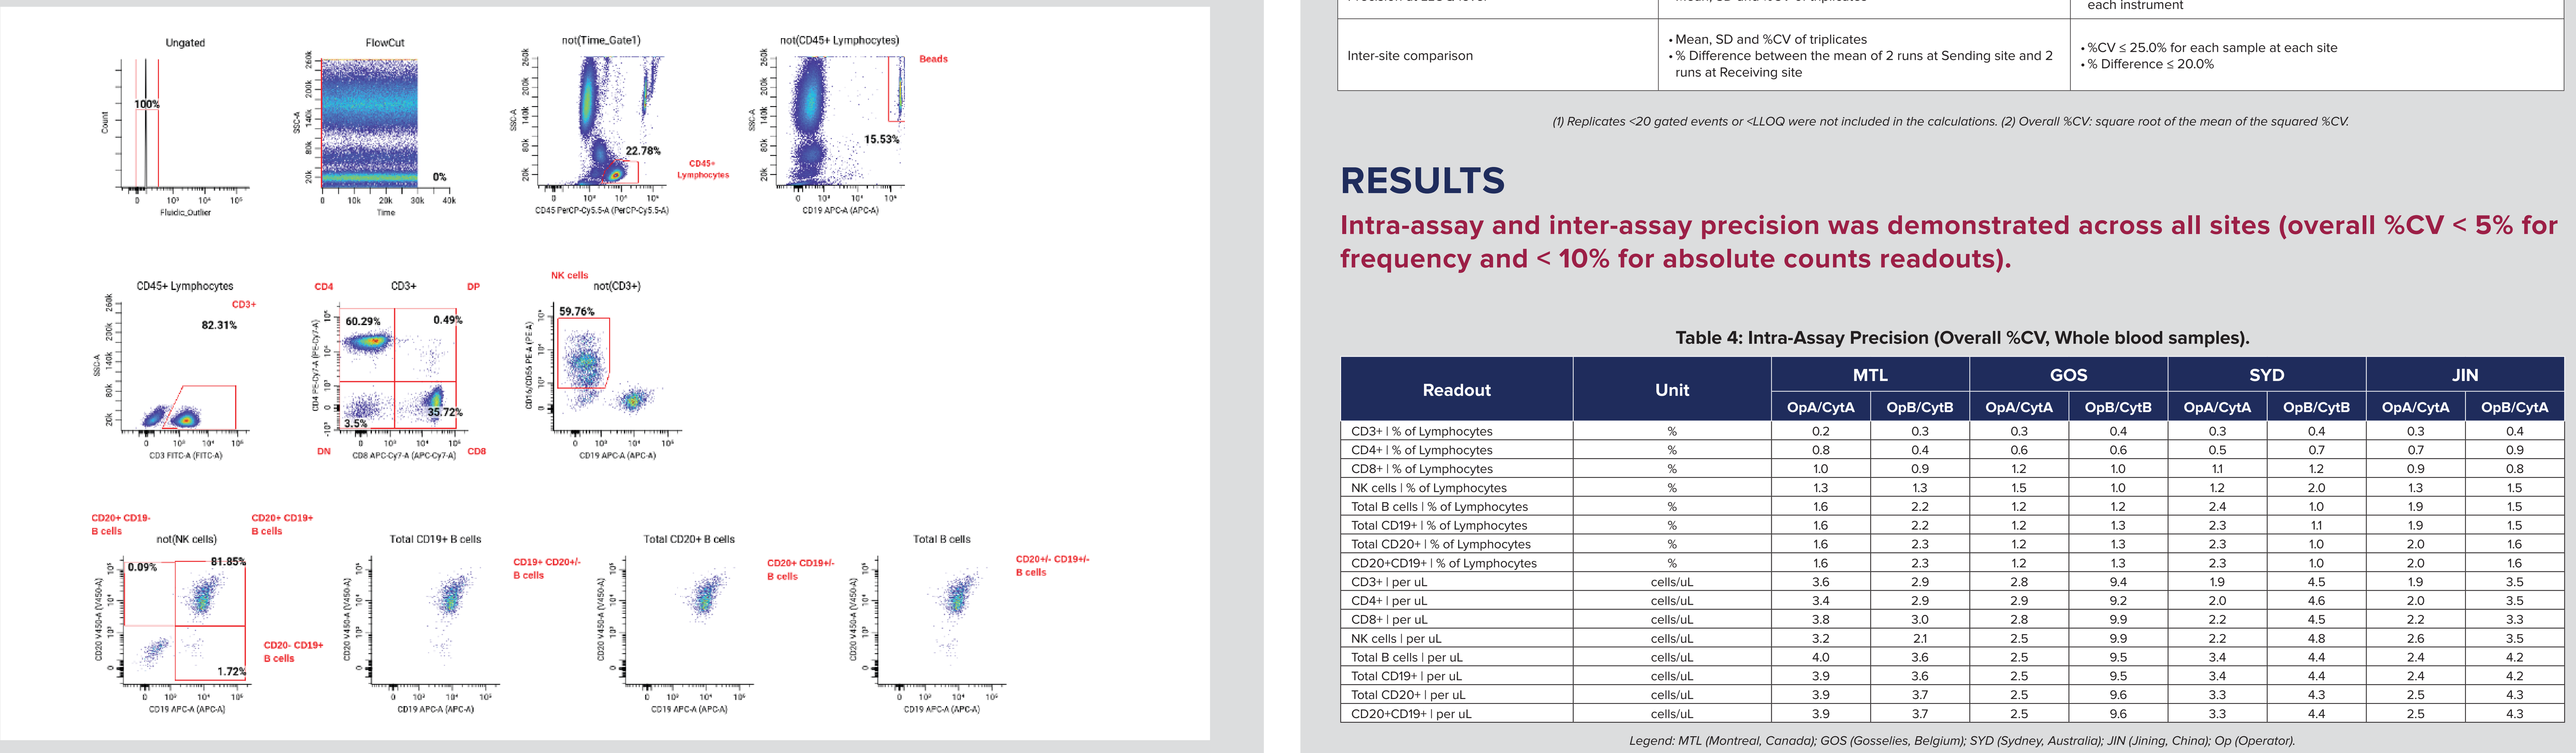
Ungated
100%
FlowCut
0%
not(Time_Gate1)
22.78%
CD45+ Lymphocytes
not(CD45+ Lymphocytes)
Beads
15.53%
CD45+ Lymphocytes
CD3+
82.31%
CD4
CD3+
DP
60.29%
0.49%
3.5%
35.72%
DN
CD8
NK cells
not(CD3+)
59.76%
CD20+ CD19- B cells
not(NK cells)
CD20+ CD19+ B cells
0.09%
81.85%
1.72%
CD20- CD19+ B cells
Total CD19+ B cells
CD19+ CD20+/- B cells
Total CD20+ B cells
CD20+ CD19+/- B cells
Total B cells
CD20+/- CD19+/- B cells

Figure 1: Gating strategy.

Table 2: Validation assessments.

Validation Assessments	Experimental Design	Validation site: MTL	Cross-validation sites: GOS	SYD	JIN (1)
Intra-assay precision	• Whole blood and controls: 2 runs on the same day • 2 operators / 2 instruments	X	X	X	X
Inter-assay precision	• Whole blood: 2 runs on the same day • Controls: 4 runs over +2 days • 2 operators / 2 instruments	X	X	X	X
Specimen stability	• Whole blood samples from Precision assessment • Stored at room temperature and at 2-8ºC for up to 7 days • 1 operator / 1 instrument	X			
Processed samples stability	• Stained samples from Precision assessment • Acquired within 2h or 16h-24h post staining • 1 operator / 1 instrument	X			
Linearity and LLOQ	• Whole blood samples diluted in plasma (8 dilution levels) • 2 operators / 2 instruments	X			
Precision at LLOQ level	• Whole blood samples diluted in plasma (7 dilution levels) • 2 operators / 2 instruments		X	X	X
Inter-site comparison	• 2 Control samples (same lot shared by all sites) • 2 runs per site • 2 operators / 2 instruments	X	X	X	X

Legend: MTL (Montreal, Canada); GOS (Gosselies, Belgium); SYD (Sydney, Australia); JIN (Jining, China); LLOQ (Lower Limit of Quantification). (1) For the Jining site, only 1 instrument was used.

Table 3: Data calculations and acceptance criteria.

Validation Assessments	Data calculations (1)	Acceptance criteria
Intra-assay precision	• Mean, SD and %CV of triplicates • Overall %CV from all samples (2)	Overall %CV ≤ 25.0%
Inter-assay precision	• Mean, SD and %CV of all replicates from all runs • Overall %CV from all whole blood samples	Overall %CV ≤ 25.0% for whole blood samples %CV ≤ 25.0% for each control
Specimen stability	• Mean, SD and %CV of triplicates • % Difference between timepoint and baseline	• %CV ≤ 25.0% at baseline & timepoint • % Difference ≤ 25.0% for 5/6 samples • For each readout, stability is defined as the latest timepoint meeting criteria
Processed samples stability	• Mean, SD and %CV of triplicates • % Difference between timepoint and baseline	• %CV ≤ 25.0% at baseline & timepoint • % Difference ≤ 20.0% for 5/6 samples • For each readout, stability is defined as the latest timepoint meeting criteria
Linearity and LLOQ	• Mean, SD and %CV of triplicates • Linear regression between measured value and theoretical value	• R2 ≥ 0.95 for each instrument • %CV ≤ 30.0% at LLOQ • For each instrument, LLOQ was defined as the lowest value from all donors/ dilution levels that meets criteria; assay LLOQ was defined as the highest value from the 2 instruments
Precision at LLOQ level	• Mean, SD and %CV of triplicates	• %CV ≤ 30.0% for at least 1 donor/dilution level for value within LLOQ range, for each instrument
Inter-site comparison	• Mean, SD and %CV of triplicates • % Difference between the mean of 2 runs at Sending site and 2 runs at Receiving site	• %CV ≤ 25.0% for each sample at each site • % Difference ≤ 20.0%

(1) Replicates <20 gated events or <LLOQ were not included in the calculations. (2) Overall %CV: square root of the mean of the squared %CV.

RESULTS

Intra-assay and inter-assay precision was demonstrated across all sites (overall %CV < 5% for frequency and < 10% for absolute counts readouts).

Table 4: Intra-Assay Precision (Overall %CV, Whole blood samples).

Readout	Unit	MTL OpA/CytA	MTL OpB/CytB	GOS OpA/CytA	GOS OpB/CytB	SYD OpA/CytA	SYD OpB/CytB	JIN OpA/CytA	JIN OpB/CytA
CD3+ \| % of Lymphocytes	%	0.2	0.3	0.3	0.4	0.3	0.4	0.3	0.4
CD4+ \| % of Lymphocytes	%	0.8	0.4	0.6	0.6	0.5	0.7	0.7	0.9
CD8+ \| % of Lymphocytes	%	1.0	0.9	1.2	1.0	1.1	1.2	0.9	0.8
NK cells \| % of Lymphocytes	%	1.3	1.3	1.5	1.0	1.2	2.0	1.3	1.5
Total B cells \| % of Lymphocytes	%	1.6	2.2	1.2	1.2	2.4	1.0	1.9	1.5
Total CD19+ \| % of Lymphocytes	%	1.6	2.2	1.2	1.3	2.3	1.1	1.9	1.5
Total CD20+ \| % of Lymphocytes	%	1.6	2.3	1.2	1.3	2.3	1.0	2.0	1.6
CD20+CD19+ \| % of Lymphocytes	%	1.6	2.3	1.2	1.3	2.3	1.0	2.0	1.6
CD3+ \| per uL	cells/uL	3.6	2.9	2.8	9.4	1.9	4.5	1.9	3.5
CD4+ \| per uL	cells/uL	3.4	2.9	2.9	9.2	2.0	4.6	2.0	3.5
CD8+ \| per uL	cells/uL	3.8	3.0	2.8	9.9	2.2	4.5	2.2	3.3
NK cells \| per uL	cells/uL	3.2	2.1	2.5	9.9	2.2	4.8	2.6	3.5
Total B cells \| per uL	cells/uL	4.0	3.6	2.5	9.5	3.4	4.4	2.4	4.2
Total CD19+ \| per uL	cells/uL	3.9	3.6	2.5	9.5	3.4	4.4	2.4	4.2
Total CD20+ \| per uL	cells/uL	3.9	3.7	2.5	9.6	3.3	4.3	2.5	4.3
CD20+CD19+ \| per uL	cells/uL	3.9	3.7	2.5	9.6	3.3	4.4	2.5	4.3

Legend: MTL (Montreal, Canada); GOS (Gosselies, Belgium); SYD (Sydney, Australia); JIN (Jining, China); Op (Operator).

Table 5: Inter-Assay Precision (Overall %CV for Whole blood samples and %CV for Controls)

Readout	Unit	MTL Whole blood	MTL Normal CTRL	MTL CD4 Low CTRL	GOS Whole blood	GOS Normal CTRL	GOS CD4 Low CTRL	SYD Whole blood	SYD Normal CTRL	SYD CD4 Low CTRL	JIN Whole blood	JIN Normal CTRL	JIN CD4 Low CTRL
CD3+ \| % of Lymphocytes	%	0.3	1.0	1.0	0.6	0.5	0.7	0.5	0.8	0.6	0.4	0.6	0.8
CD4+ \| % of Lymphocytes	%	0.7	1.5	2.7	0.8	0.5	1.6	0.7	1.0	2.3	0.8	1.0	1.3
CD8+ \| % of Lymphocytes	%	1.1	1.4	1.0	1.1	1.2	0.8	1.1	1.7	1.0	1.0	1.0	0.9
NK cells \| % of Lymphocytes	%	1.4	3.7	1.5	1.6	1.1	1.4	2.0	2.6	1.6	1.5	2.5	1.6
Total B cells \| % of Lymphocytes	%	1.9	1.4	1.5	1.5	1.3	1.2	2.3	1.2	1.0	1.7	1.5	0.7
Total CD19+ \| % of Lymphocytes	%	1.9	1.4	1.5	1.5	1.3	1.2	2.3	1.2	1.2	1.7	1.5	1.1
Total CD20+ \| % of Lymphocytes	%	2.1	1.3	1.6	1.6	1.6	1.5	2.3	1.3	1.1	1.8	1.4	1.0
CD20+CD19+ \| % of Lymphocytes	%	2.0	1.3	1.7	1.6	1.5	1.5	2.3	1.3	1.3	1.8	1.4	1.3
CD3+ \| per uL	cells/uL	3.9	4.6	4.2	7.7	2.4	4.2	6.2	3.9	8.9	4.4	3.9	1.6
CD4+ \| per uL	cells/uL	4.1	4.6	5.5	7.5	2.6	3.7	6.2	4.1	9.4	4.5	4.2	1.7
CD8+ \| per uL	cells/uL	3.9	5.2	3.9	7.9	2.1	4.7	6.3	4.1	8.3	4.6	3.8	2.4
NK cells \| per uL	cells/uL	4.2	4.2	3.8	8.4	3.0	5.3	7.6	3.1	7.9	4.8	5.4	2.1
Total B cells \| per uL	cells/uL	4.8	4.3	4.2	7.6	3.4	3.9	7.4	3.4	9.5	5.1	4.6	2.4
Total CD19+ \| per uL	cells/uL	4.8	4.3	4.2	7.6	3.5	3.9	7.5	3.4	9.7	5.2	4.7	2.6
Total CD20+ \| per uL	cells/uL	4.9	4.3	4.6	7.6	3.6	4.4	7.4	3.4	9.5	5.2	4.6	2.6
CD20+CD19+ \| per uL	cells/uL	4.9	4.4	4.6	7.6	3.6	4.4	7.5	3.4	9.7	5.2	4.6	2.9

Legend: MTL (Montreal, Canada); GOS (Gosselies, Belgium); SYD (Sydney, Australia); JIN (Jining, China); CTRL (Control).

All frequency and absolute counts readouts were shown to be stable for up to 7 days of sample storage at room temperature or 2-8ºC.

Table 6: Specimen Stability.

Readout	Unit	#HD with %CV≤25% & %Difference with baseline ≤25%: Day 1 (24h)	Day 2 (48h)	Day 3 (72h)	Day 4 (96h)	Day 7 (168h)	VALIDATED STABILITY (RT and 2-8ºC)
CD3+ \| % of Lymphocytes	%	6	6	6	6	6	168h
CD4+ \| % of Lymphocytes	%	6	6	6	6	6	168h
CD8+ \| % of Lymphocytes	%	6	6	6	6	6	168h
NK cells \| % of Lymphocytes	%	6	6	6	6	6	168h
Total B cells \| % of Lymphocytes	%	6	6	6	6	6	168h
Total CD19+ \| % of Lymphocytes	%	6	6	6	6	6	168h
Total CD20+ \| % of Lymphocytes	%	6	6	6	6	6	168h
CD20+CD19+ \| % of Lymphocytes	%	6	6	6	6	6	168h
CD3+ \| per uL	cells/uL	6	6	6	6	6	168h
CD4+ \| per uL	cells/uL	6	6	6	6	6	168h
CD8+ \| per uL	cells/uL	6	6	6	6	6	168h
NK cells \| per uL	cells/uL	6	6	6	6	6	168h
Total B cells \| per uL	cells/uL	6	6	6	6	6	168h
Total CD19+ \| per uL	cells/uL	6	6	6	6	6	168h
Total CD20+ \| per uL	cells/uL	6	6	6	6	6	168h
CD20+CD19+ \| per uL	cells/uL	6	6	6	6	6	168h

Legend: #HD (Number of Healthy Donors); %CV (coefficient of variation); RT (room temperature); ºC (degree Celcius); h (hour).

Processed samples were found to be stable for up to 21h post-staining for all frequency and absolute counts readouts.

Table 7: Processed samples Stability.

Readout	Unit	#HD with %CV≤25% & %Difference with baseline ≤20%	VALIDATED STABILITY
CD3+ \| Percent of Lymphocytes	%	6	21h39m
CD4+ \| Percent of Lymphocytes	%	6	21h39m
CD8+ \| Percent of Lymphocytes	%	6	21h39m
NK cells \| Percent of Lymphocytes	%	6	21h39m
Total B cells \| Percent of Lymphocytes	%	6	21h39m
Total CD19+ \| Percent of Lymphocytes	%	6	21h39m
Total CD20+ \| Percent of Lymphocytes	%	6	21h39m
CD20+CD19+ \| Percent of Lymphocytes	%	6	21h39m
CD3+ \| per uL	cells/uL	6	21h39m
CD4+ \| per uL	cells/uL	6	21h39m
CD8+ \| per uL	cells/uL	6	21h39m
NK cells \| per uL	cells/uL	6	21h39m
Total B cells \| per uL	cells/uL	6	21h39m
Total CD19+ \| per uL	cells/uL	6	21h39m
Total CD20+ \| per uL	cells/uL	6	21h39m
CD20+CD19+ \| per uL	cells/uL	6	21h39m

Legend: #HD (Number of Healthy Donors); %CV (coefficient of variation); h (hour); m (minutes).

LLOQ values were below 5 cells/uL for all absolute counts readouts, and linearity was demonstrated.

Table 8: Lower Limit of Quantification (LLOQ).

Readout	Unit	LLOQ (CytA)	LLOQ (CytB)	Final assay LLOQ (1)
CD3+ \| per uL	cells/uL	4.2	1.9	4.2
CD4+ \| per uL	cells/uL	1.1	1.0	1.1
CD8+ \| per uL	cells/uL	0.8	1.6	1.6
NK cells \| per uL	cells/uL	1.2	2.0	2.0
Total B cells \| per uL	cells/uL	0.9	1.4	1.4
Total CD19+ \| per uL	cells/uL	0.9	1.4	1.4
Total CD20+ \| per uL	cells/uL	2.1	1.3	2.1
CD20+CD19+ \| per uL	cells/uL	2.1	1.3	2.1

Legend: CYT: Cytometer. (1) Precision at LLOQ level was verified across all CellCarta sites and cytometers (data not shown).

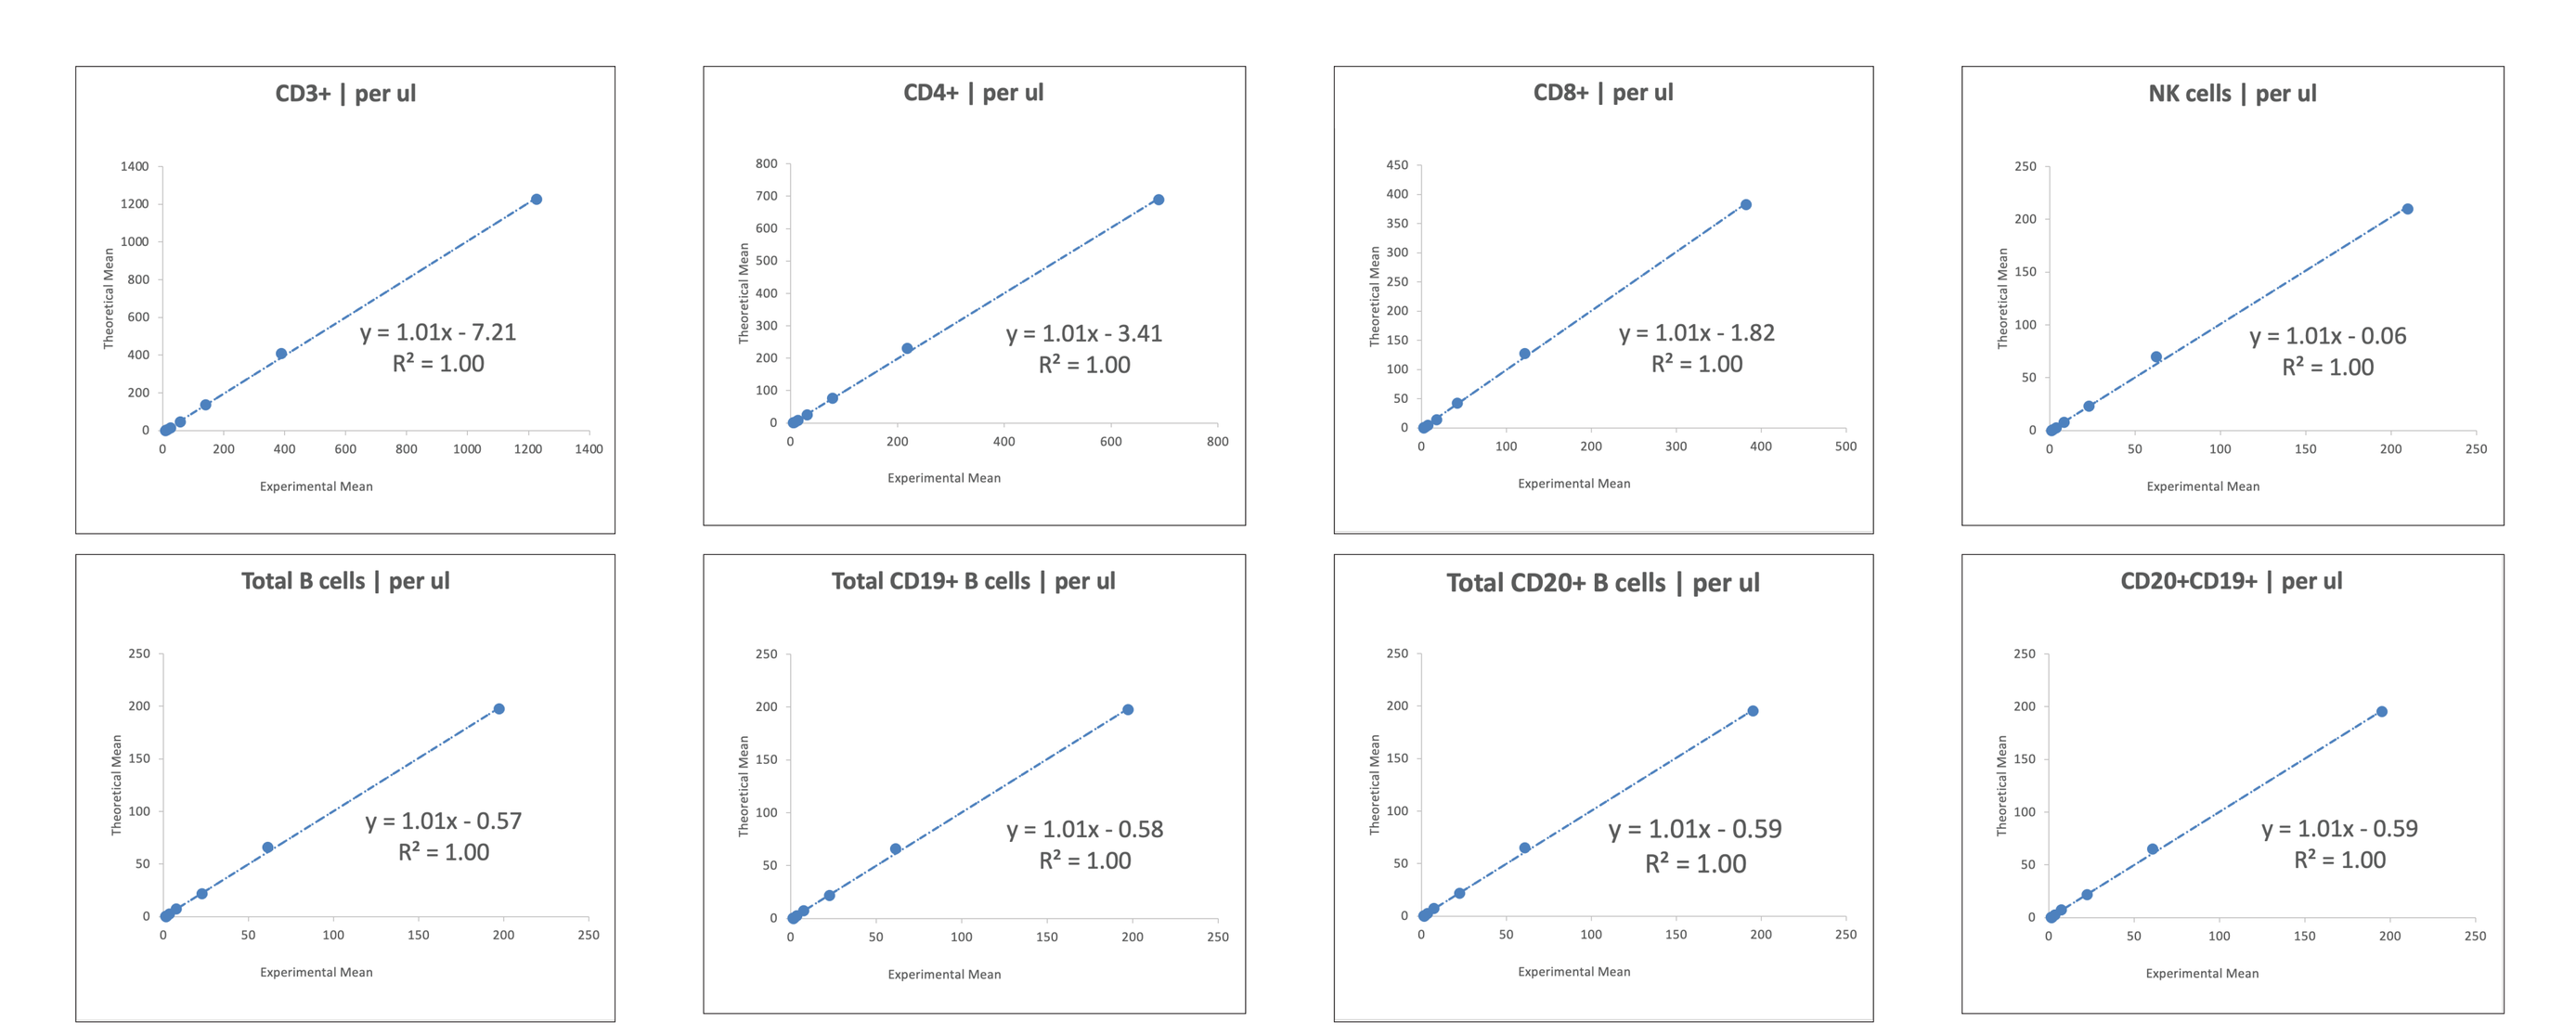
CD3+ | per ul
y = 1.01x - 7.21
R² = 1.00
CD4+ | per ul
y = 1.01x - 3.41
R² = 1.00
CD8+ | per ul
y = 1.01x - 1.82
R² = 1.00
NK cells | per ul
y = 1.01x - 0.06
R² = 1.00
Total B cells | per ul
y = 1.01x - 0.57
R² = 1.00
Total CD19+ B cells | per ul
y = 1.01x - 0.58
R² = 1.00
Total CD20+ B cells | per ul
y = 1.01x - 0.59
R² = 1.00
CD20+CD19+ | per ul
y = 1.01x - 0.59
R² = 1.00
Experimental Mean
Theoretical Mean

Figure 2: Linearity. Data from 1 healthy donor and from 1 cytometer are shown.

Consistent results were obtained (less than 15% difference for all readouts) when control samples were tested at 4 CellCarta laboratories located in Canada, Belgium, Australia and China.

Table 9: Inter-site comparison (% Difference).

Readout	Unit	GOS vs MTL Normal CTRL	GOS vs MTL CD4 Low CTRL	SYD vs MTL Normal CTRL	SYD vs MTL CD4 Low CTRL	JIN vs MTL Normal CTRL	JIN vs MTL CD4 Low CTRL
CD3+ \| % of Lymphocytes	%	0.1	0.5	0.2	0.1	0.3	0.5
CD4+ \| % of Lymphocytes	%	0.4	0.7	0.7	0.4	0.2	0.7
CD8+ \| % of Lymphocytes	%	0.2	3.1	0.2	1.5	0.3	0.3
NK cells \| % of Lymphocytes	%	1.0	2.1	0.5	1.1	1.3	1.1
Total B cells \| % of Lymphocytes	%	0.1	0.6	0.8	1.2	0.0	1.6
Total CD19+ \| % of Lymphocytes	%	0.3	0.0	0.5	1.6	0.2	1.8
Total CD20+ \| % of Lymphocytes	%	0.1	0.4	0.6	1.8	0.3	1.2
CD20+CD19+ \| % of Lymphocytes	%	0.1	0.3	0.3	2.3	0.6	1.4
CD3+ \| per uL	cells/uL	7.2	9.6	6.8	0.3	0.0	4.9
CD4+ \| per uL	cells/uL	7.5	8.2	7.3	0.5	0.1	4.9
CD8+ \| per uL	cells/uL	7.3	12.5	6.4	1.7	0.0	4.7
NK cells \| per uL	cells/uL	6.2	11.5	6.2	1.3	1.4	5.5
Total B cells \| per uL	cells/uL	7.3	8.4	5.7	1.4	0.2	2.6
Total CD19+ \| per uL	cells/uL	7.5	9.1	6.1	1.8	0.0	2.4
Total CD20+ \| per uL	cells/uL	7.1	8.6	6.0	2.1	0.1	3.1
CD20+CD19+ \| per uL	cells/uL	7.3	9.3	6.3	2.5	0.3	2.8

Legend: MTL (Montreal, Canada); GOS (Gosselies, Belgium); SYD (Sydney, Australia); JIN (Jining, China); CTRL (Control).

CONCLUSION

In this study, a new TBNK assay with expanded B cell markers coverage was successfully validated. The assay demonstrated strong, reliable performance across multiple sites, enabling lymphocyte subset enumeration in global clinical trials.

REFERENCES

[1]CLSI. Validation of assays performed by flow cytometry. 1st ed. CLSI guideline H62. Clinical Laboratory Standards Institute; 2021.

Send inquiries to *isales@cellcarta.com* with WRIB 2026 in email subject.

Day2-P18

www.cellcarta.com