Efficient tracking of electronic noise in BD LSR Fortessa™ without routine baseline checks.

Dominic Gagnon

Associate Director, Global Flow Cytometry, CellCarta

POSTER NUMBER: P336

CellCarta

INTRODUCTION

Electronic noise affecting flow cytometry data comes from several sources, including external electromagnetic fields emanated from the cytometer electronics and cabling, thermionic noise, photon shot noise, and dynode noise. Electronic noise can be very detrimental to sample analysis when affecting important readouts that are dim and hard to detect. In figure 1, the sensitivity of the detection of TIE2 is reduced by the electronic noise spreading the negative population toward the positive gate.

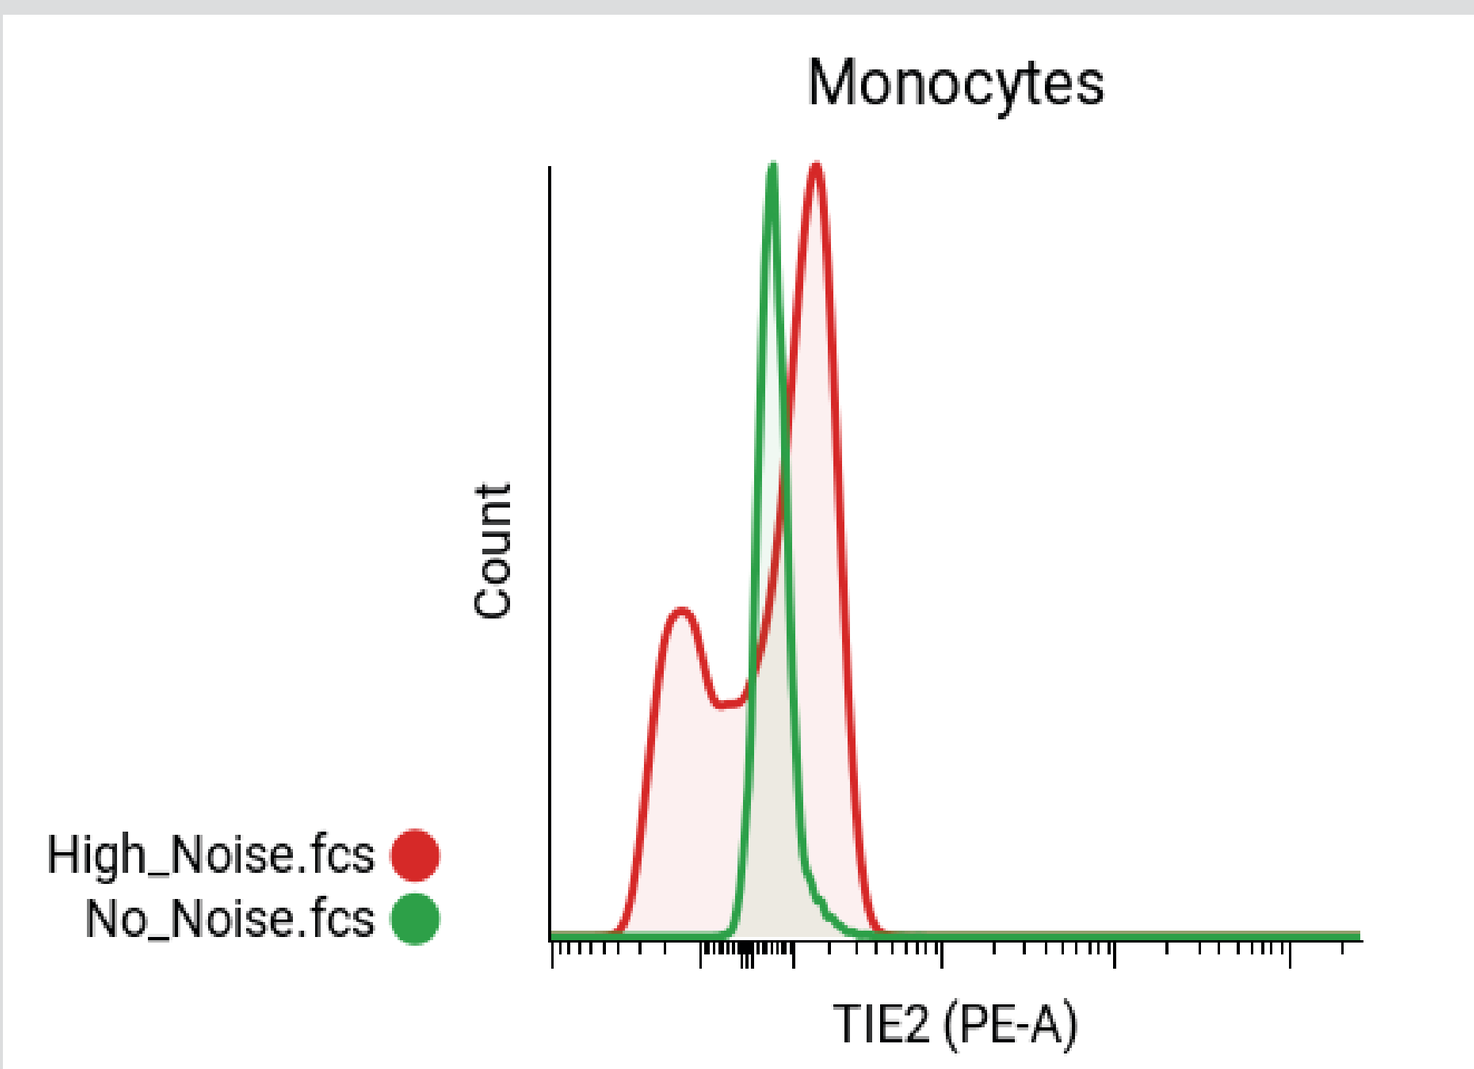

Monocytes
Count
High_Noise.fcs
No_Noise.fcs
TIE2 (PE-A)

Figure 1: Effect of electronic noise on real samples. In green, normal readout without electronic noise. In red, acquisition affected by high electronic noise spreading the negative signal in both directions.

At CellCarta, we have a fleet of 14 BD LSR Fortessa™ running BD FACSDiva™ software version 9, operating 12-16 hours a day, 7 days a week. We maintain them using best practices and they are under service contracts. We aim to minimize downtime and provide the best quality and reproducibility of data through lengthy clinical trials.

That's why we have implemented in the first step, multi-instrument and multi-site alignment using lyophilised beads as demonstrated in our **2024 CYTO poster: Multi- Site Instrument Alignment Using Lyophilised BD™ CompBeads**.

METHODS

In the same spirit of maintaining fully aligned instruments, we have implemented two types of trending. The first one is the daily trending using BD® CS&T beads recorded with a harmonised application settings. MFI from bright beads is tracked over time to detect any drift. (Fig. 2) The second type of trending is running the lyophilised compensation beads (real fluorescence) on all instruments to confirm that settings are maintained. (Fig. 3) We centrally store and visualize all trending data by loading it into CellEngine cytometry analysis software (CellCarta). CellEngine allows us to quickly visualize the data from numerous perspectives, including by date, by instrument, by operator, by site, and by reagent lots.

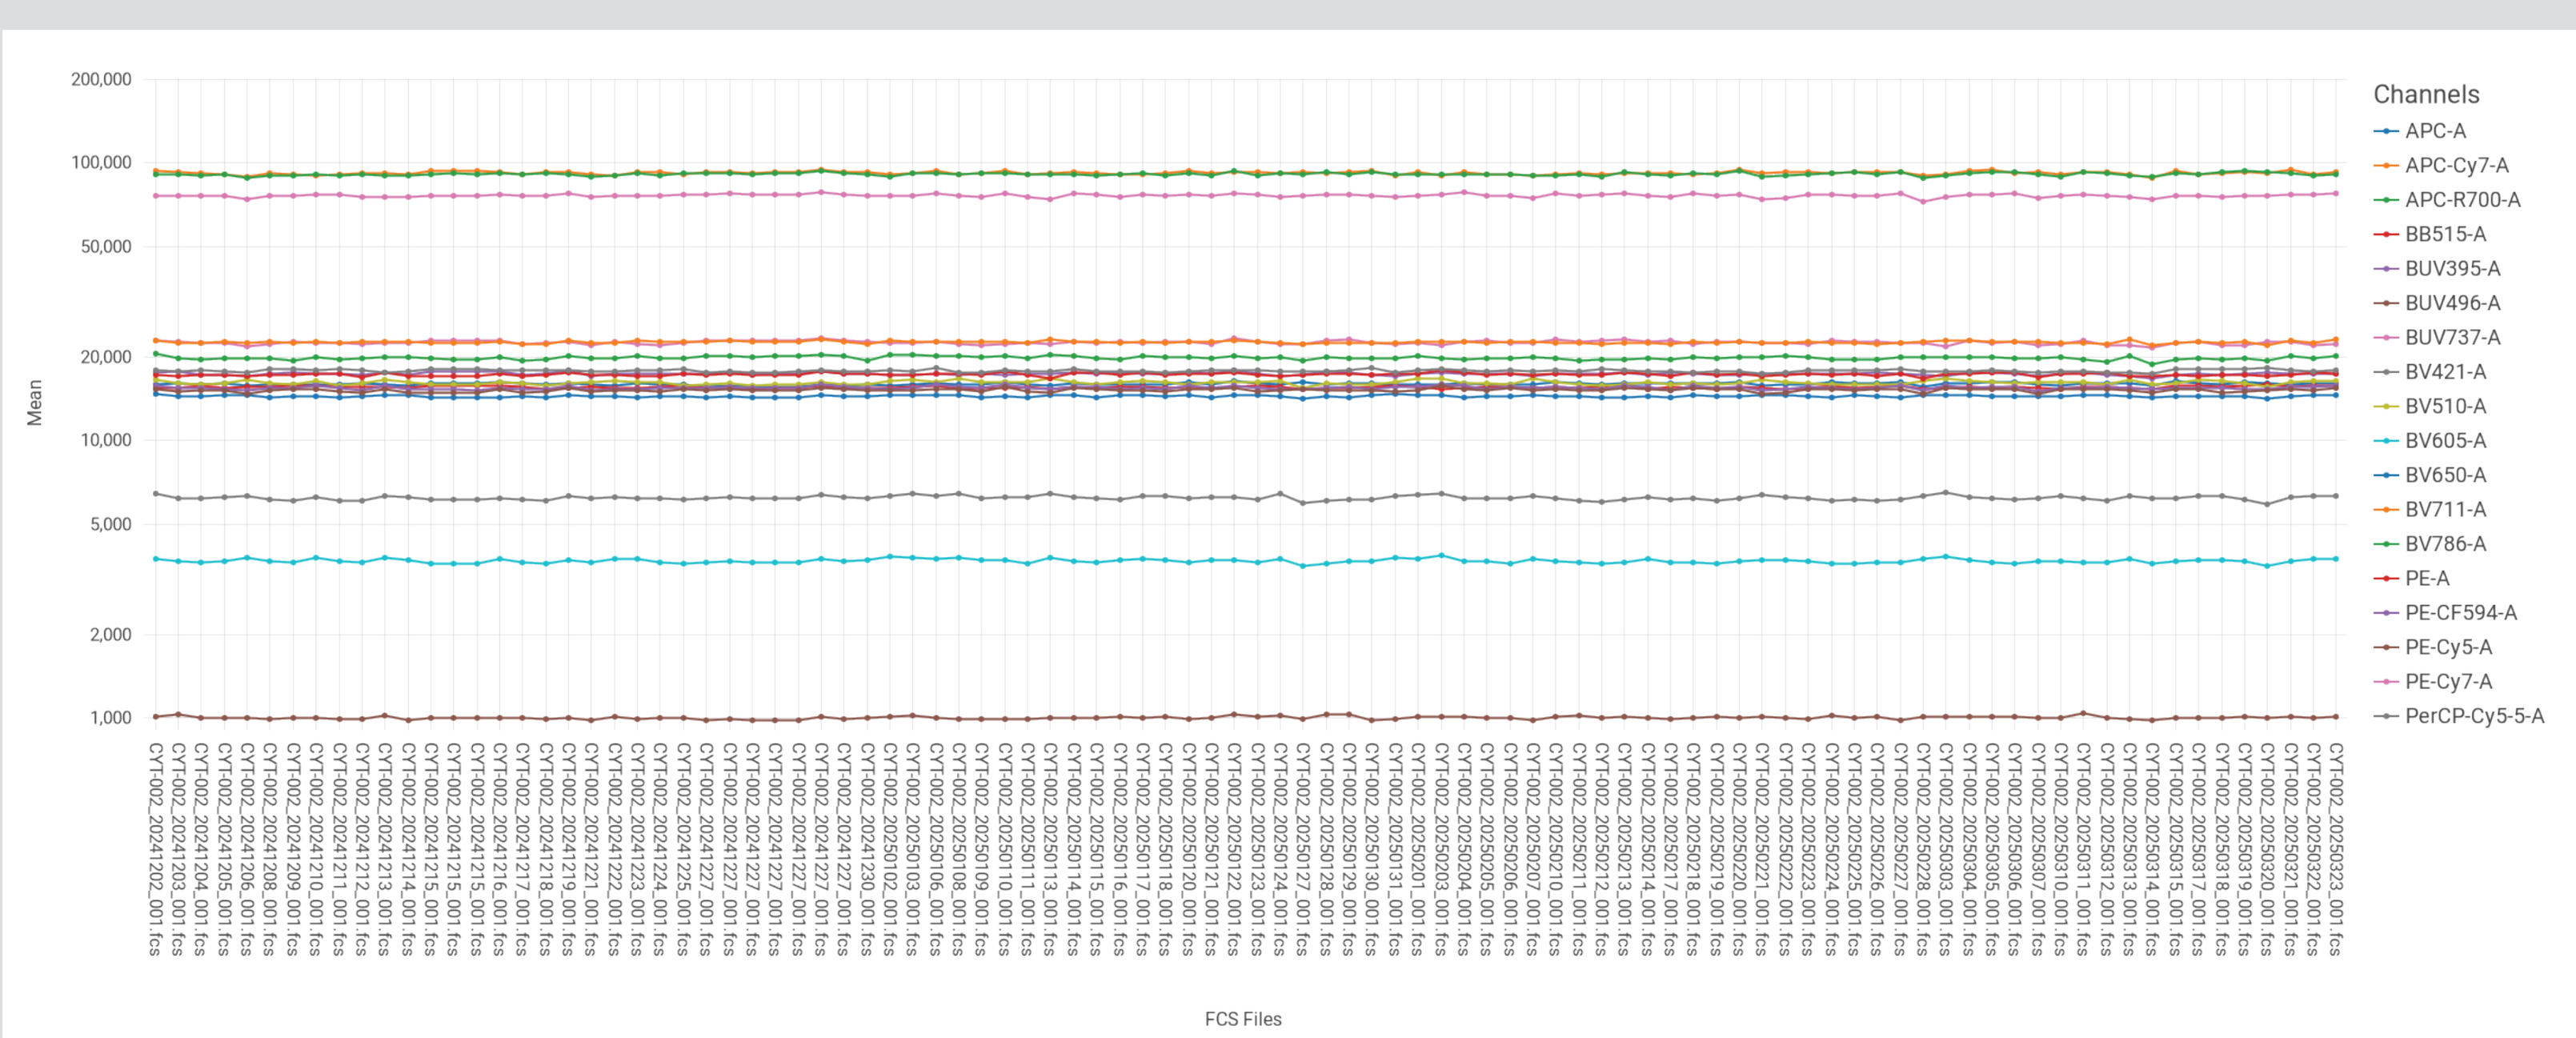

Mean
FCS Files
Channels
APC-A
APC-Cy7-A
APC-R700-A
BB515-A
BUV395-A
BUV496-A
BUV737-A
BV421-A
BV510-A
BV605-A
BV650-A
BV711-A
BV786-A
PE-A
PE-CF594-A
PE-Cy5-A
PE-Cy7-A
PerCP-Cy5-5-A

Figure 2: Trending of the bright peak of BD® CS&T beads recorded daily on one cytometer with a harmonised application settings.

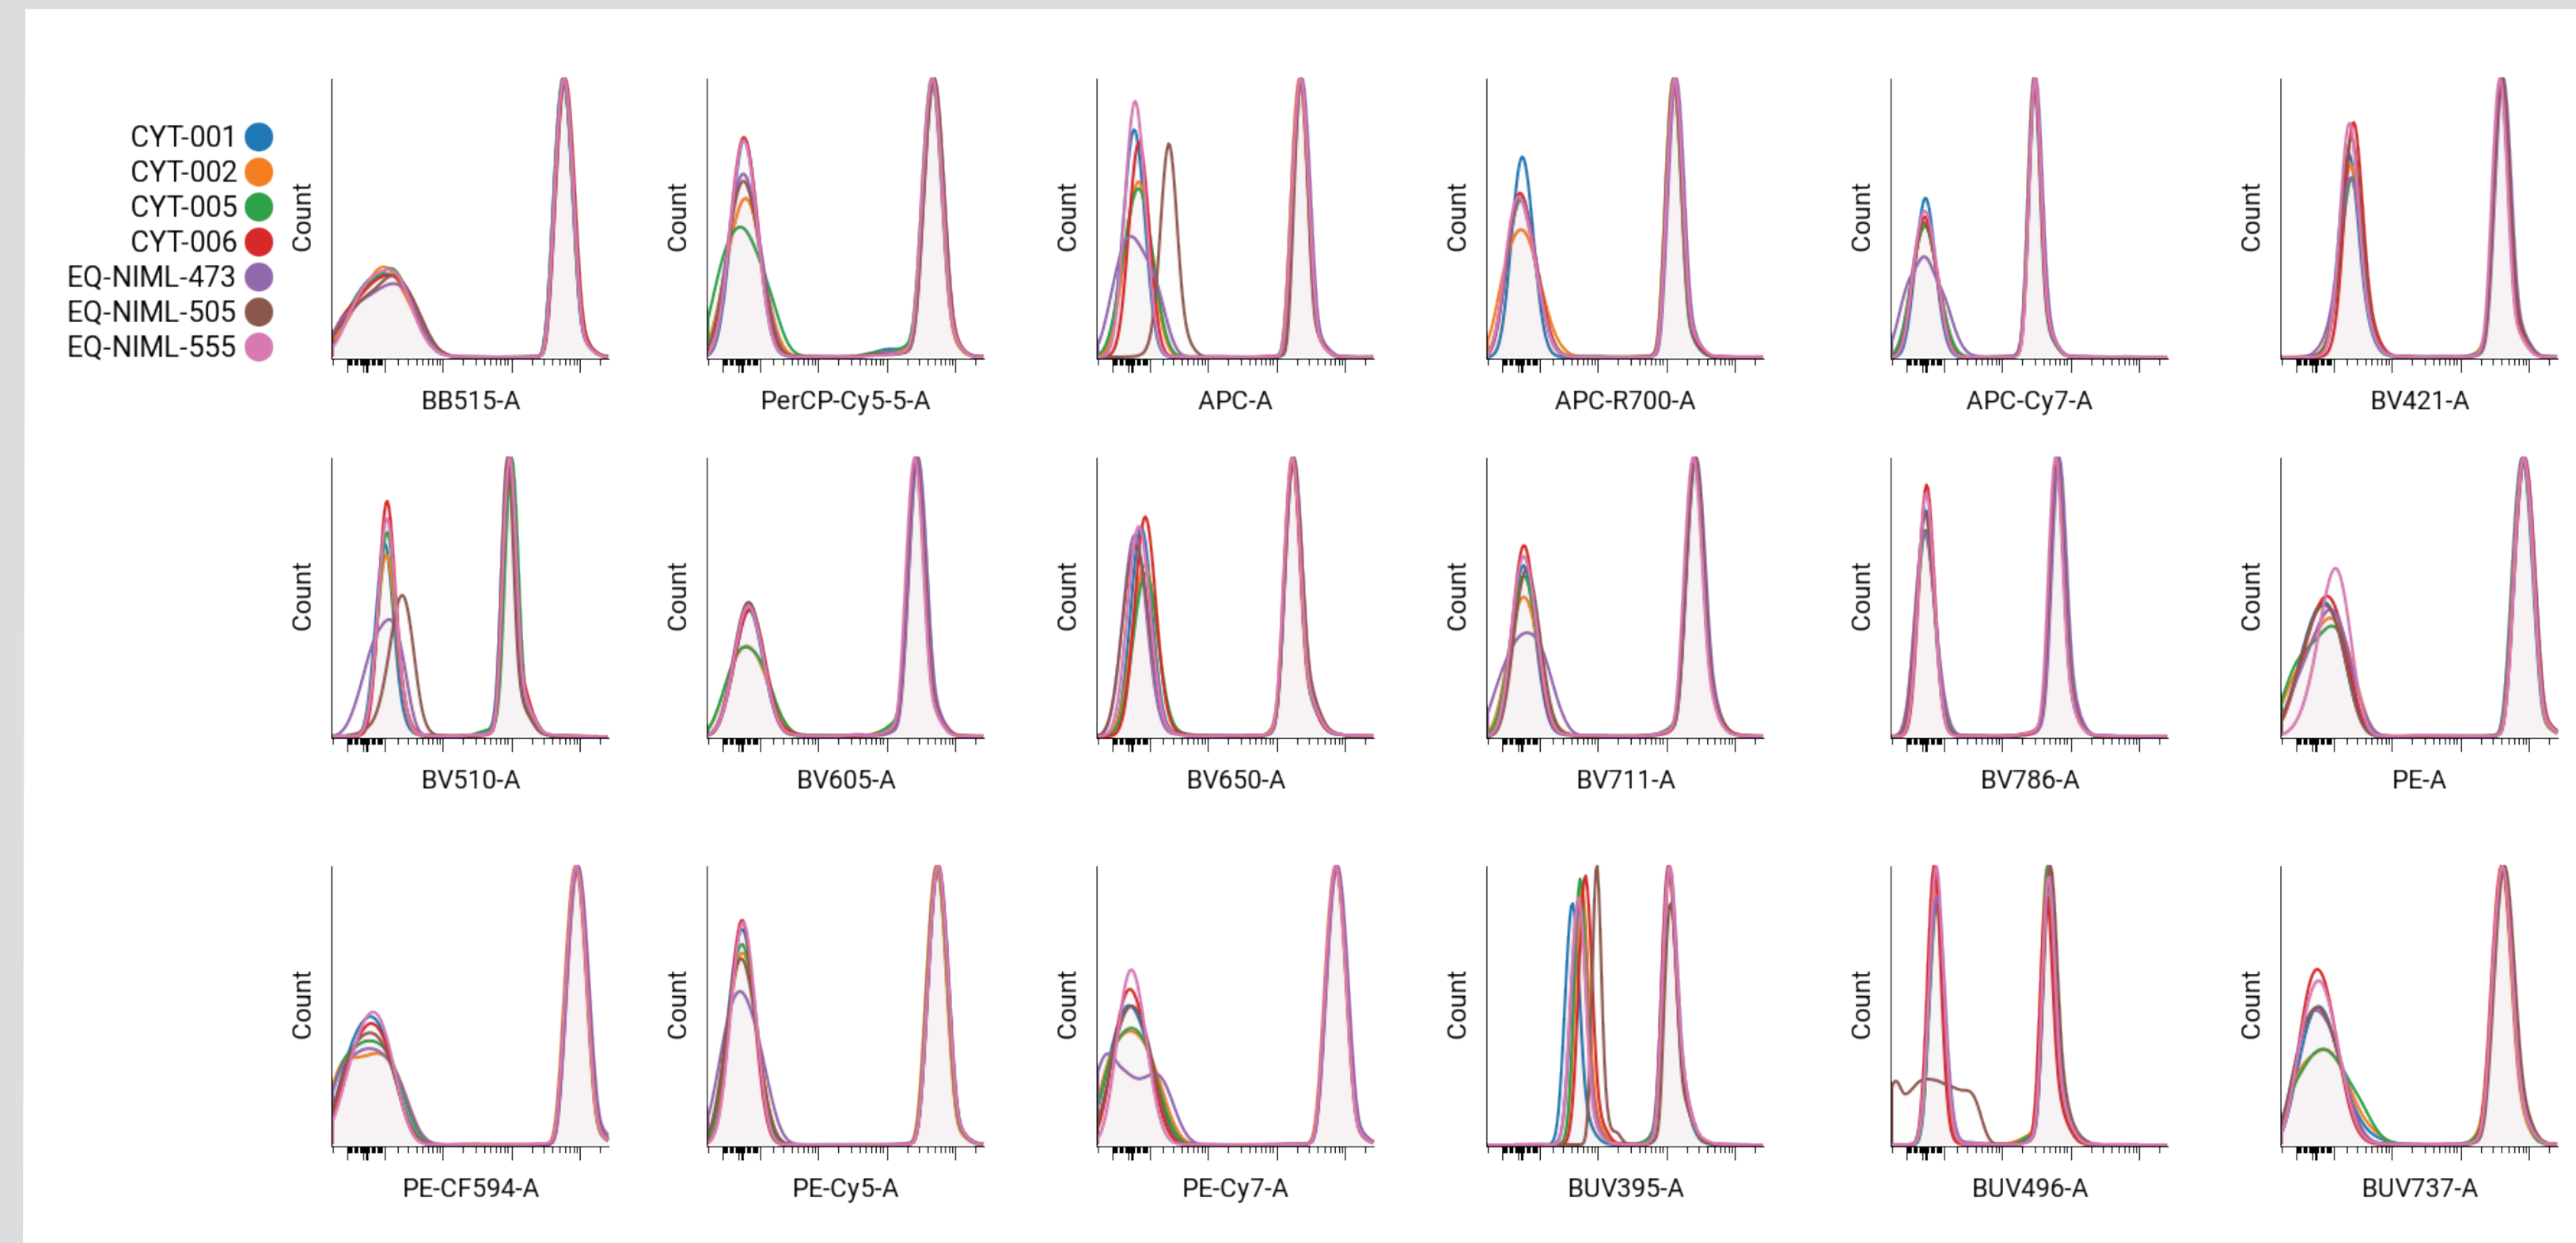

CYT-001
CYT-002
CYT-005
CYT-006
EQ-NIML-473
EQ-NIML-505
EQ-NIML-555
Count
BB515-A
PerCP-Cy5-5-A
APC-A
APC-R700-A
APC-Cy7-A
BV421-A
BV510-A
BV605-A
BV650-A
BV711-A
BV786-A
PE-A
PE-CF594-A
PE-Cy5-A
PE-Cy7-A
BUV395-A
BUV496-A
BUV737-A

Figure 3: Inter-instrument stability. Recording of the 18-color Lyophilised BD™ CompBeads on seven different cytometers using an application settings harmonised 4 months ago.

RESULTS

Running a baseline takes around 45 minutes, which is too long to perform daily or even weekly on all our fleet instruments to detect issue with electronic noise. We needed a solution that was fast and does not require much additional reagents. Thus, we sought to utilize our daily trending of the MFI using BD® CS&T beads.

Visually, electronic noise can be detected in BD FACSDiva software by looking at the recording of the CS&T beads (Fig. 4). This is easy to implement but has several downsides: because it's not quantitative, there's potential for variation between operators, and it's a one-time verification instead of a trending over time.

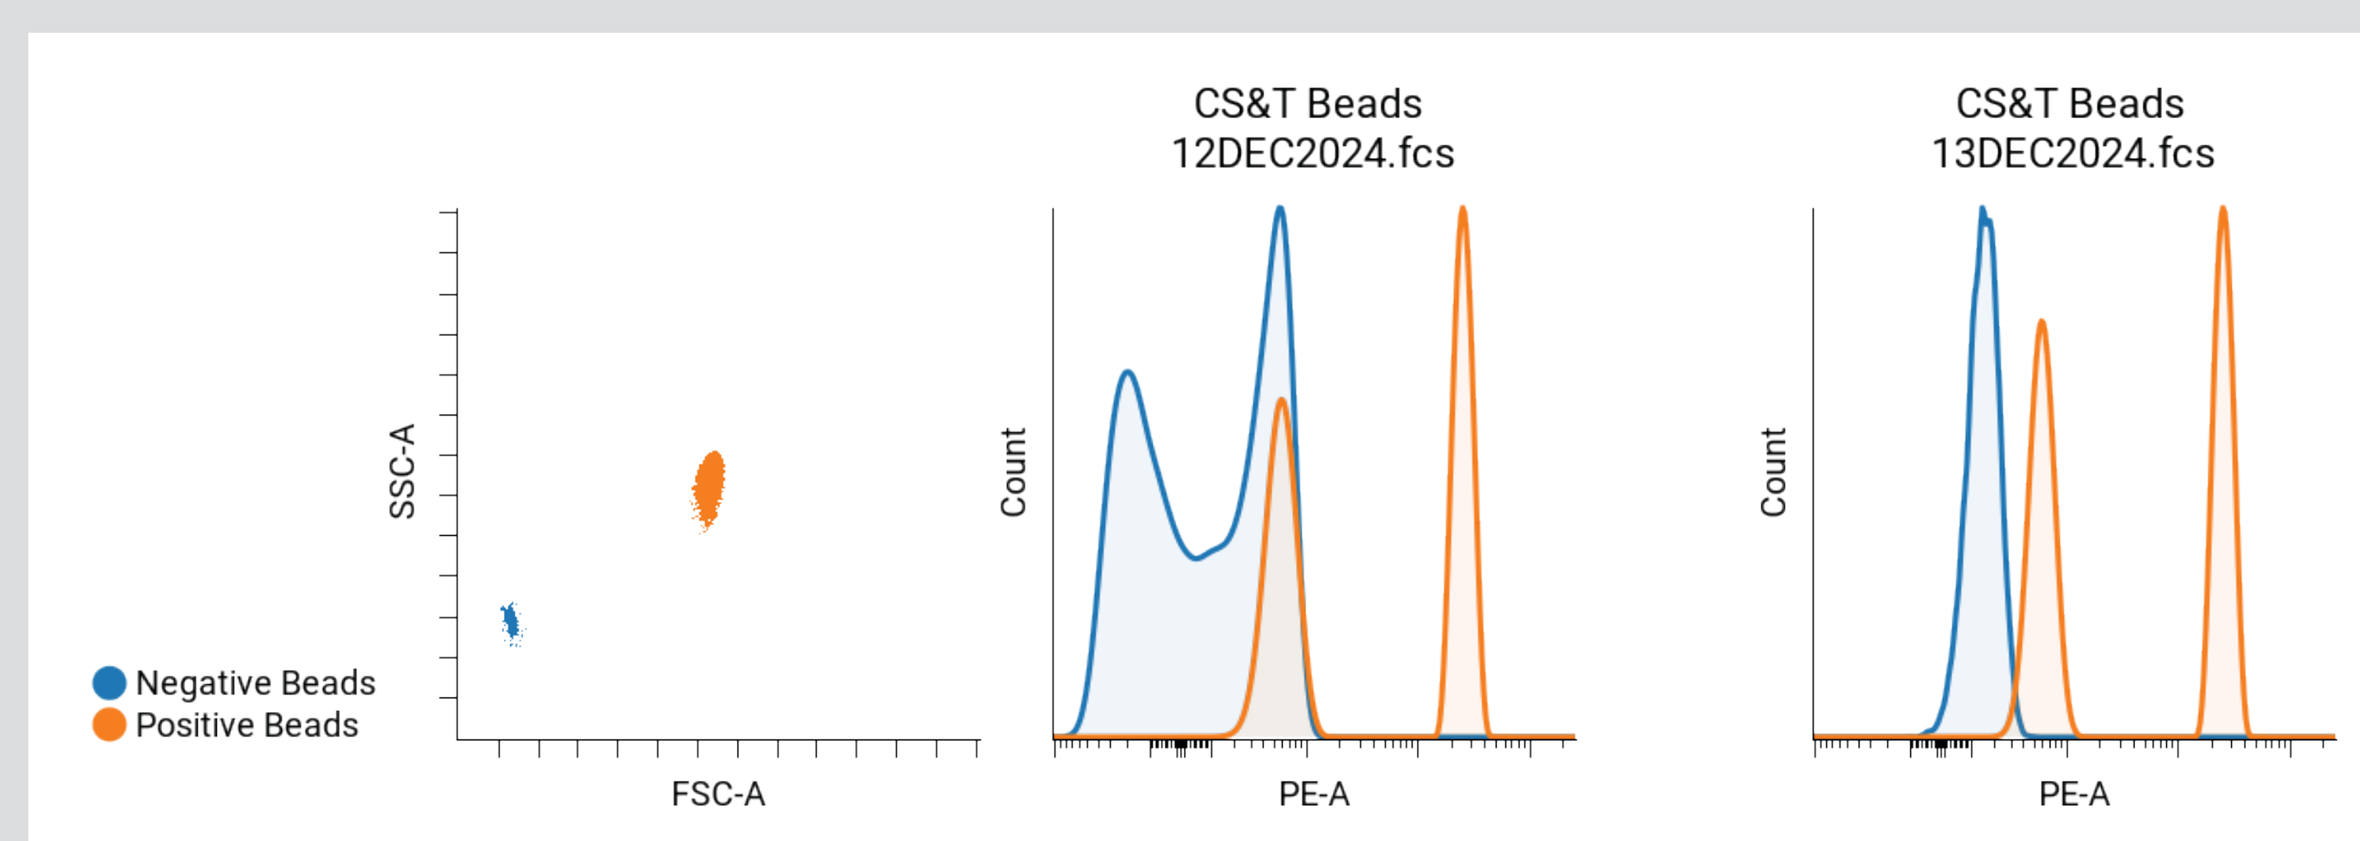

Negative Beads
Positive Beads
SSC-A
FSC-A
CS&T Beads
12DEC2024.fcs
Count
PE-A
CS&T Beads
13DEC2024.fcs
Count
PE-A

Figure 4: Visualisation in CellEngine of the electronic noise on negative BD® CS&T beads (in blue). Negative starts to spread and can even have double peaks (middle figure). After removal of electronic noise, the negative peak is clearly unimodal (right figure) restoring sensitivity.

DISCUSSION

Since we were recording CS&T beads daily and they have a negative bead, we have tried to look at trending those data. Trending the MFI of the negative population did not reliably flag high noise. The best approach that we have found is using the Median Absolute Deviation (MAD). The MAD is a measure of the dispersion (or variation) of the data and correlates with higher electronic noise that spread the negative signal. When we observe clear excursion from the normal trend for each specific parameters per cytometer, it will be further investigated by performing a Baseline QC if deem necessary. While the MAD doesn't provide the electronic noise readouts directly, it performs well as a surrogate for monitoring. If Baseline fails, a service call is made to correct the situation. Figure 5 show a trending without any excursion (almost all our instruments are in this situation). Figure 6 show a know "problematic" instrument with a PE detector excursion (57 of robust Standard Deviation of Electronic Noise (rSDEN)), PE-Cy7 excursion (84 of rSDEN), and initial BV510 excursion (33 rSDEN). The latter was below BD Biosciences threshold for service call (40 rSDEN).

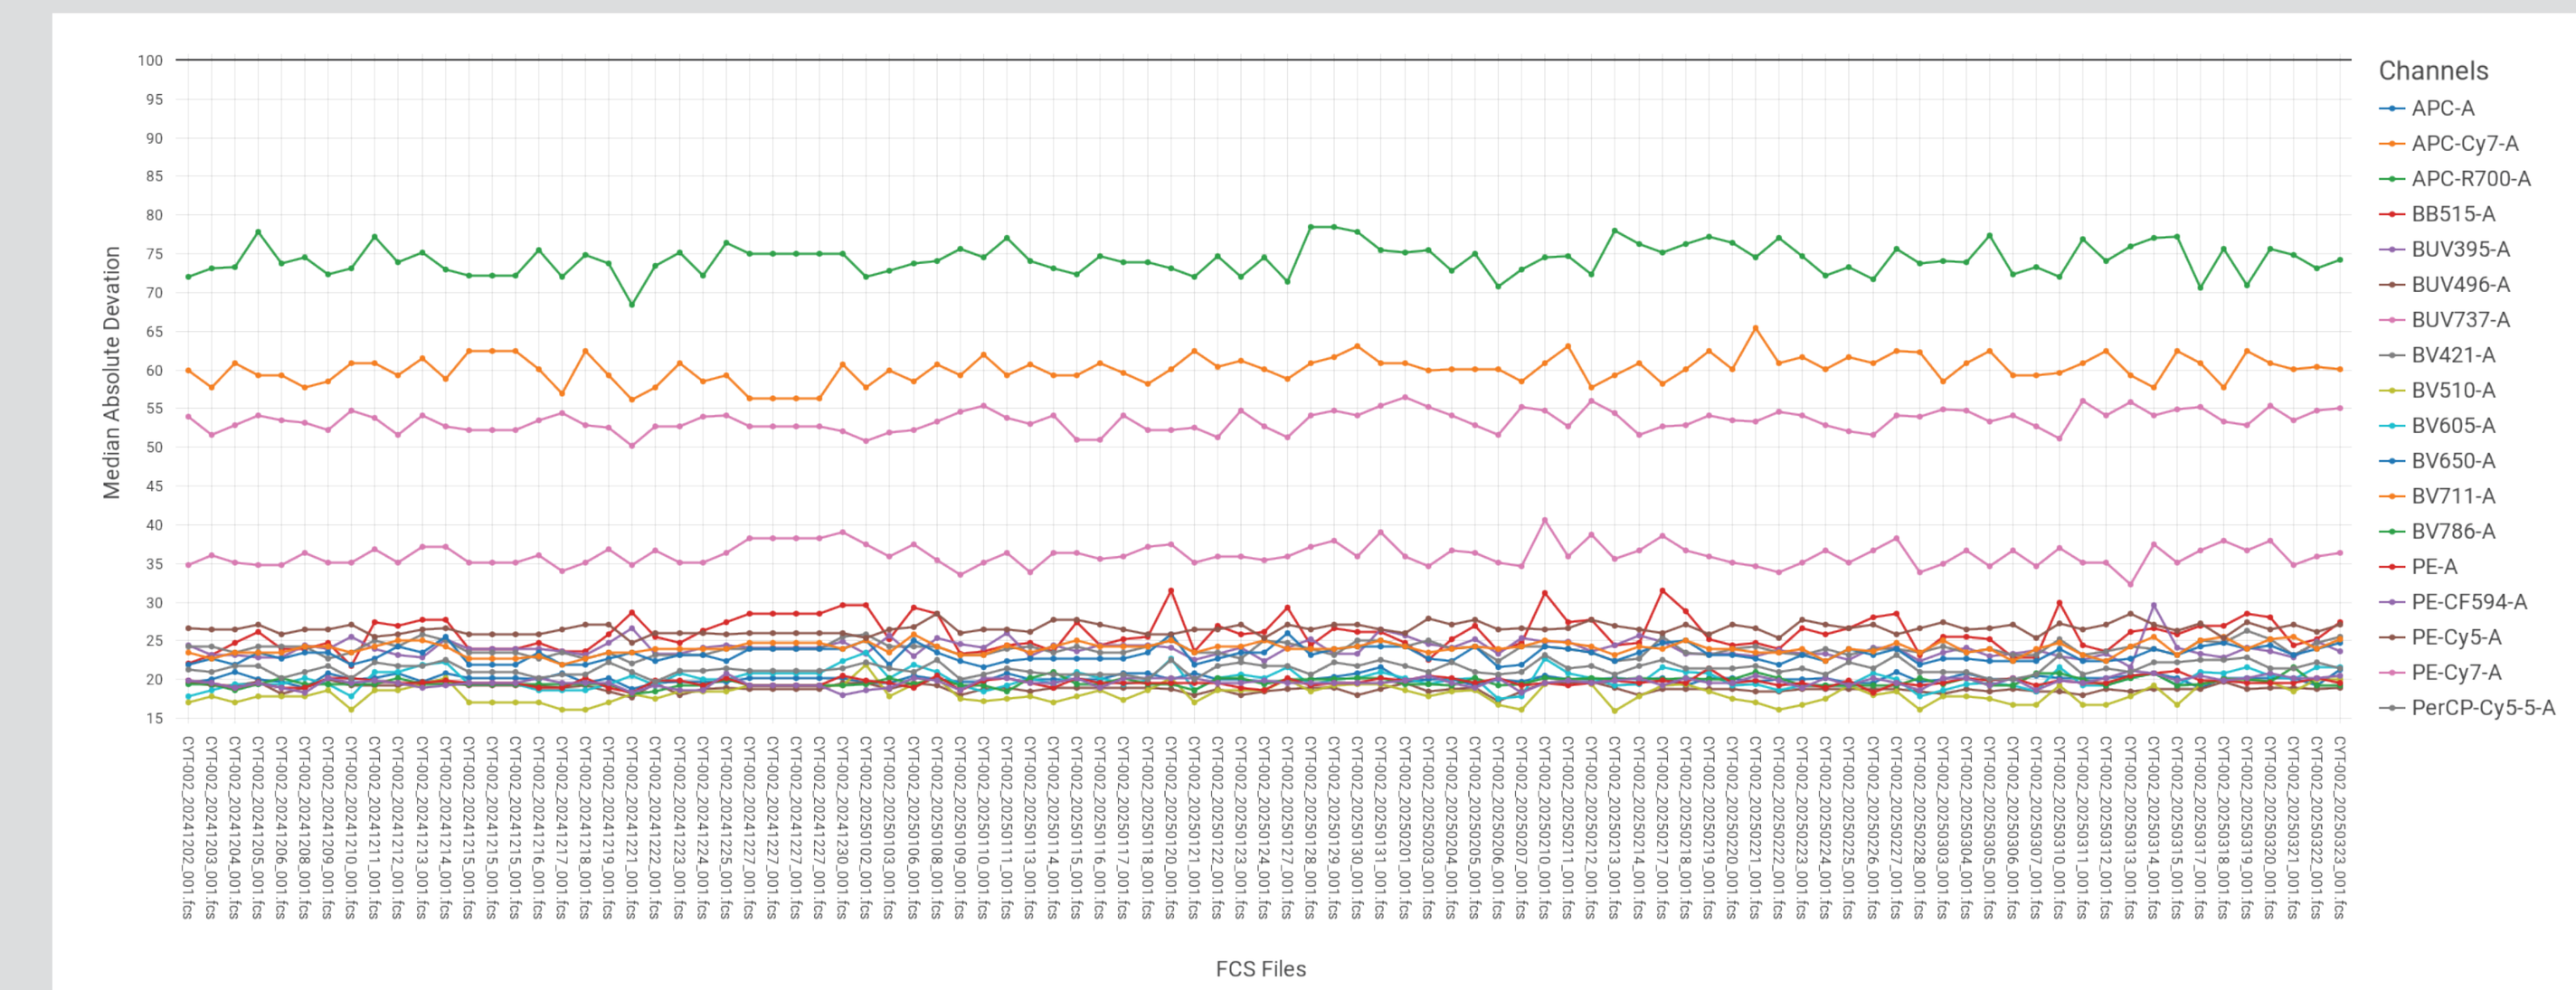

Median Absolute Deviation
FCS Files
Channels
APC-A
APC-Cy7-A
APC-R700-A
BB515-A
BUV395-A
BUV496-A
BUV737-A
BV421-A
BV510-A
BV605-A
BV650-A
BV711-A
BV786-A
PE-A
PE-CF594-A
PE-Cy5-A
PE-Cy7-A
PerCP-Cy5-5-A

Figure 5: Four months trending of the MAD on negative signal of CS&T beads showing no issues.

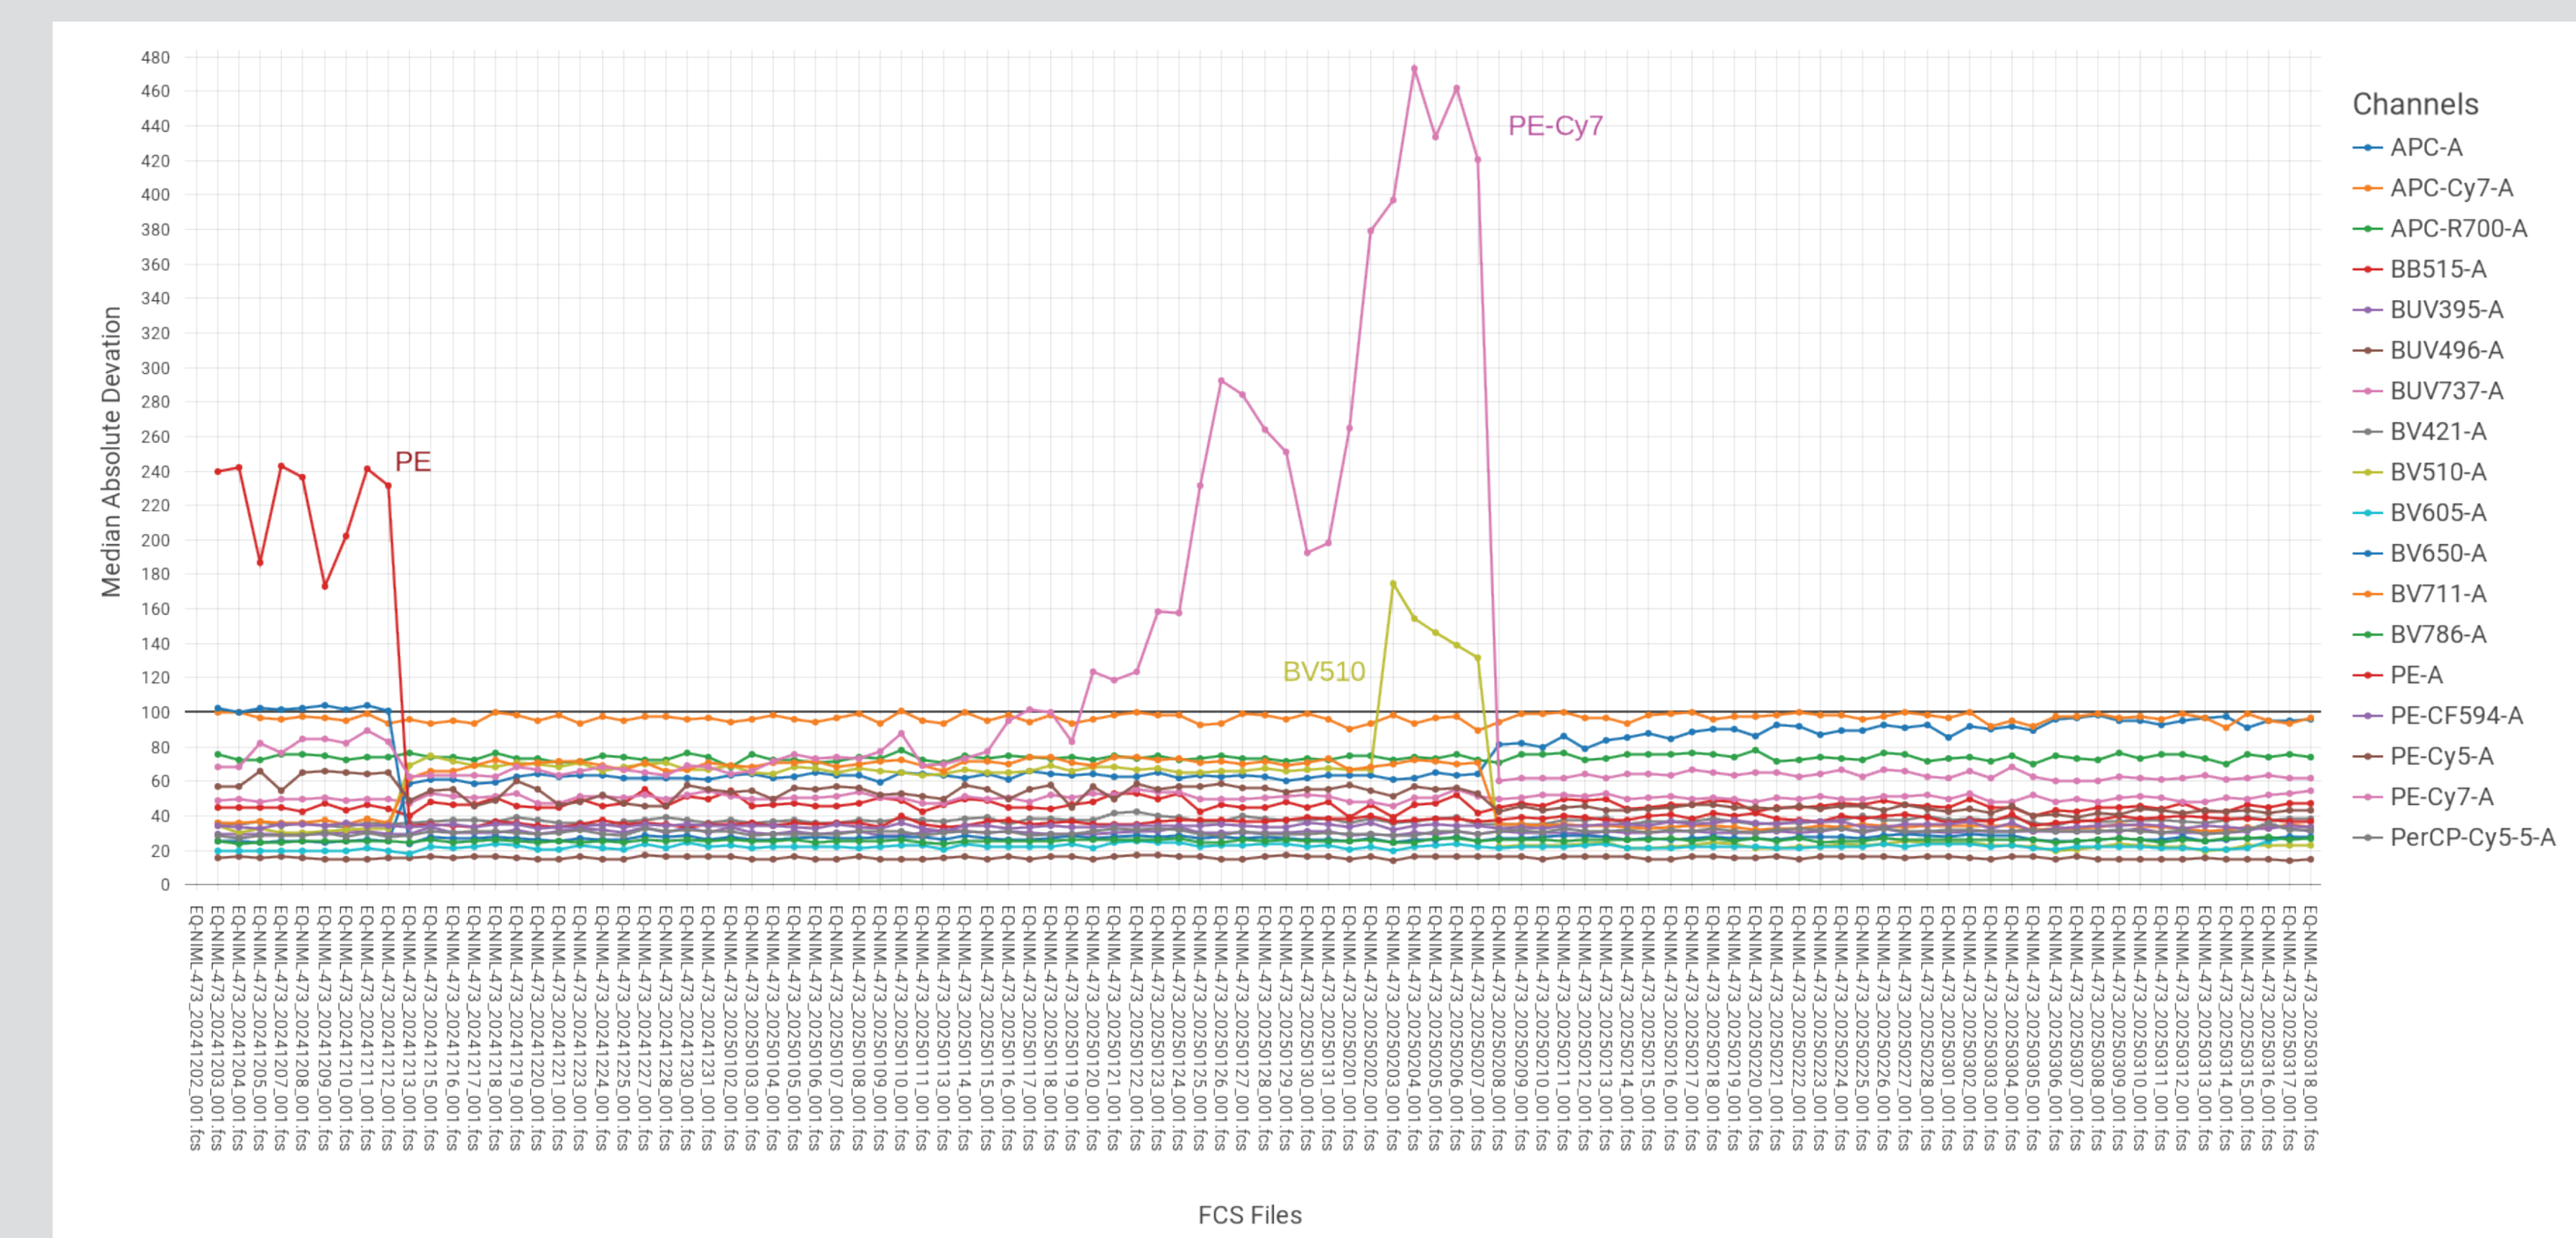

PE-Cy7
PE
BV510
Median Absolute Deviation
FCS Files
Channels
APC-A
APC-Cy7-A
APC-R700-A
BB515-A
BUV395-A
BUV496-A
BUV737-A
BV421-A
BV510-A
BV605-A
BV650-A
BV711-A
BV786-A
PE-A
PE-CF594-A
PE-Cy5-A
PE-Cy7-A
PerCP-Cy5-5-A

Figure 6: Four months trending of the MAD on negative signal of CS&T beads showing high noise for PE (red line, left) and PE-Cy7 (purple line, middle). BV510 was starting to become problematic and was solved at the same time as PE-Cy7 noise (pale green, middle). Noise was back to normal after service call.

CONCLUSION

We have adopted the monitoring of the MAD of the negative peak from CS&T beads as a way to quickly and economically detect high electronic noise across our fleet of cytometers. This serves as a highly sensitive initial signal that can trigger a more intensive baseline procedure and potentially a service call. We centrally collect and visualize all trending data in CellEngine so that we can detect drift over time, as well as find issues that correlate with instrument, operator, reagent lot and/or laboratory site. This enables CellCarta to provide high-quality, high-consistency data, even during multi-year and multi-site clinical trials.

Please send inquiries to
Dominic Gagnon
Associate Director, Global Flow Cytometry, CellCarta
dominic.gagnon@cellcarta.com

P336 www.cellcarta.com