Beyond Affinity: Simplifying ADC quantitation without compromising sensitivity.

Ons Ousji, Mark Watson, Gwenael Pottiez, Luca Genovesi
CellCarta, Montreal, Quebec, Canada

POSTER NUMBER: DAY3-P14

INTRODUCTION

- Antibody–drug conjugates (ADCs) consist of a biologically active payload covalently bound to a monoclonal antibody via a chemical linker. By merging the specific binding properties of antibodies to the potency of the payload, ADCs are designed to selectively deliver cell-killing agents while limiting systemic toxicity[1].
- Ligand-Binding Assays (LBA) and Hybrid Ligand-Binding Assay–Mass Spectrometry (LBA-LC/MS) are two prominent techniques for analyzing antibody-drug conjugates (ADCs). LBAs, particularly ELISA, offer high sensitivity and a broad quantification range without extraction steps. LBA/LC-MS combines the strengths of both technologies, providing sensitive quantitative bioanalysis with multiple analyte detection and high specificity within a single sample thanks to the physical detection of one or multiple surrogate peptides. Both technologies can utilize various capture reagents, including generic anti-human antibodies or more specific anti-idiotype antibodies, for total antibody quantification. However, since both methods rely on capture reagents they can be negatively affected by their availability (too early into the development stage), lack of access to the epitope with the payload of ADCs hindering capture, and potential cross-reactivity with pre-clinical IgGs.
- To address these challenges, we describe a mass spectrometry-based workflow for the quantitation of ADCs using a direct digestion method, implemented with an automated spiking procedure via the Bravo AssayMap. By eliminating affinity capture steps, this simplified protocol offers a sensitive and straightforward approach that is robust, scalable, and easy to implement, ensuring reliable quantification of ADCs across a broad dynamic range without compromising performances.

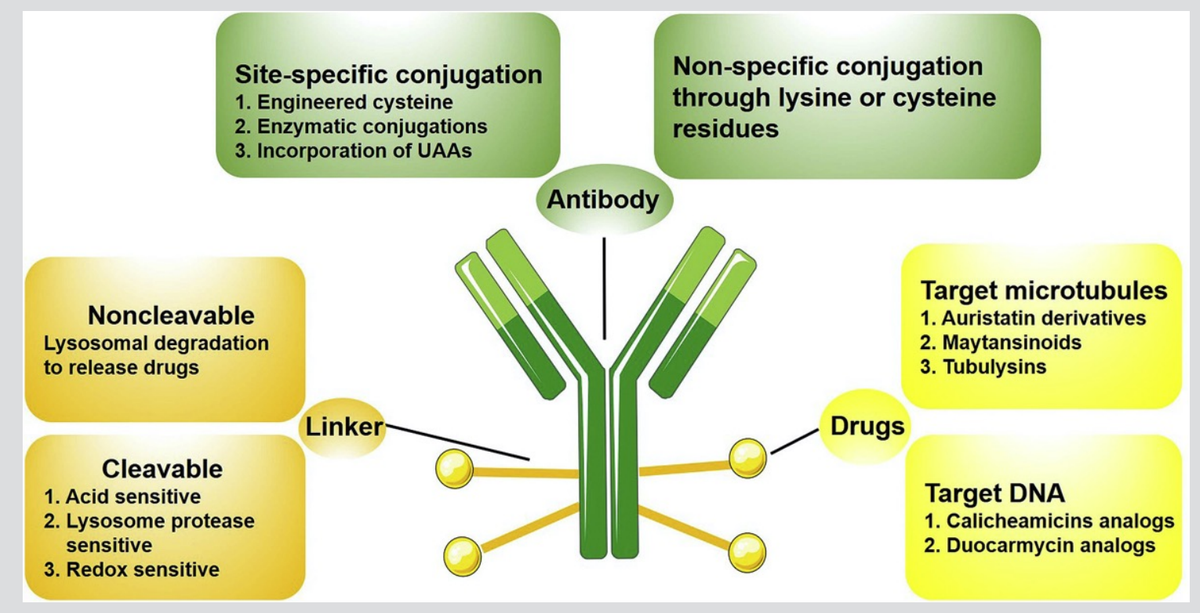
Site-specific conjugation
1. Engineered cysteine
2. Enzymatic conjugations
3. Incorporation of UAAs
Non-specific conjugation through lysine or cysteine residues
Antibody
Noncleavable
Lysosomal degradation to release drugs
Cleavable
1. Acid sensitive
2. Lysosome protease sensitive
3. Redox sensitive
Linker
Drugs
Target microtubules
1. Auristatin derivatives
2. Maytansinoids
3. Tubulysins
Target DNA
1. Calicheamicins analogs
2. Duocarmycin analogs

Figure 1. ADC Building Blocks[2].

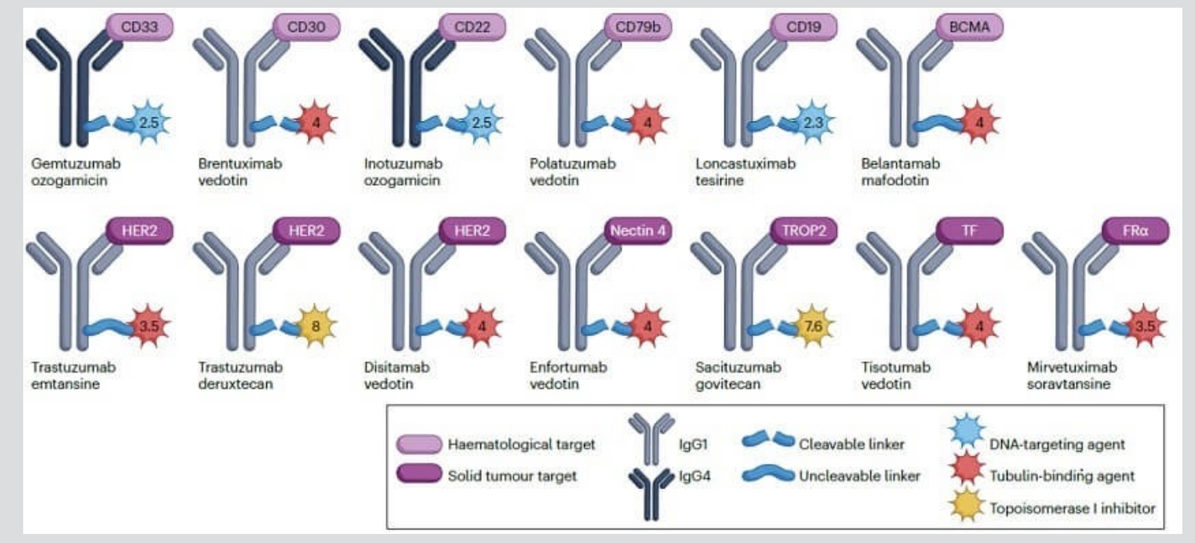

Figure 2. FDA Approved ADC[3].

AFFINITY-FREE METHOD DEVELOPMENT

The initial goal of this study was to find the limits of the direct digestion approach by using a common workflow that eliminates affinity purification while maintaining sensitivity comparable to generic or specific affinity capture methods. Initial efforts were made to reduce the amount of plasma used compared to standard affinity protocol, with sub 10µL samples, while trying to keep the process on a 1-day turnaround time, if possible.

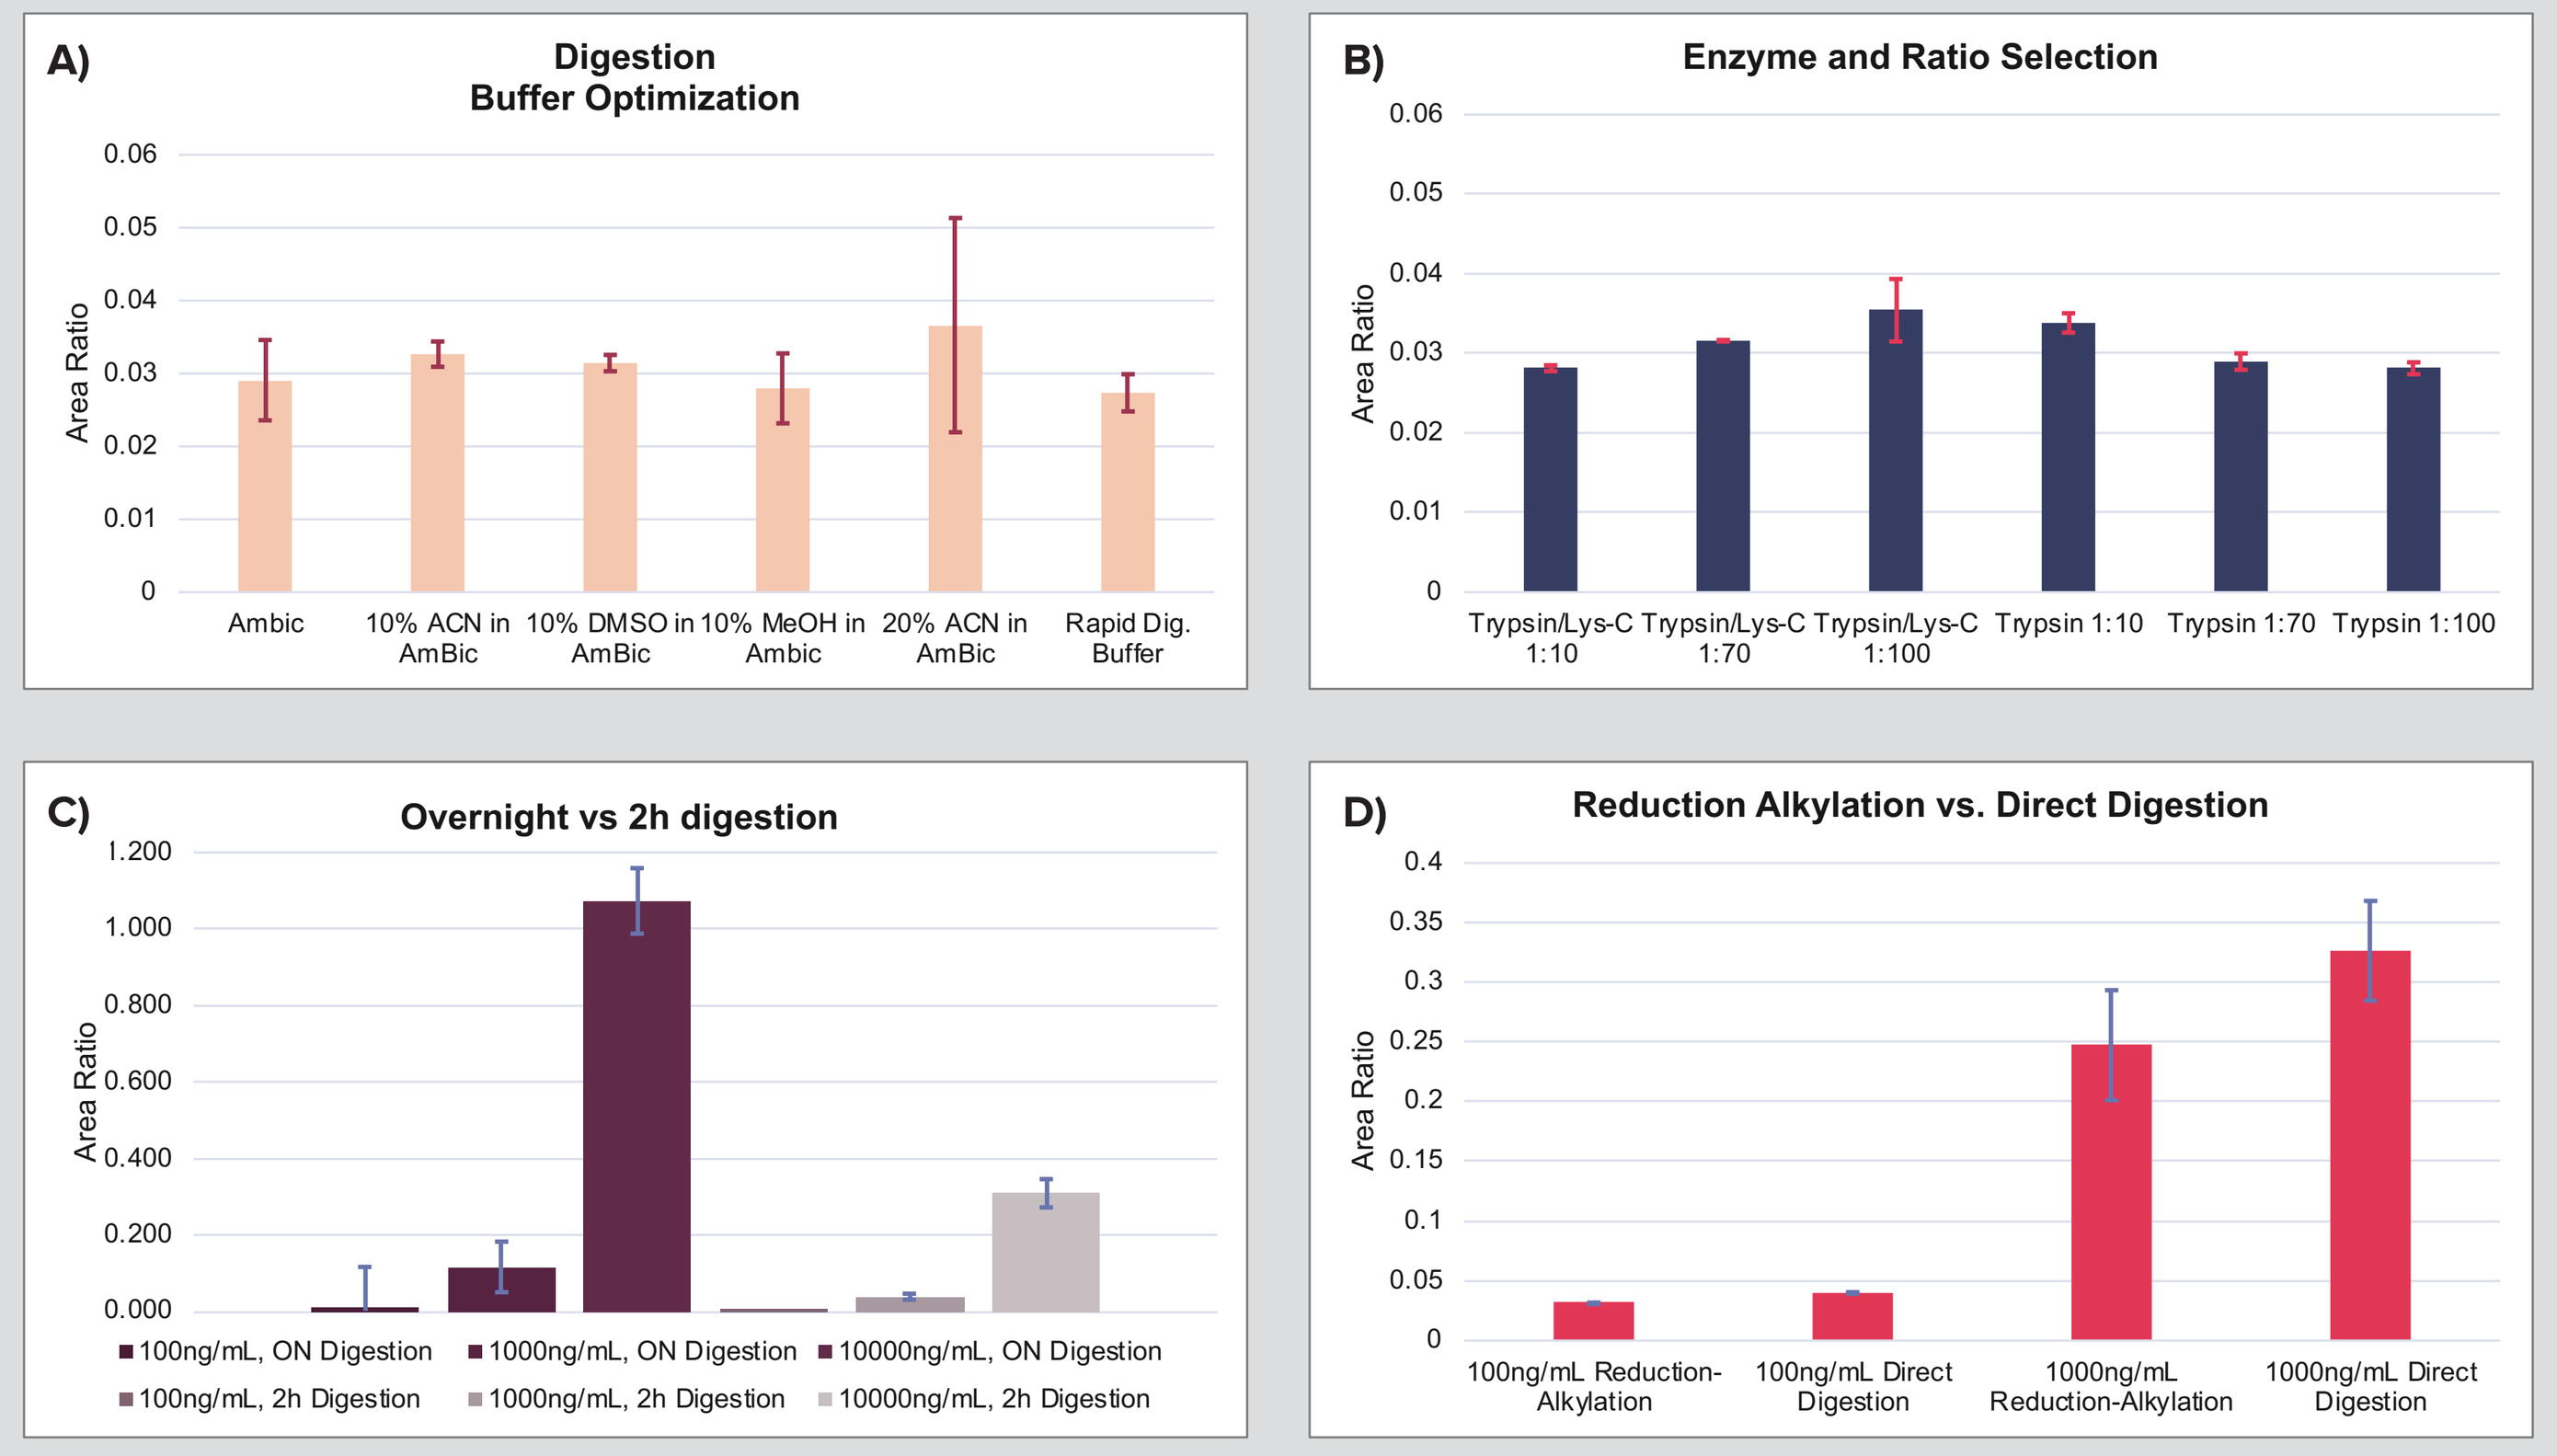
A)
Digestion Buffer Optimization
Area Ratio
Ambic
10% ACN in AmBic
10% DMSO in AmBic
10% MeOH in Ambic
20% ACN in AmBic
Rapid Dig. Buffer
B)
Enzyme and Ratio Selection
Trypsin/Lys-C 1:10
Trypsin/Lys-C 1:70
Trypsin/Lys-C 1:100
Trypsin 1:10
Trypsin 1:70
Trypsin 1:100
C)
Overnight vs 2h digestion
100ng/mL, ON Digestion
1000ng/mL, ON Digestion
10000ng/mL, ON Digestion
100ng/mL, 2h Digestion
1000ng/mL, 2h Digestion
10000ng/mL, 2h Digestion
D)
Reduction Alkylation vs. Direct Digestion
100ng/mL Reduction-Alkylation
100ng/mL Direct Digestion
1000ng/mL Reduction-Alkylation
1000ng/mL Direct Digestion

Figure 3. A) Digestion buffer optimization; **B)** Enzyme and ratio selection; **C)** Digestion time optimization; **D)** Reduction and Alkylation vs. Direct digestion. ON: Overnight.

To ensure that current performance matches or surpasses the standard set by hybrid methods, we aimed for an LLOQ of 100 ng/mL or lower while maximizing the analytical range to cover early time points without requiring sample dilution. For this reason, the initial method was tested between 0.1 and 500 µg/mL, starting with 5 µL of plasma. The use of a C4 column in the LC/MS method ensured minimal carryover issues.

EARLY PERFORMANCE EVALUATION

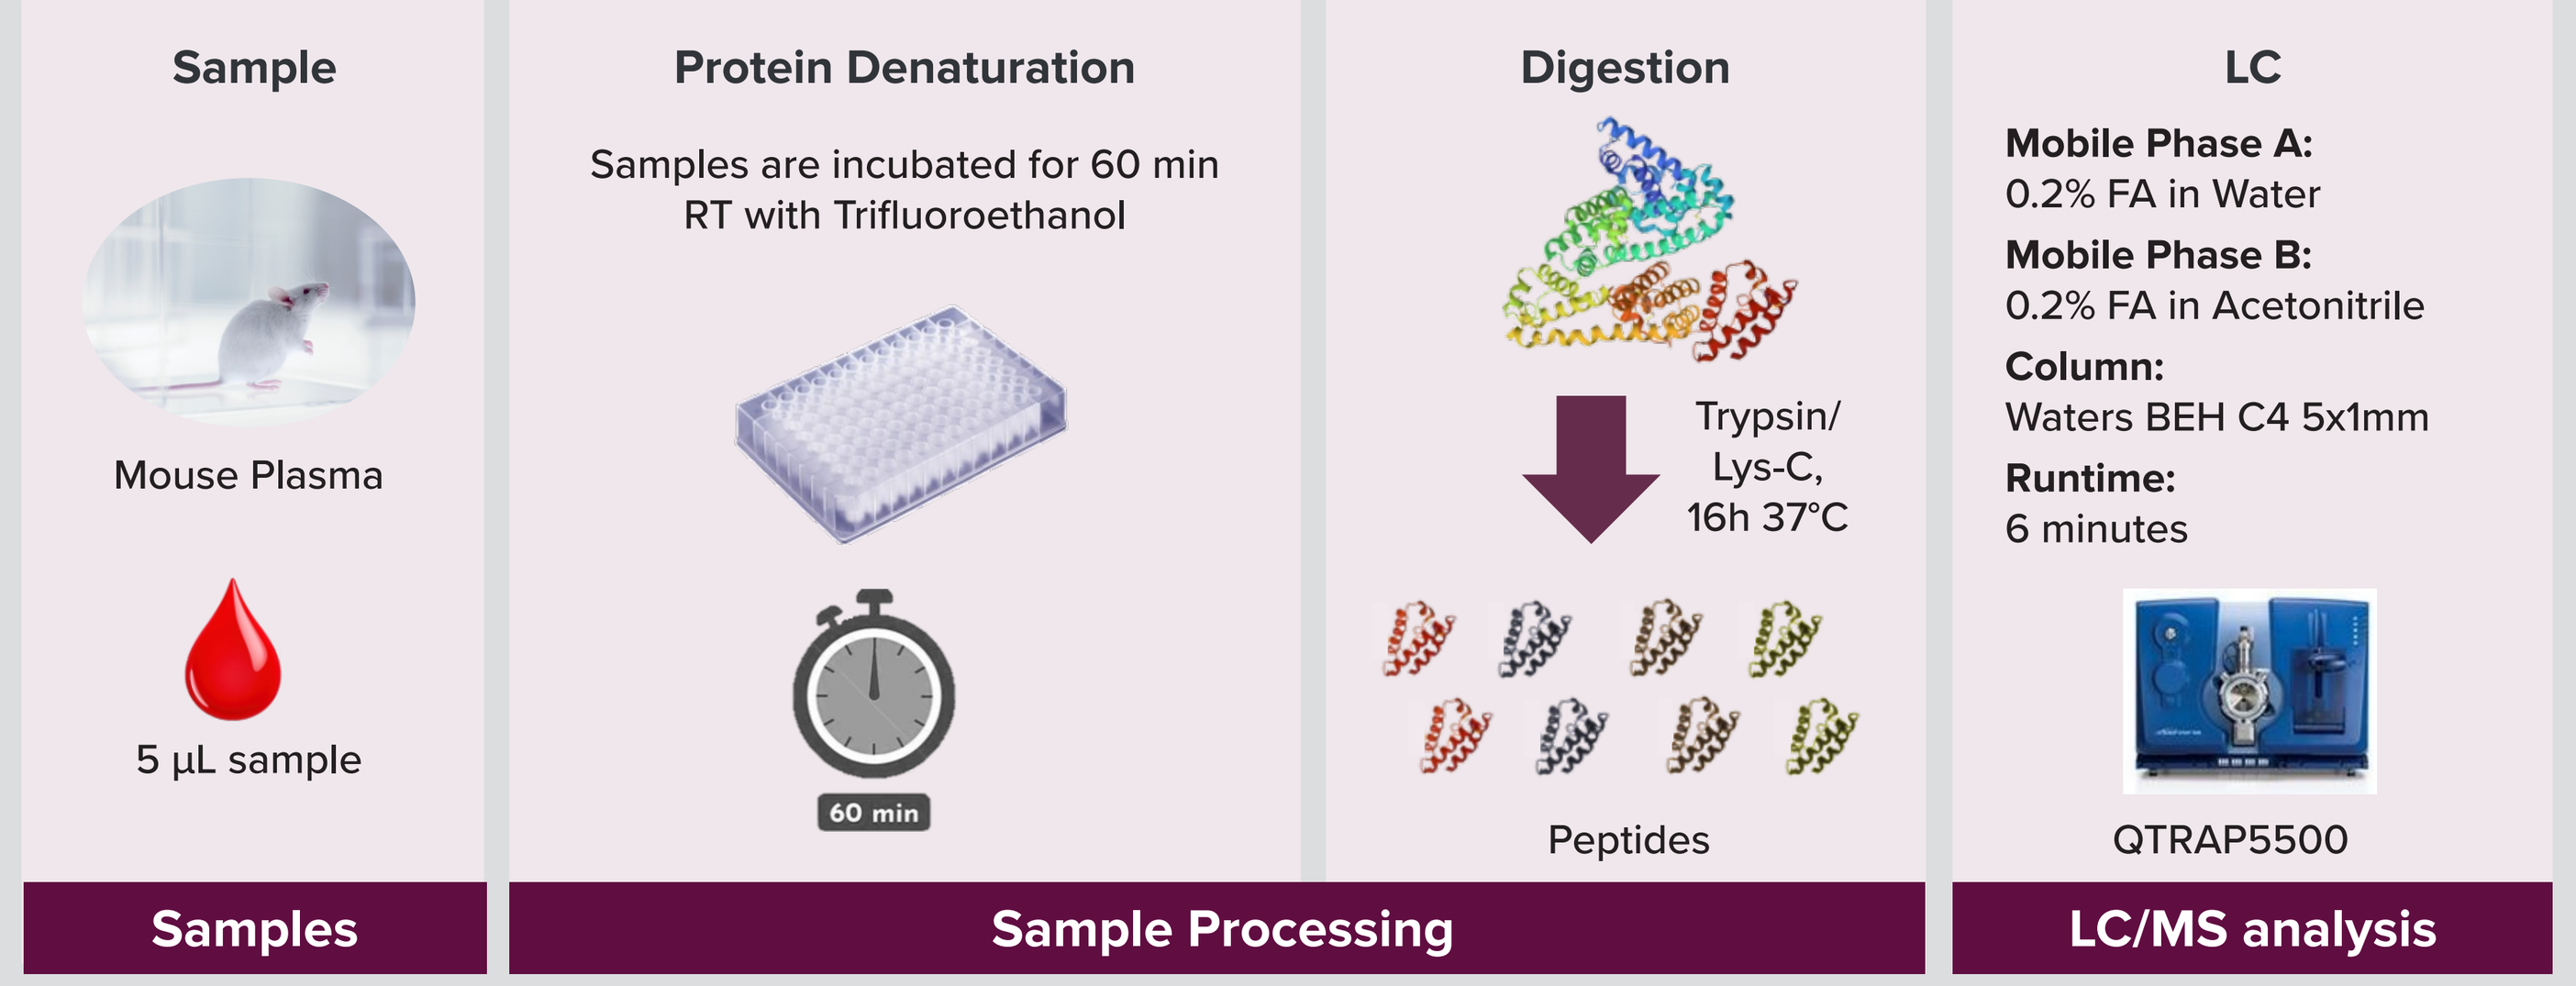
Sample
Mouse Plasma
5 µL sample
Protein Denaturation
Samples are incubated for 60 min RT with Trifluoroethanol
60 min
Digestion
Trypsin/ Lys-C, 16h 37°C
Peptides
LC
Mobile Phase A:
0.2% FA in Water
Mobile Phase B:
0.2% FA in Acetonitrile
Column:
Waters BEH C4 5x1mm
Runtime:
6 minutes
QTRAP5500
Samples
Sample Processing
LC/MS analysis

Figure 4. Optimized Analytical Workflow.

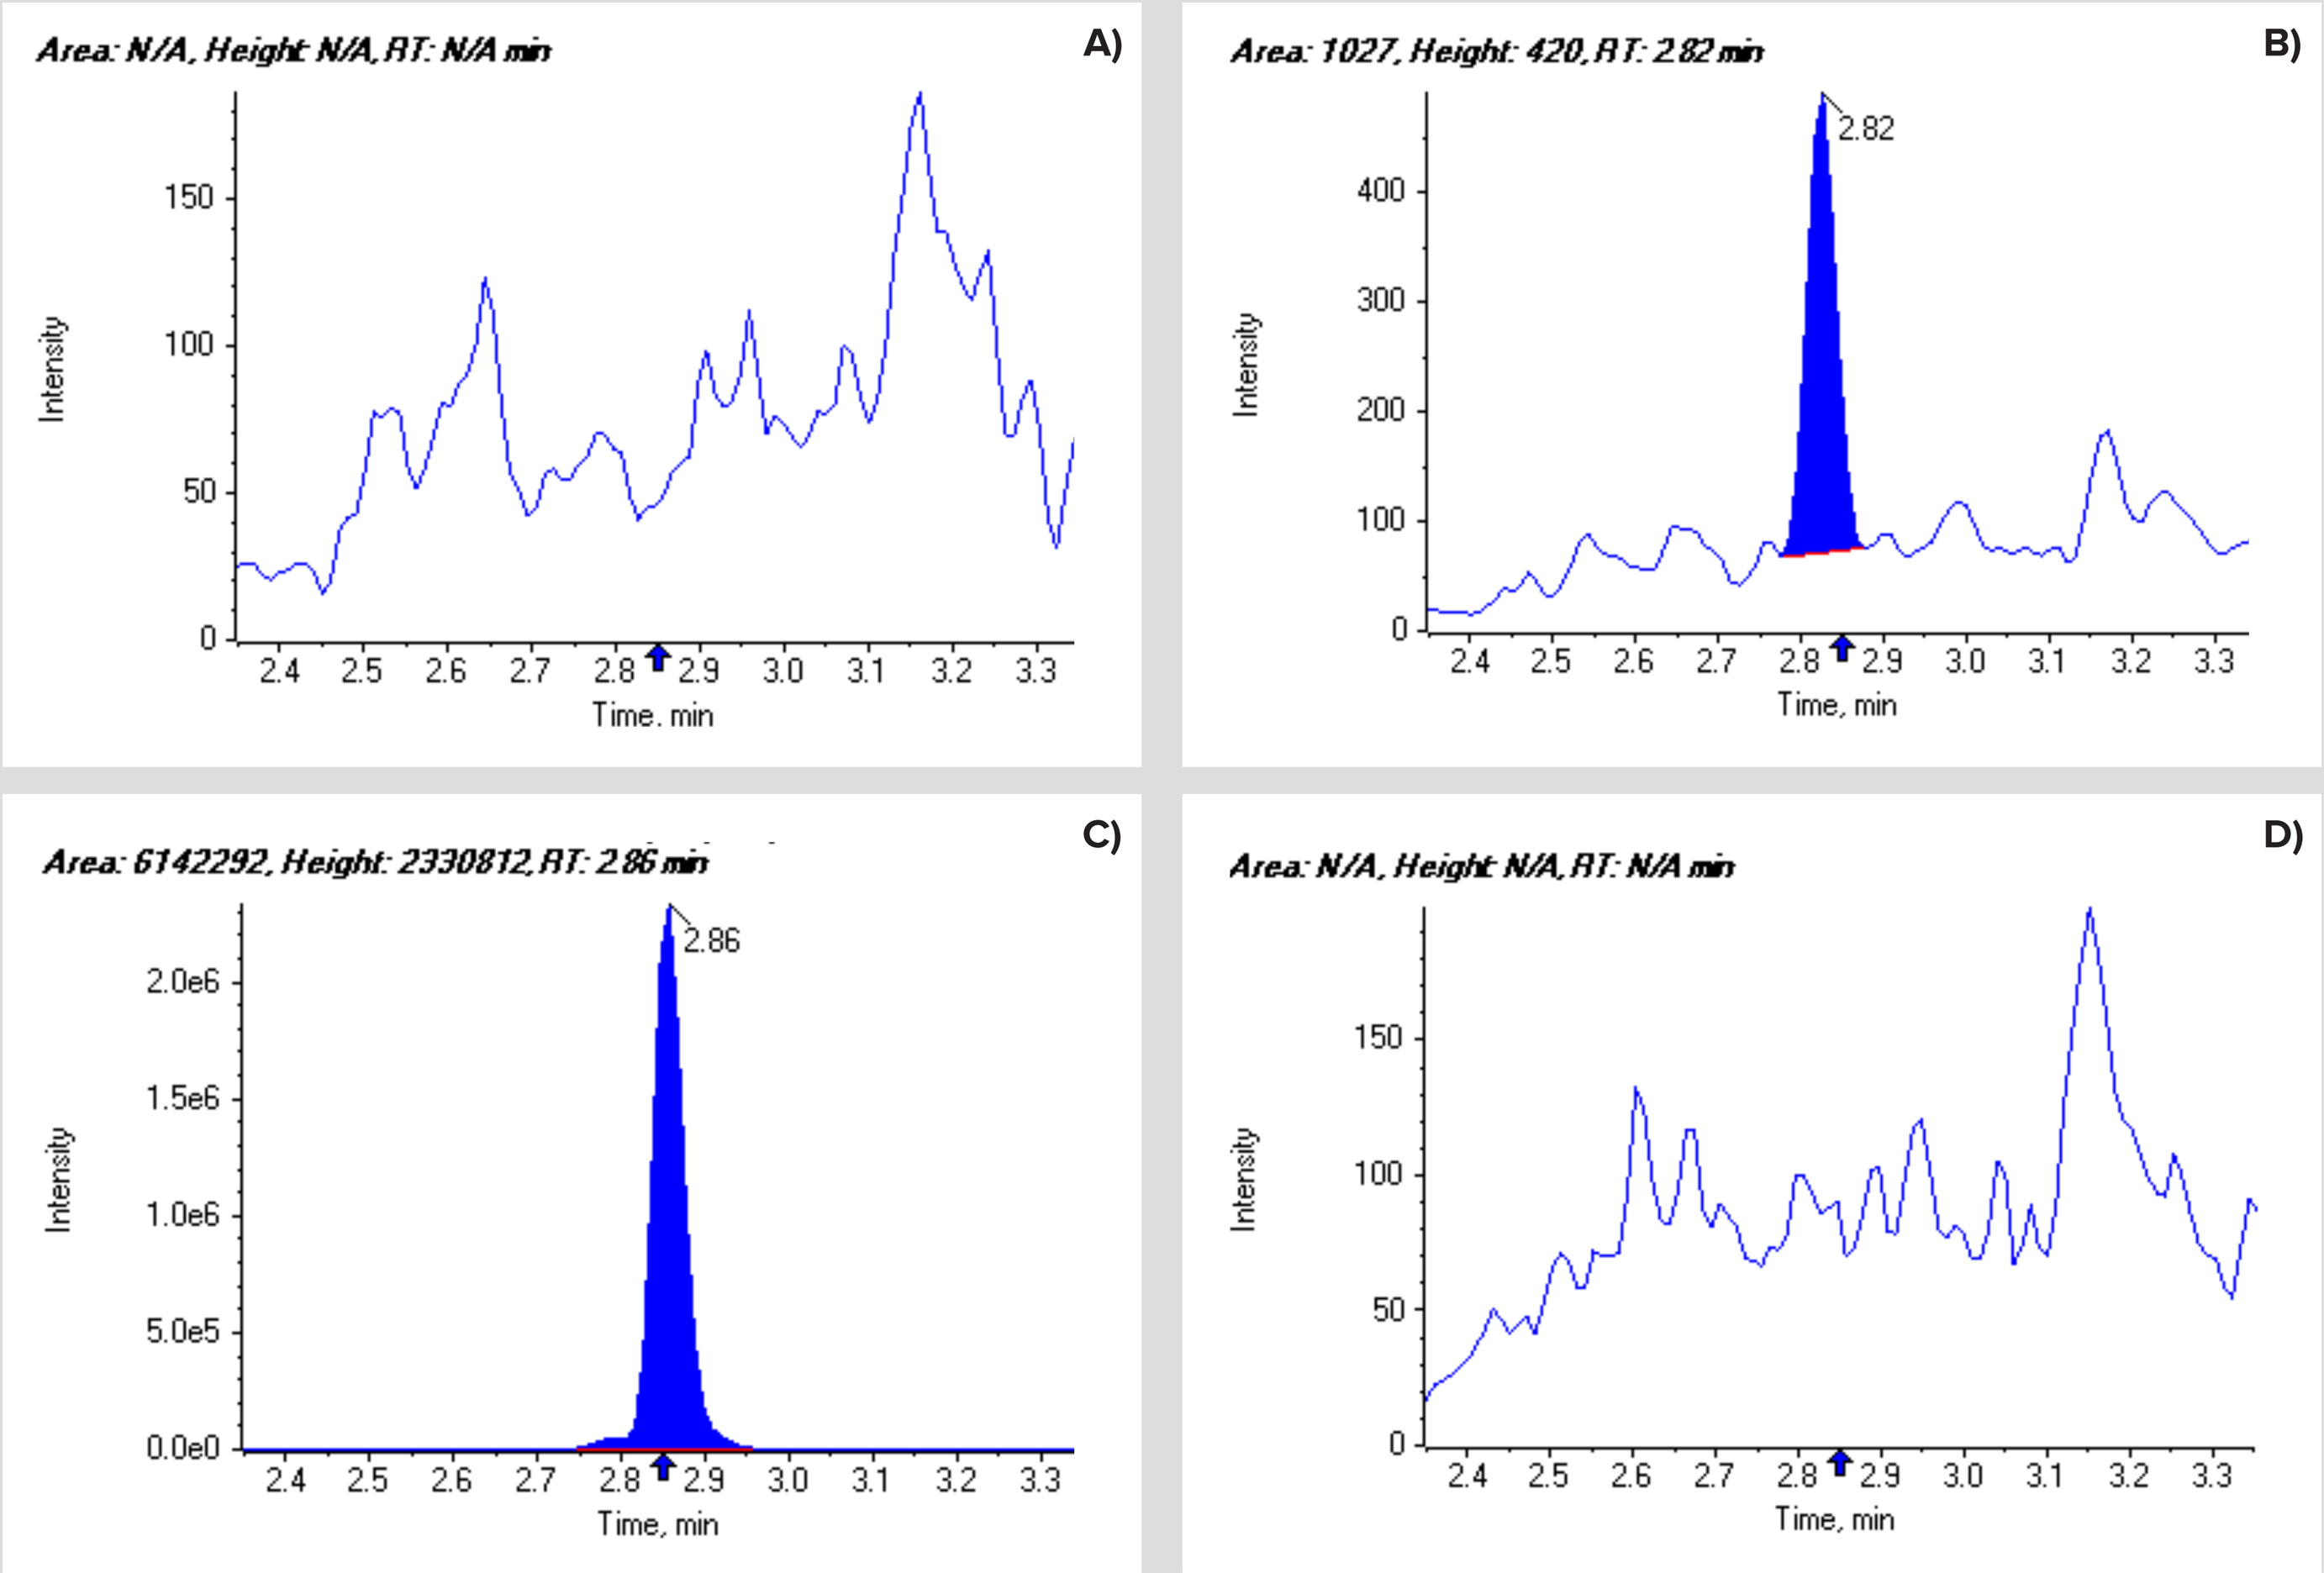
Area: N/A, Height: N/A, RT: N/A min
A)
Area: 1027, Height: 420, RT: 2.82 min
B)
2.82
Area: 6142292, Height: 2330812, RT: 2.86 min
C)
2.86
Area: N/A, Height: N/A, RT: N/A min
D)
Intensity
Time, min

Figure 5. A) Mouse plasma, double blank; **B)** LLOQ spiked at 100ng/mL; **C)** ULOQ spiked at 500µg/mL; **D)** 1st carryover blank after 3 ULOQ injected.

ASSAY OPTIMIZATION, AUTOMATION

Building on our implementation of a generic and sensitive assay for ADC analysis by direct digestion, we have further enhanced our workflow by incorporating the Agilent AssayMAP Bravo liquid handler from Agilent. Using the built-in reformatting software, standard and QC samples were prepared independently, with volumes easily adjusted based on the number of plates to be extracted.

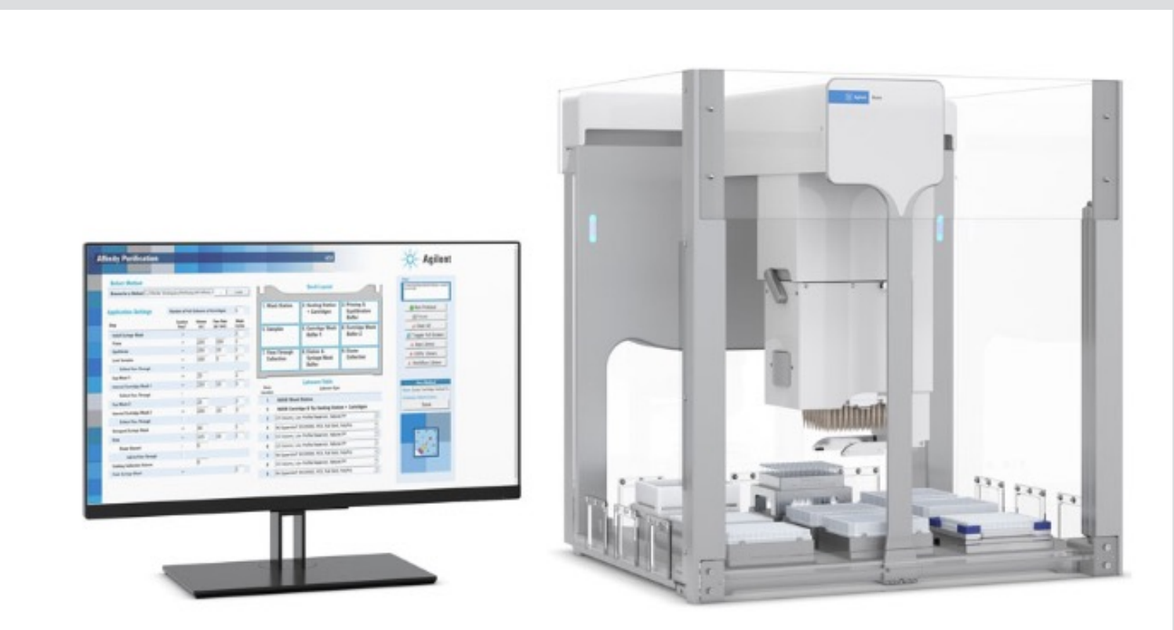

Figure 6. Agilent Bravo Assay Map standard configuration and software[4].

The integration of the AssayMAP Bravo was shown to improve overall efficiency by reducing manual errors and ensuring consistent sample treatment. To evaluate performance, the same calibration curve was spiked manually and using the AssayMAP Bravo, then extracted with the developed protocol. Results were compared for precision and accuracy.

PRECISION AND ACCURACY, MANUAL VS. AUTOMATED SPIKING

Table 1. Precision and accuracy run, manual spiking, Peak area only.

Manual Spiking	Standards (µg/mL)										Quality Controls matrix (µg/mL)				
	STD1	STD2	STD3	STD4	STD5	STD6	STD7	STD8	STD9	STD10	QC-LLOQ	QC-1	QC-2	QC-3	QC-4
	0.100	**0.200**	**0.500**	**1.00**	**4.00**	**12.50**	**50.00**	**150.00**	**400.00**	**500.00**	**0.100**	**0.300**	**1.50**	**10.00**	**375.00**
											0.106	0.260	1.52	10.98	355.75
TTPPVLDSDGSFFLYSK	0.107	0.182	0.435	1.03	4.10	12.29	56.97	147.52	416.39	469.94	0.096	0.293	1.55	10.47	361.11
											0.102	0.307	1.52	10.29	362.48
n	1	1	1	1	1	1	1	1	1	1	3	3	3	3	3
Mean	0.107	0.182	0.435	1.030	4.100	12.290	56.970	147.52	416.390	469.94	0.101	0.287	1.530	10.580	359.78
SD	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	0.005	0.024	0.017	0.358	3.557
% C.V.	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	5.0	8.4	1.1	3.4	1.0
% bias	7.0	-9.0	-13.0	3.0	2.5	-1.7	13.9	-1.7	4.1	-6.0	1.3	-4.4	2.0	5.8	-4.1

Table 2. Precision and accuracy run, Assay Map Spiking, Peak area only.

AssayMap Spiking	Standards (µg/mL)										Quality Controls matrix (µg/mL)				
	STD1	STD2	STD3	STD4	STD5	STD6	STD7	STD8	STD9	STD10	QC-LLOQ	QC-1	QC-2	QC-3	QC-4
	0.100	**0.200**	**0.500**	**1.00**	**4.00**	**12.50**	**50.00**	**150.00**	**400.00**	**500.00**	**0.100**	**0.300**	**1.50**	**10.00**	**375.00**
											0.114	0.306	1.64	10.30	376.01
TTPPVLDSDGSFFLYSK	0.108	0.172	0.503	0.938	3.88	11.98	48.69	158.19	437.72	533.56	0.103	0.279	1.68	9.96	406.64
											0.110	0.279	1.33	9.66	381.51
n	1	1	1	1	1	1	1	1	1	1	3	3	3	3	3
Mean	0.108	0.172	0.503	0.938	3.88	11.98	48.69	158.19	437.72	533.56	0.109	0.288	1.546	9.981	388.057
SD	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	0.006	0.015	0.189	0.320	16.327
% C.V.	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	N/AP	5.3	5.3	12.2	3.2	4.2
% bias	7.5	-13.8	0.6	-6.2	-3.0	-4.2	-2.6	5.5	9.4	6.7	9.0	-4.0	3.1	-0.2	3.5

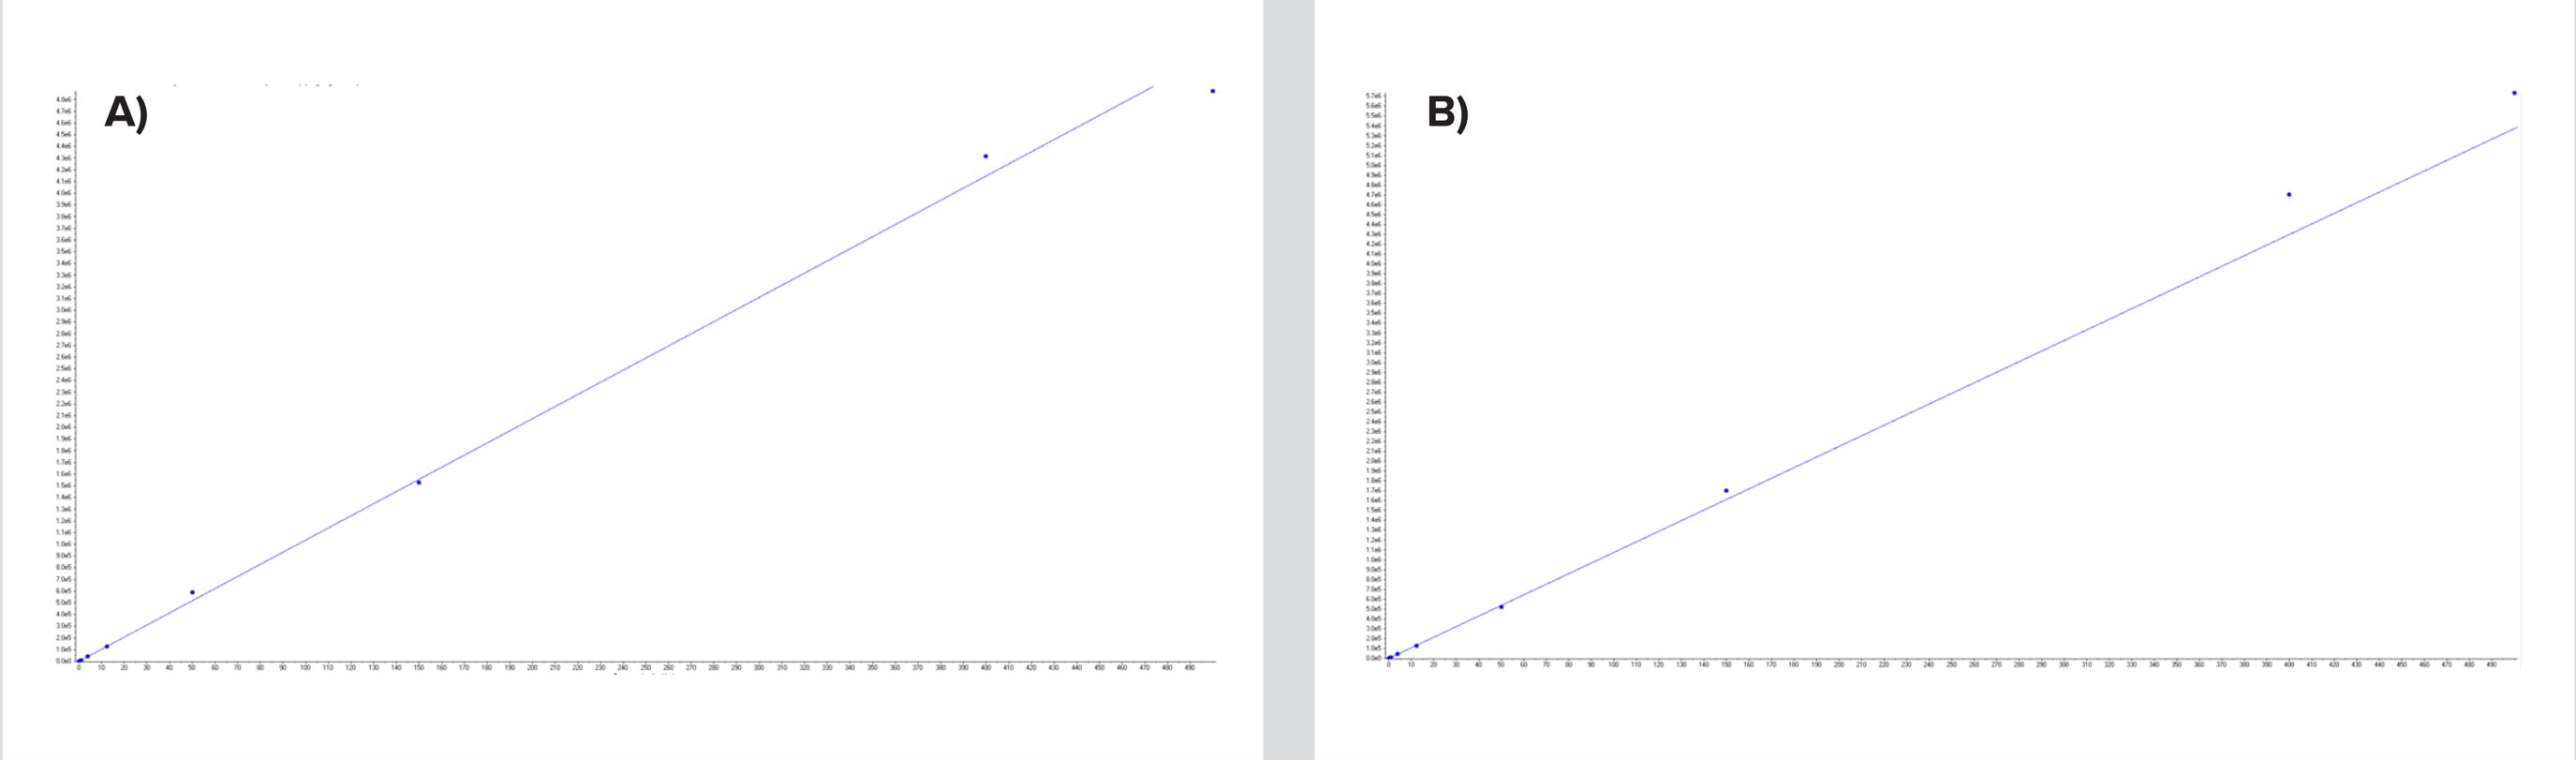
A)
B)

Figure 7. A) Standard curve generated by manually spiking a test molecule into mouse plasma at different concentrations; **B)** Standard curve generated by spiking a test molecule into mouse plasma at different concentrations using Bravo Assay Map platform.

ASSAY OPTIMIZATION, ROBUSTNESS

In mass spectrometry, physical divert valves have traditionally been used to prevent contamination, but they face significant challenges in capillary HPLC (capLC) systems. These valves introduce dead volume, causing peak dispersion and reduced resolution, and disrupt low-flow stability, risking electrospray ionization instability. Moreover, the valves are prone to failure due to high backpressure, require frequent cleaning, and are often incompatible with narrow capillaries. To improve method robustness, making this method suitable for back-to-back analysis, despite having a sample processing with minimal clean-up, we have introduced a virtual divert valve, based on the principle of voltage modulation. This solution offers an elegant approach, by temporarily disabling the ionization voltage, preventing contamination without introducing additional hardware. This technique preserves chromatographic integrity, maintains stable flow, and is independent of backpressure constraints.

Software-controlled voltage modulation significantly reduces source fouling, extending maintenance intervals and enhancing long-term robustness, making it a highly effective and efficient contamination control method for capLC-MS systems.

METHOD ROBUSTNESS

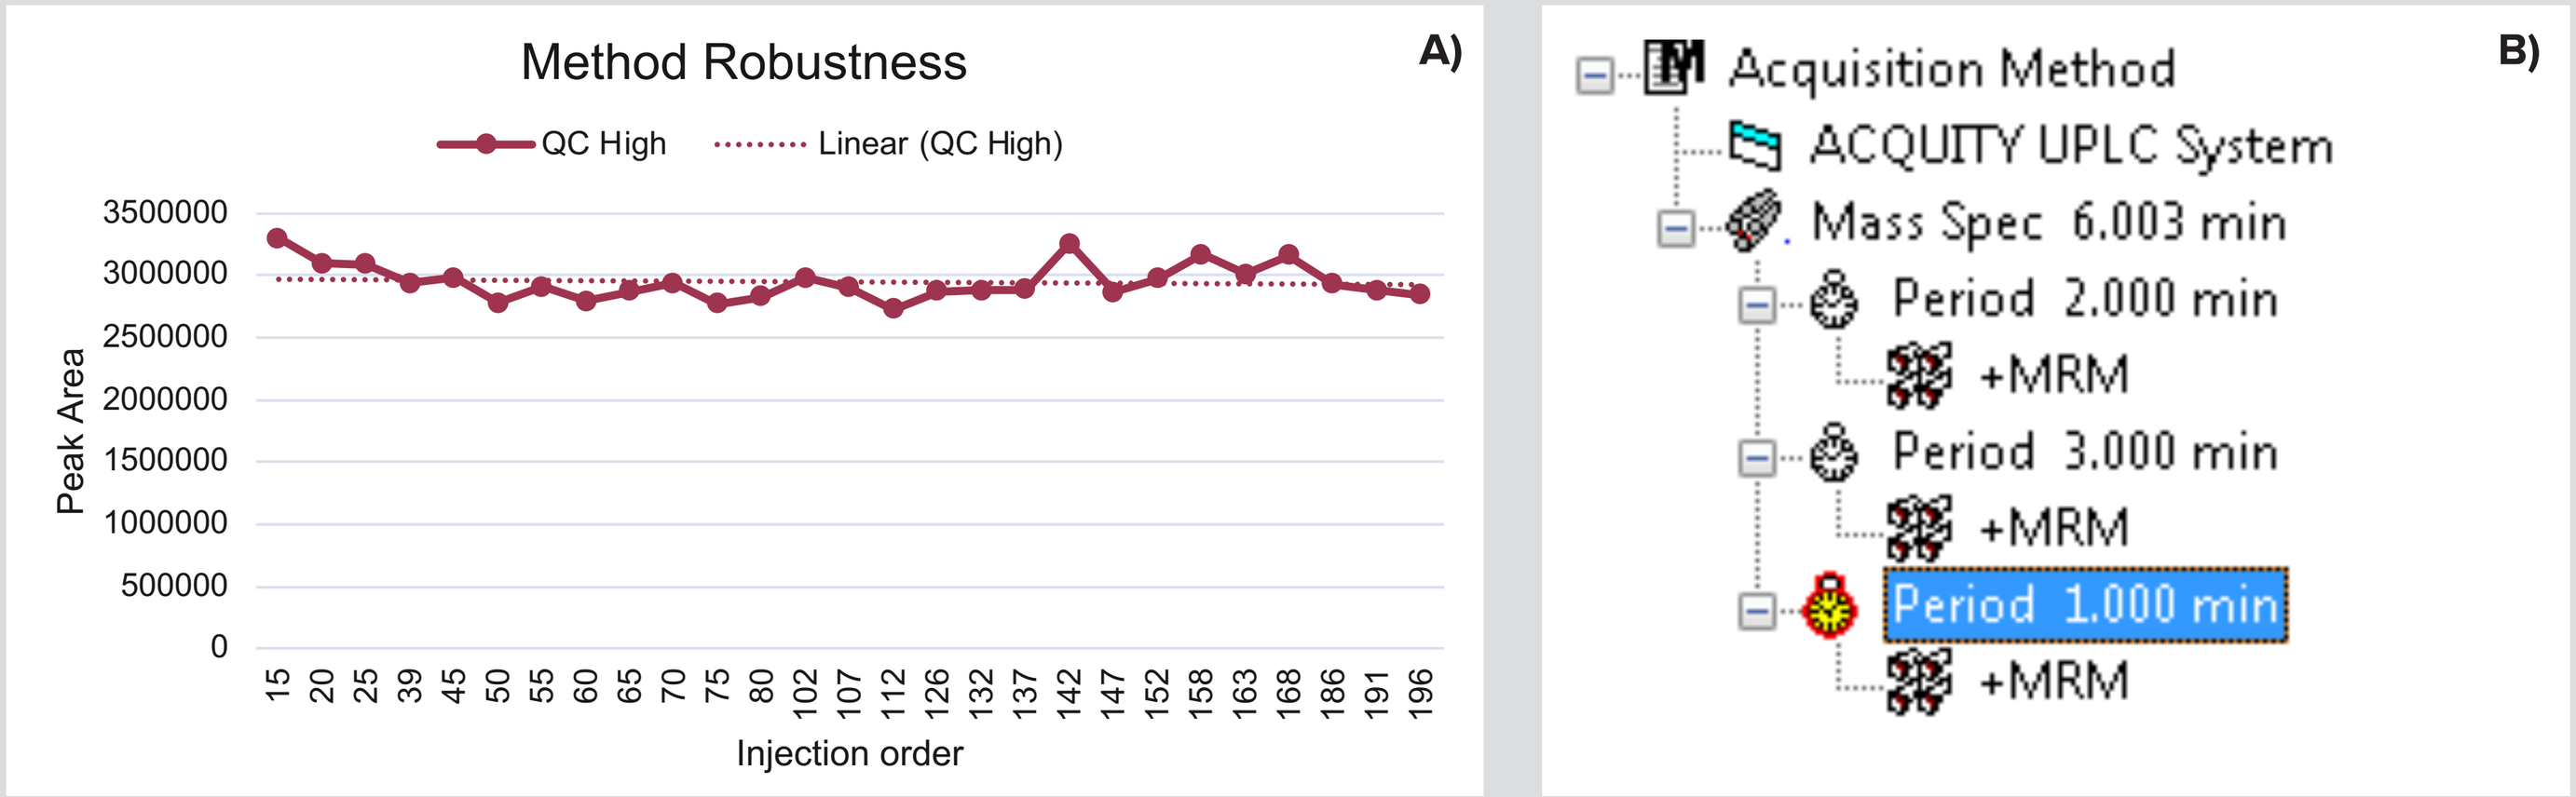
Method Robustness
A)
QC High
Linear (QC High)
Peak Area
Injection order
B)
Acquisition Method
ACQUITY UPLC System
Mass Spec 6.003 min
Period 2.000 min
+MRM
Period 3.000 min
+MRM
Period 1.000 min
+MRM

Figure 8. A) Peak area of 27 QC-High independently extracted and continuously injected with the virtual divert valve; **B)** Structure of the virtual divert valve

PLATFORM COMPARISON

An initial platform comparison demonstrated a promising increase in sensitivity, with a 2X enhancement observed when transitioning from the QTRAP5500 to the QTRAP6500+, resulting in an impressive reduction of the method's LLOQ to the sub 100ng/mL range. To leverage available resources, the method was subsequently further developed and optimized on the QTRAP5500.

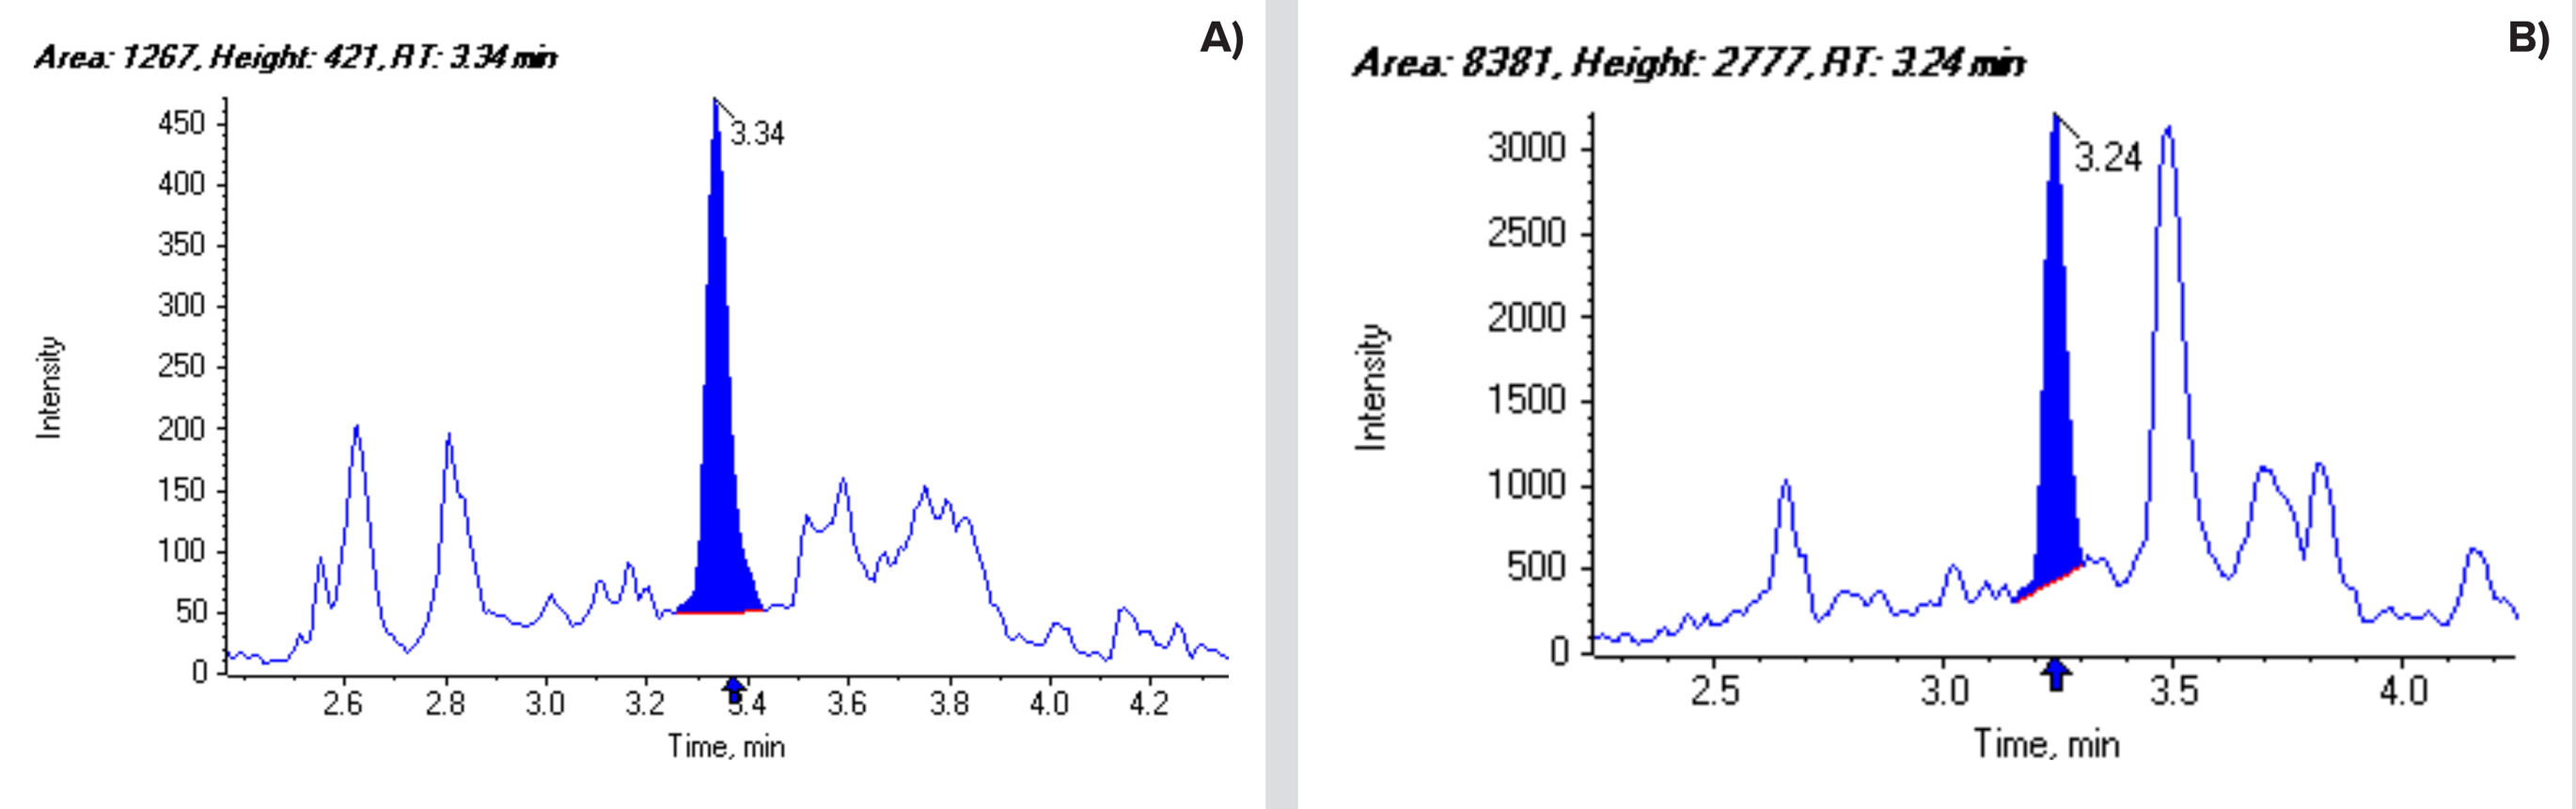
Area: 1267, Height: 421, RT: 3.34 min
A)
3.34
Area: 8381, Height: 2777, RT: 3.24 min
B)
3.24
Intensity
Time, min

Figure 9. A) LLOQ spiked at 100ng/mL and injected on QTRAP5500; **B)** LLOQ spiked at 100ng/mL and injected on QTRAP6500+.

CONCLUSION

- The developed method proved to be fit for purpose, achieving a sensitivity in line with affinity capture assays of 100 ng/mL from just 5 µL of plasma, with good robustness, precision, and accuracy (small molecule criteria when spiking using the AssayMap).
- The current analytical range spans from 0.1 to 500 µg/mL in just 6 minutes, demonstrating broad applicability for ADC and therapeutic antibody quantification. Access to state-of-the-art triple quadrupole instruments could push even further the current level of sensitivity obtained on a QTRAP5500, matching results obtainable by Hybrid LBA/LC-MS or LBA assay.
- Due to the minimal sample preparation steps (no reduction, alkylation, or affinity capture), automated spiking, and low reagent costs (1:100 trypsin ratio), this approach represents a low-cost, high-efficiency, and easily deployable solution for preclinical bioanalysis, that is unaffected by the increased complexity of ADC's.

REFERENCES

1 Cahuzac, H., & Devel, L. (2020). Analytical methods for the detection and quantification of ADCs in biological matrices. Pharmaceuticals, 13(12), 462.
2 Zhao, P., Zhang, Y., Li, W., Jeanty, C., Xiang, G., & Dong, Y. (2020). Recent advances of antibody drug conjugates for clinical applications. Acta Pharmaceutica Sinica B, 10(9), 1589-1600.
3 Image retrieved from Summary of ADC Targets For Solid Tumors & Hematological Tumors, https://www.biochempeg.com/article/356.html
4 Image retrieved from https://www.agilent.com/cs/library/usermanuals/public/AssayMAP_BravoPlatform_GetStarted_D0000352.pdf

Please send inquiries to
Luca Genovesi, Ph.D.
R&D Group Leader
luca.genovesi@Cellcarta.com

DAY3-P14
www.cellcarta.com